An Engineered Bi-Functional Fusion Protein (CD86-Fc-NKG2a) to Reverse NK Cell Tolerance of Malignant Cells

Casey Shuptrine, John Etchart, Zachary Opheim, Arpita Patel, Kyung Jin Yoo, Suresh de Silva, George Fromm, Josiah Hornblower, Taylor Schreiber ***Shattuck Labs, Inc.*** Austin, TX & Research Triangle Park, NC

Contact: TSchreiber@shattucklabs.com

#P810

Abstract

Background: As clinical outcomes improve for immunotherapeutic regimens based on PD-1/L1 blockade, the proportion of cancer patients who develop resistance to PD-1/L1 blockade will increase. Some resistance mechanisms, including MHC-I and β2M downregulation, reflecting the nature of PD-1/L1 as a T-cell centric checkpoint. Recent clinical success by groups targeting non-T cell immune cells has highlighted the potential of enhancing macrophage or NK cell targeted immunotherapies for the treatment of a wide variety of cancers. Future combinatorial approaches will likely utilize agents targeting multiple different immune cells to effectively arm both the adaptive and the innate immune systems. As a prime example of this, the anti-NKG2a monoclonal antibody Monalizumab, has shown promising preclinical and clinical anti-cancer effects, especially when combined with an ADCC competent antibody [1]. Therefore, targeting non-classical innate immune checkpoints has the potential to synergize with currently approved cancer therapies. Here we report the generation of a two-sided fusion protein, CD86-Fc-NKG2a, which was designed to provide competitive inhibition of the NK-centric HLA-E checkpoint, which providing co-stimulation to T cells via CD28.

Methods: Human and mouse variants of CD86-Fc-NKG2a were produced and characterized using a variety of biochemical assays to determine the correct molecular weight, subunit composition and binding affinity; molecular assays to characterize in vitro cell binding, in vitro functional activity, in vitro lysis capabilities; and anti-tumor efficacy in multiple syngeneic model systems.

Results: The NKG2a domain bound recombinant and cell-expressed HLA-E with high affinity. The CD86 domain bound recombinant CD28 and CTLA-4, with a higher affinity for CTLA-4. When tested head-to-head in a therapeutic CT26 murine tumor model, mCD86-Fc-NKG2a demonstrated stronger anti-cancer activity compared to anti-NKG2a antibody controls. The combination of the CD86-Fc-NKG2a with an anti-PD1 antibody was effective in controlling tumor growth in immune competent mice. In vitro cell lysis assays, with PBMCs, have further elucidated the effect of CD86-Fc-NKG2a on NK cell mediated tumor cell death.

Fusion Protein Platform

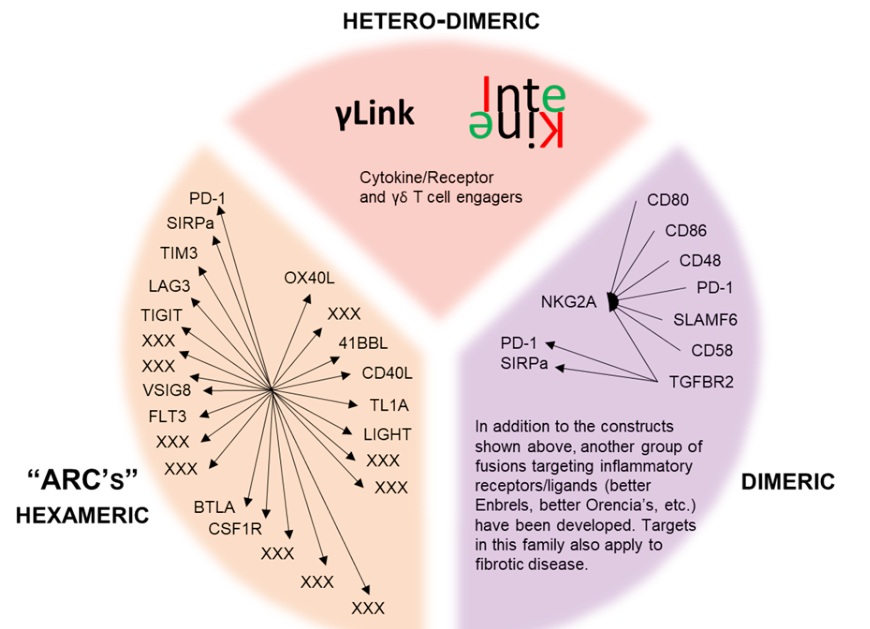

HETERO-DIMERIC
γLink
Cytokine/Receptor and γδ T cell engagers
"ARC's"
HEXAMERIC
DIMERIC
PD-1
SIRPa
TIM3
LAG3
TIGIT
VSIG8
FLT3
BTLA
CSF1R
OX40L
41BBL
CD40L
TL1A
LIGHT
XXX
NKG2A
CD80
CD86
CD48
SLAMF6
CD58
TGFBR2
In addition to the constructs shown above, another group of fusions targeting inflammatory receptors/ligands (better Enbrels, better Orencia's, etc.) have been developed. Targets in this family also apply to fibrotic disease.

