Co-Stimulation of OX40 or LTβR Reprograms Exhausted Lymphocytes to Acquire an Effector Phenotype in the Setting of Combined TIGIT and Checkpoint Blockade

George Fromm, Suresh de Silva, Arpita Patel, Kyung Jin Yoo, Louis Gonzalez, Casey Shuptrine, Kaiwen Huang, Kellsey Johannes, Sarah Cooper, Kinsley Evans, Zach Opheim, Cameron Garrett, Josiah C. Hornblower & Taylor H. Schreiber

Shattuck Labs, Inc. Austin, TX & Research Triangle Park, NC

Contact: tschreiber@shattucklabs.com

#P703

Background

Recent studies regarding TOX as a key transcription factor guiding T cell exhaustion have shed important light on strategies to reprogram tumor infiltrating lymphocytes. While inhibition of TOX prevented exhaustion, it did not result in acquisition of an effector phenotype; which may explain why additional inhibition of TIGIT/LAG3/TIM3 in the setting of PD-1 blockade has not yielded marked clinical improvement. Instead, these results hint that while checkpoint blockade may be required to destabilize exhaustion, pushing exhausted lymphocytes toward an effector phenotype may require co-stimulatory signaling. To explore this, we developed two ***Agonist Redirected Checkpoint (ARC)*** constructs which combine TIGIT/PVR inhibition with either OX40L or LIGHT mediated co-stimulation.

Shattuck synthesized both murine and human versions of **TIGIT-Fc-OX40L** and **TIGIT-Fc-LIGHT**, and each domain of the human ARCs bound human/cyno PVR, OX40, LTβR, and HVEM with low nanomolar affinity. Due to the hexameric structure, the OX40L domain clustered OX40 receptors and stimulated Fc receptor-independent NFκB signaling, and induced IL-2, TNFα, and IFNγ secretion from human PBMCs in the presence of SEB. **TIGIT-Fc-LIGHT** uniquely induced non-canonical NFκB/NIK signaling, stimulated CCL2/CXCL8 expression and IL-8 secretion from A375 cells, and induced antigen-independent proliferation and differentiation of CD8-depleted human PBMCs in AIMV media. Both murine TIGIT-Fc-OX40L and TIGIT-Fc-LIGHT were effective at controlling CT26 and B16.F10 growth and programed a memory response that mediated the rejection of secondary tumor re-challenge. Anti-tumor efficacy of both ARCs was enhanced when combined with checkpoint blockade of PD1 or CTLA4; resulting in complete tumor rejection of large established tumors. Lastly, both ARCs have concluded non-human primate dose-range finding studies and have demonstrated distinct on-target PD activity.

ARC Platform

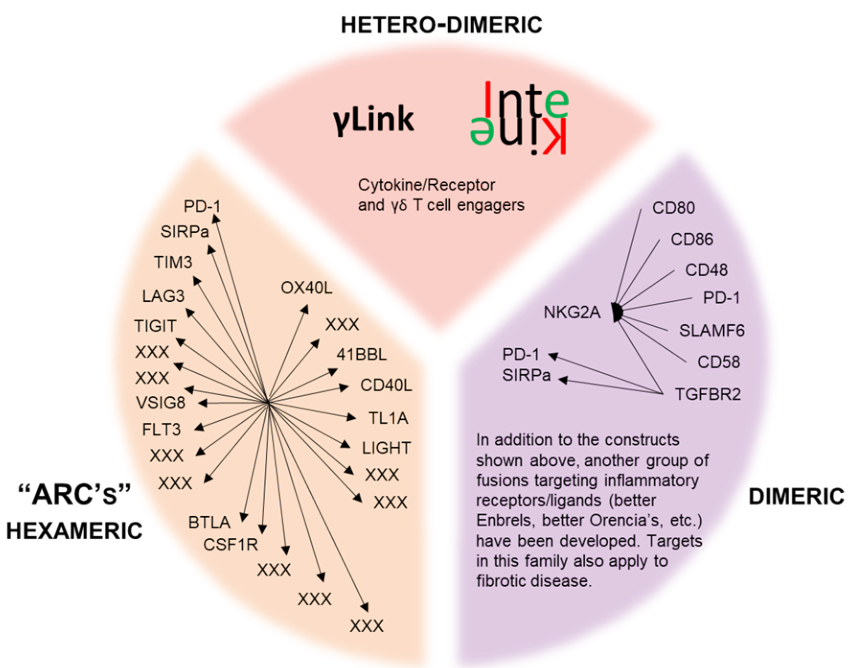

HETERO-DIMERIC
γLink
Cytokine/Receptor and γδ T cell engagers
"ARC's"
HEXAMERIC
PD-1
SIRPa
TIM3
LAG3
TIGIT
VSIG8
FLT3
BTLA
CSF1R
OX40L
41BBL
CD40L
TL1A
LIGHT
XXX
DIMERIC
NKG2A
CD80
CD86
CD48
PD-1
SLAMF6
CD58
TGFBR2
PD-1
SIRPa
In addition to the constructs shown above, another group of fusions targeting inflammatory receptors/ligands (better Enbrels, better Orencia's, etc.) have been developed. Targets in this family also apply to fibrotic disease.