Figure 1. ARC Platform. Agonist Redirected Checkpoints consist of a type I membrane protein extra-cellular domain (ECD), linked to a type II ECD, via a central domain (i.e. Fc). Over 250 unique constructs have been synthesized to date, and include combinations of all checkpoint molecules, the entire family of TNFSF ligands, and NK stimulatory molecules.

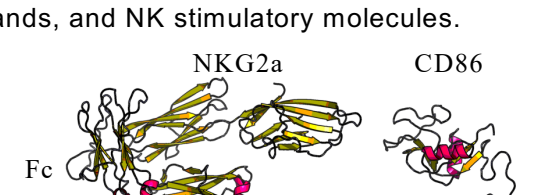

NKG2a
CD86
Fc

Figure 2. Dimeric ARC Structure. Tertiary structure prediction of the CD86-Fc-NKG2a ARC.

In Vitro Characterization and Function

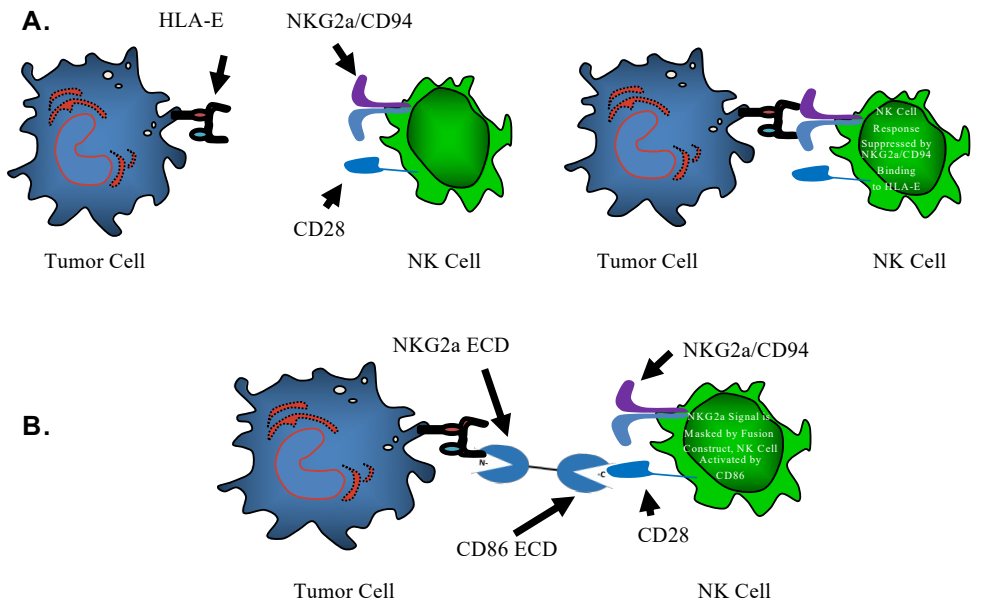

A.
HLA-E
NKG2a/CD94
CD28
Tumor Cell
NK Cell
NK Cell Response Suppressed by NKG2a/CD94 Binding to HLA-E
B.
NKG2a ECD
CD86 ECD
CD28
NKG2a Signal is Masked by Fusion Construct, NK Cell Activated by CD86

Figure 3. **MOA.** (A) HLA-E can interact with NKG2a expressed on NK cells or T cells to suppress cytolytic cell activation. (B) CD86-Fc-NKG2a can block the immune suppressive signals and simultaneously provide NK/T cell co-stimulation.