Figure 1. ARC Platform. Agonist Redirected Checkpoints consist of one arm of the Shattuck platform, and link type I membrane protein extra-cellular domains (ECD) to type II ECDs, via a central domain (i.e. Fc). Over 250 unique constructs have been synthesized to date, and include combinations of nearly all checkpoint molecules, and the entire family of TNFSF ligands.

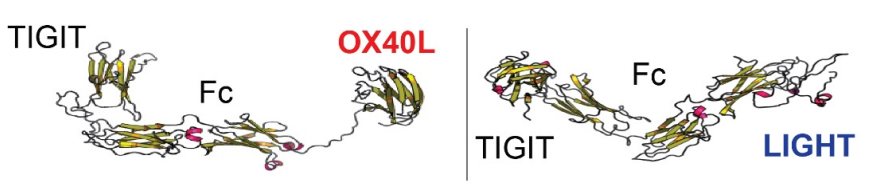

TIGIT
Fc
OX40L
Fc
TIGIT
LIGHT

Figure 2. TIGIT ARC Structures. Tertiary structure prediction of the TIGIT-Fc-OX40L and TIGIT-Fc-LIGHT ARCs.

In Vitro Characterization and Function

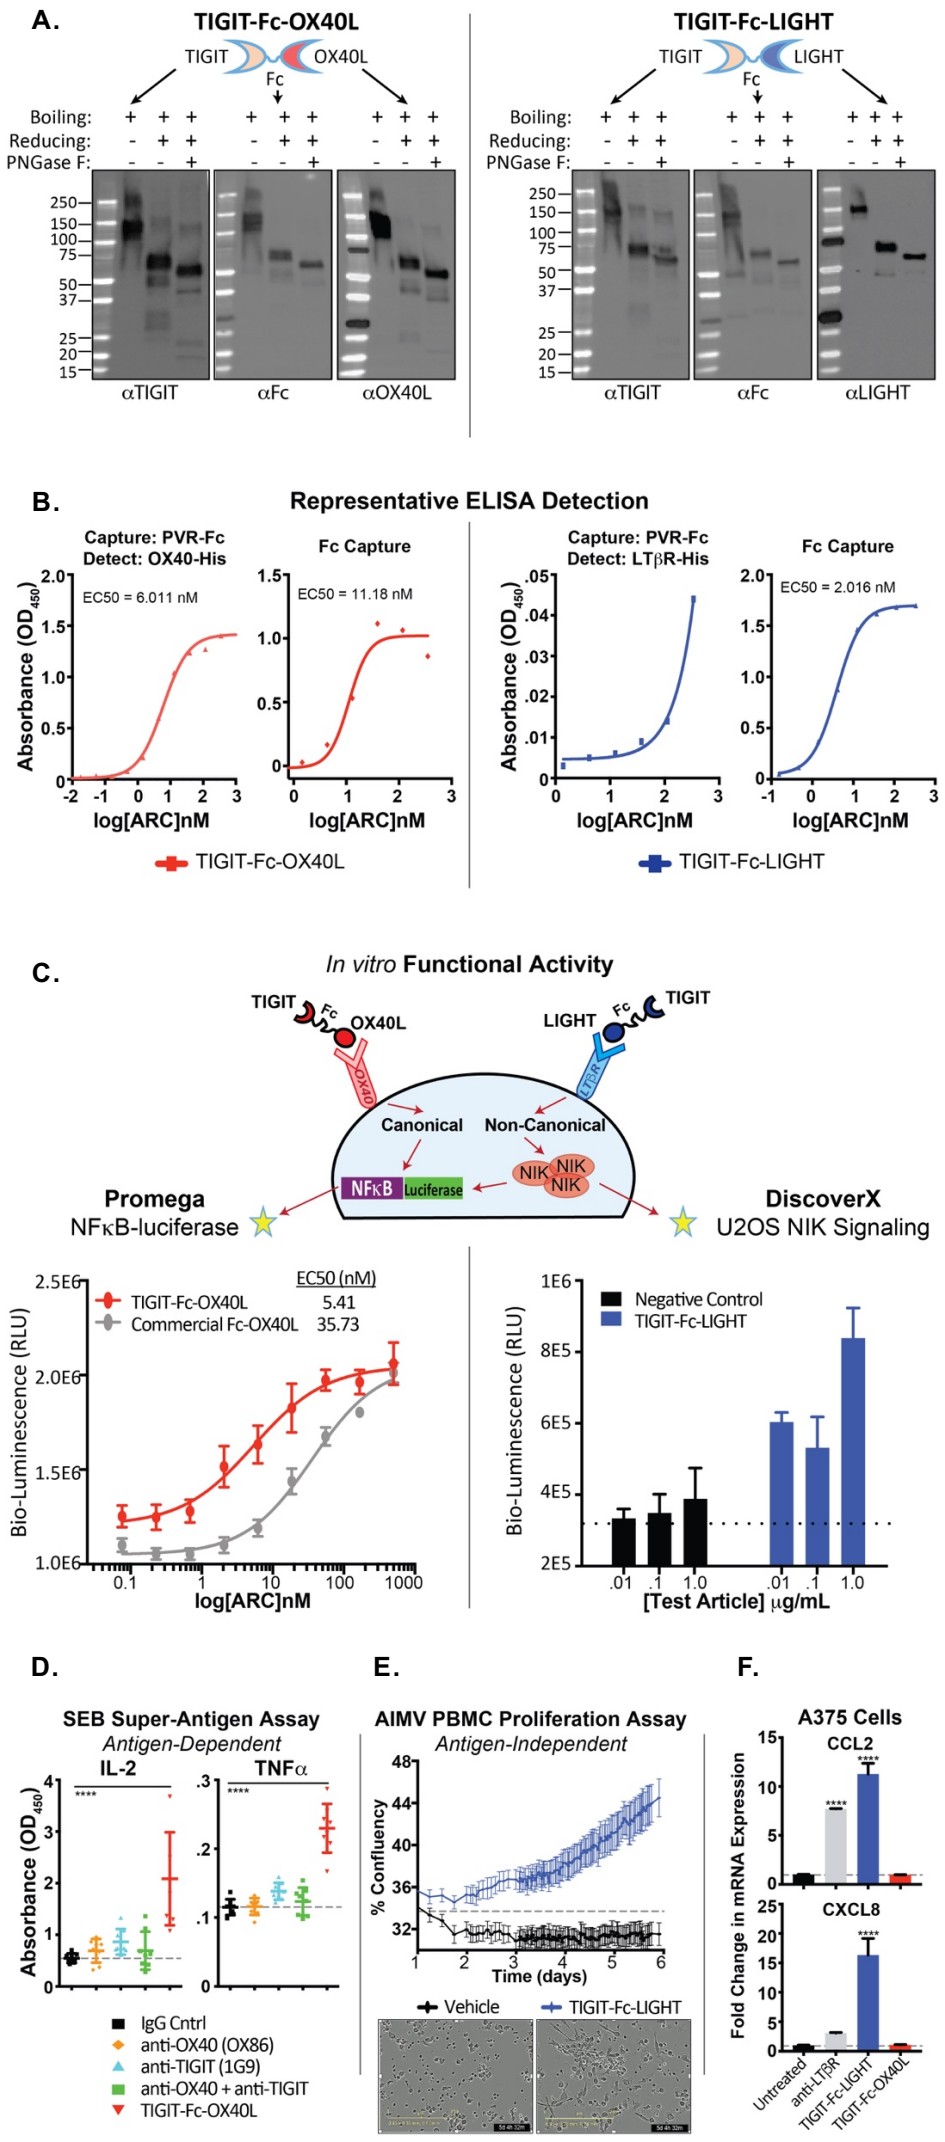

A.
TIGIT-Fc-OX40L
TIGIT-Fc-LIGHT
Boiling:
Reducing:
PNGase F:
αTIGIT
αFc
αOX40L
αLIGHT
B.
Representative ELISA Detection
Capture: PVR-Fc Detect: OX40-His
EC50 = 6.011 nM
Fc Capture
EC50 = 11.18 nM
Capture: PVR-Fc Detect: LTβR-His
EC50 = 2.016 nM
log[ARC]nM
TIGIT-Fc-OX40L
TIGIT-Fc-LIGHT
C.
In vitro Functional Activity
Canonical
Non-Canonical
Promega NFκB-luciferase
DiscoverX U2OS NIK Signaling
EC50 (nM)
TIGIT-Fc-OX40L 5.41
Commercial Fc-OX40L 35.73
Bio-Luminescence (RLU)
Negative Control
TIGIT-Fc-LIGHT
[Test Article] μg/mL