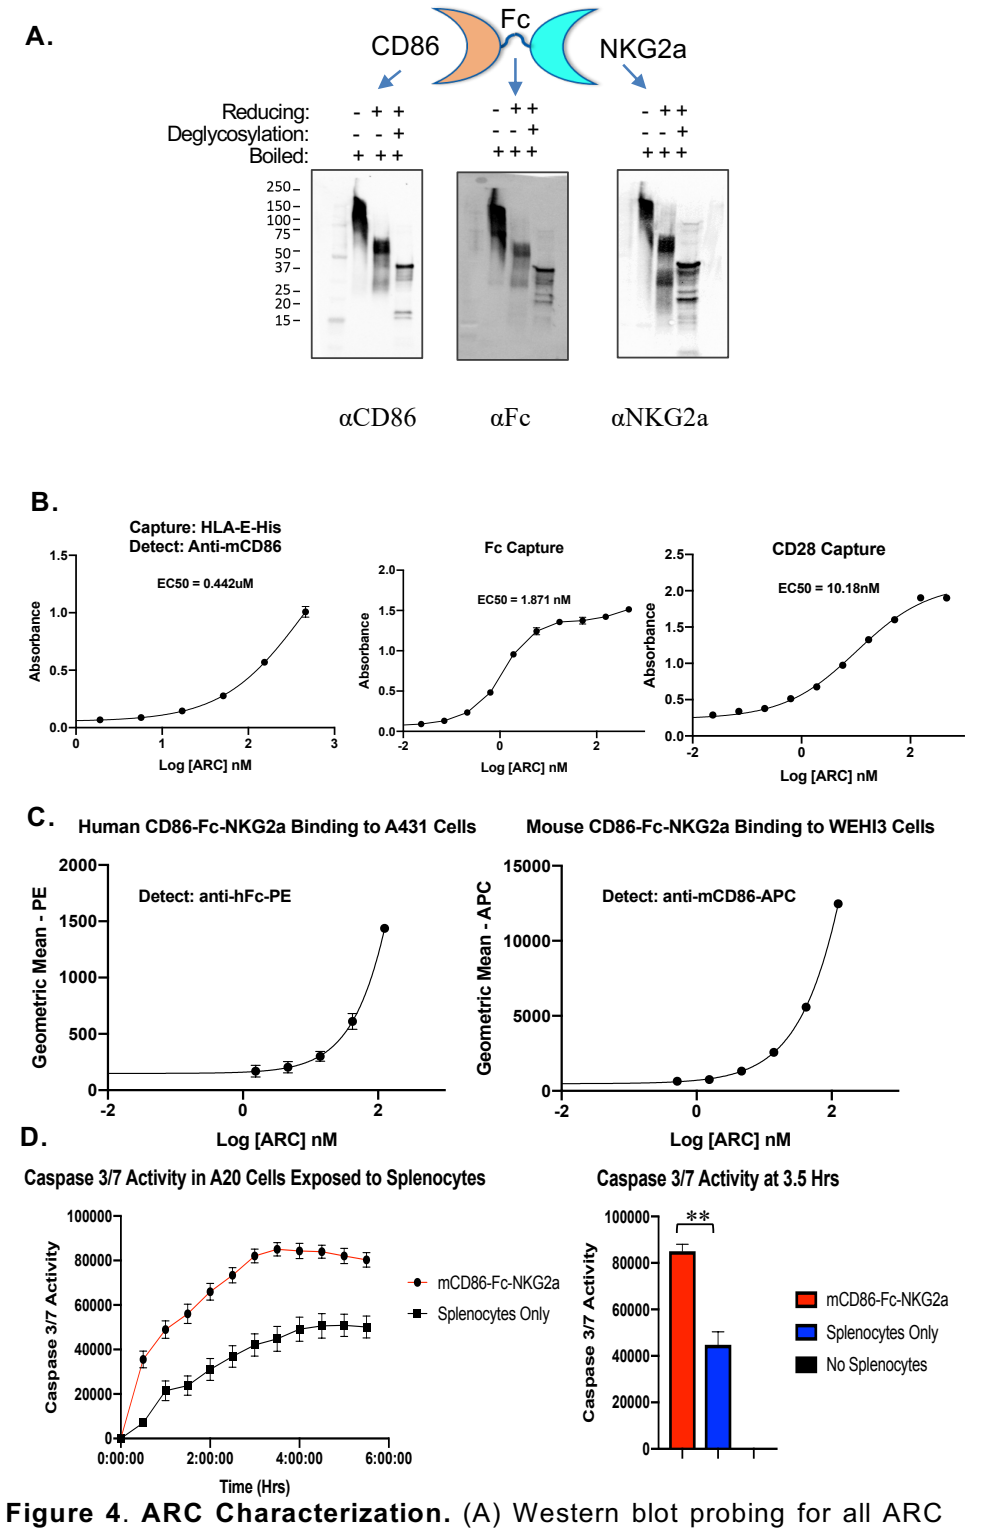

A.
CD86
Fc
NKG2a
Reducing:
Deglycosylation:
Boiled:
αCD86
αFc
αNKG2a
B.
Capture: HLA-E-His
Detect: Anti-mCD86
EC50 = 0.442uM
Fc Capture
EC50 = 1.871 nM
CD28 Capture
EC50 = 10.18nM
Absorbance
Log [ARC] nM
C.
Human CD86-Fc-NKG2a Binding to A431 Cells
Detect: anti-hFc-PE
Geometric Mean - PE
Mouse CD86-Fc-NKG2a Binding to WEHI3 Cells
Detect: anti-mCD86-APC
Geometric Mean - APC
D.
Caspase 3/7 Activity in A20 Cells Exposed to Splenocytes
Caspase 3/7 Activity
Time (Hrs)
mCD86-Fc-NKG2a
Splenocytes Only
Caspase 3/7 Activity at 3.5 Hrs
No Splenocytes

Figure 4. **ARC Characterization.** (A) Western blot probing for all ARC domains. (B) Representative ELISA binding of HLA-E and CD28. (C) CD86-Fc-NKG2a binds to the cell surface of HLA-E (Qa-1 for mouse) positive tumor cells. (D) CD86-Fc-NKG2a enhances splenocyte mediated killing of murine lymphoma cells in co-culture at 0.34 ug/ml. **p<0.01.

Anti-Tumor Efficacy and Synergy with Checkpoint Blockade

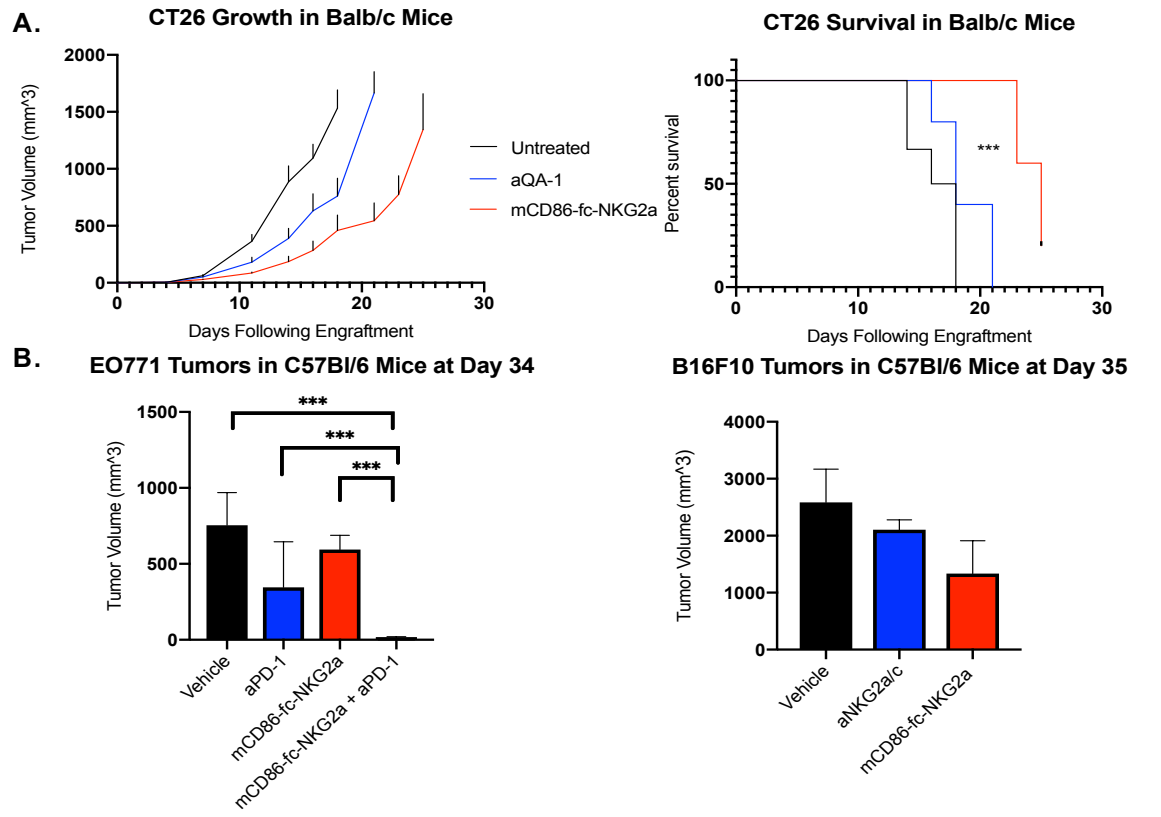

A.
CT26 Growth in Balb/c Mice
Tumor Volume (mm^3)
Days Following Engraftment
Untreated
aQA-1
mCD86-fc-NKG2a
CT26 Survival in Balb/c Mice
Percent survival
B.
EO771 Tumors in C57Bl/6 Mice at Day 34
Vehicle
aPD-1
mCD86-fc-NKG2a
mCD86-fc-NKG2a + aPD-1
B16F10 Tumors in C57Bl/6 Mice at Day 35
aNKG2a/c

Figure 5. Anti-tumor efficacy (CT26, EO771, B16F10). 3 treatments of ARCs (300 μg each) and/or antibodies (100 μg each) were given on days 7, 10, and 14 following tumor inoculations, when starting tumor volumes were **equivalent**. Monotherapies and combinations with (B) anti-PD1 (RMP1-14) are shown, along with survival.

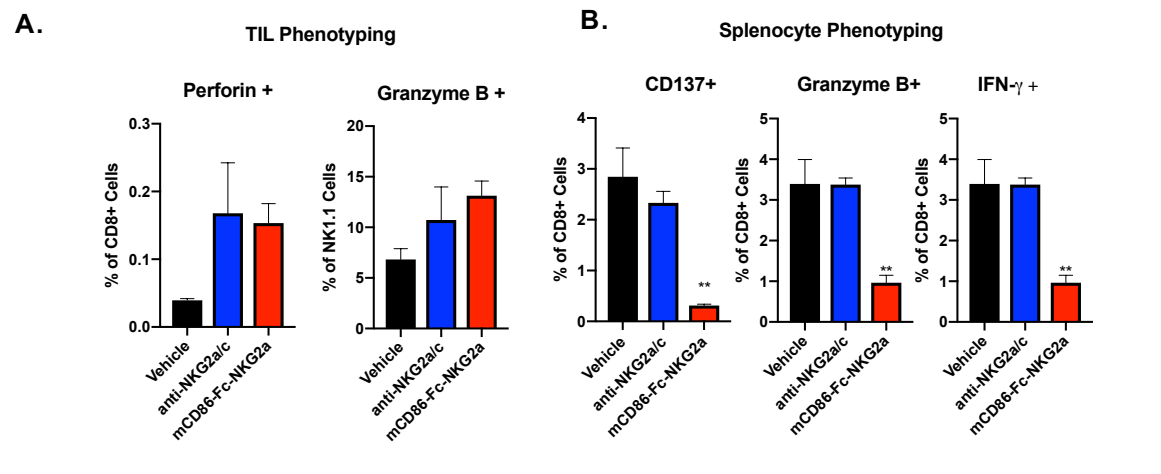

A.
TIL Phenotyping
Perforin +
Granzyme B +
% of CD8+ Cells
% of NK1.1 Cells
B.
Splenocyte Phenotyping
CD137+
Granzyme B+
IFN-γ +
Vehicle
anti-NKG2a/c
mCD86-Fc-NKG2a

Figure 5. Immune Phenotyping. (A) B16F10 tumors (day 7 post treatment) and Splenocytes (day 7 post treatment) collected from B16F10 bearing mice treated with mCD86-Fc-NKG2a and/antibodies as above, and phenotyped using flow cytometry. **p<0.01

Summary

- ***ARCs*** are a novel class of bi-functional biologics capable of targeting type-I and type-II membrane proteins; and can target all checkpoint molecules, the entire family of TNFR superfamily receptors, and NK stimulatory molecules.

- **CD86-Fc-NKG2a** can block HLA-E/NKG2a interactions on NK and T cells, while simultaneously delivering a potent co-stimulatory signal through CD28, resulting in enhanced tumor cell killing and antitumor in vivo activity.

References

1. Andre´ P, Denis C, Soulas C, *et al*. Anti-NKG2A mAb is a Checkpoint Inhibitor that Promotes Anti-tumor Immunity by Unleashing Both T and NK Cells. Cell. 2018 Dec 13; 175(7): 1731-1743.e.13.

PhDPosters.com
Your Research Poster, Printed For Less