D.
SEB Super-Antigen Assay
Antigen-Dependent
IL-2
TNFα
IgG Cntrl
anti-OX40 (OX86)
anti-TIGIT (1G9)
anti-OX40 + anti-TIGIT
TIGIT-Fc-OX40L
E.
AIMV PBMC Proliferation Assay
Antigen-Independent
% Confluency
Time (days)
Vehicle
TIGIT-Fc-LIGHT
F.
A375 Cells
CCL2
CXCL8
Fold Change in mRNA Expression
Untreated
anti-LTβR
TIGIT-Fc-LIGHT
TIGIT-Fc-OX40L

Figure 3. ARC Characterization. (A) Western blot probing for all ARC domains. (B) Representative ELISA binding of TIGIT ARCs to PVR, OX40, LTβR, and anti-Fc. (C) TIGIT ARCs signal through both the canonical and non-canonical NFkB-signaling pathways; shown here are example assays performed using the Promega and DiscoverX platforms. (D) Mouse splenocytes were cultured with test articles and 200 ng/mL SEB for 3 days, and IL-2/TNFα was assessed from culture supernatant by ELISA. (E) Human CD8-depleted PBMCs were cultured with test articles for 6 days, and proliferation was assessed using IncuCyte. (F) A375 cells were cultured with test articles for 6 hours and gene expression was assessed using qRT-PCR (ΔΔCT vs. GAPDH of untreated; **** p<.0001).

Anti-Tumor Efficacy and Synergy with Checkpoint Blockade

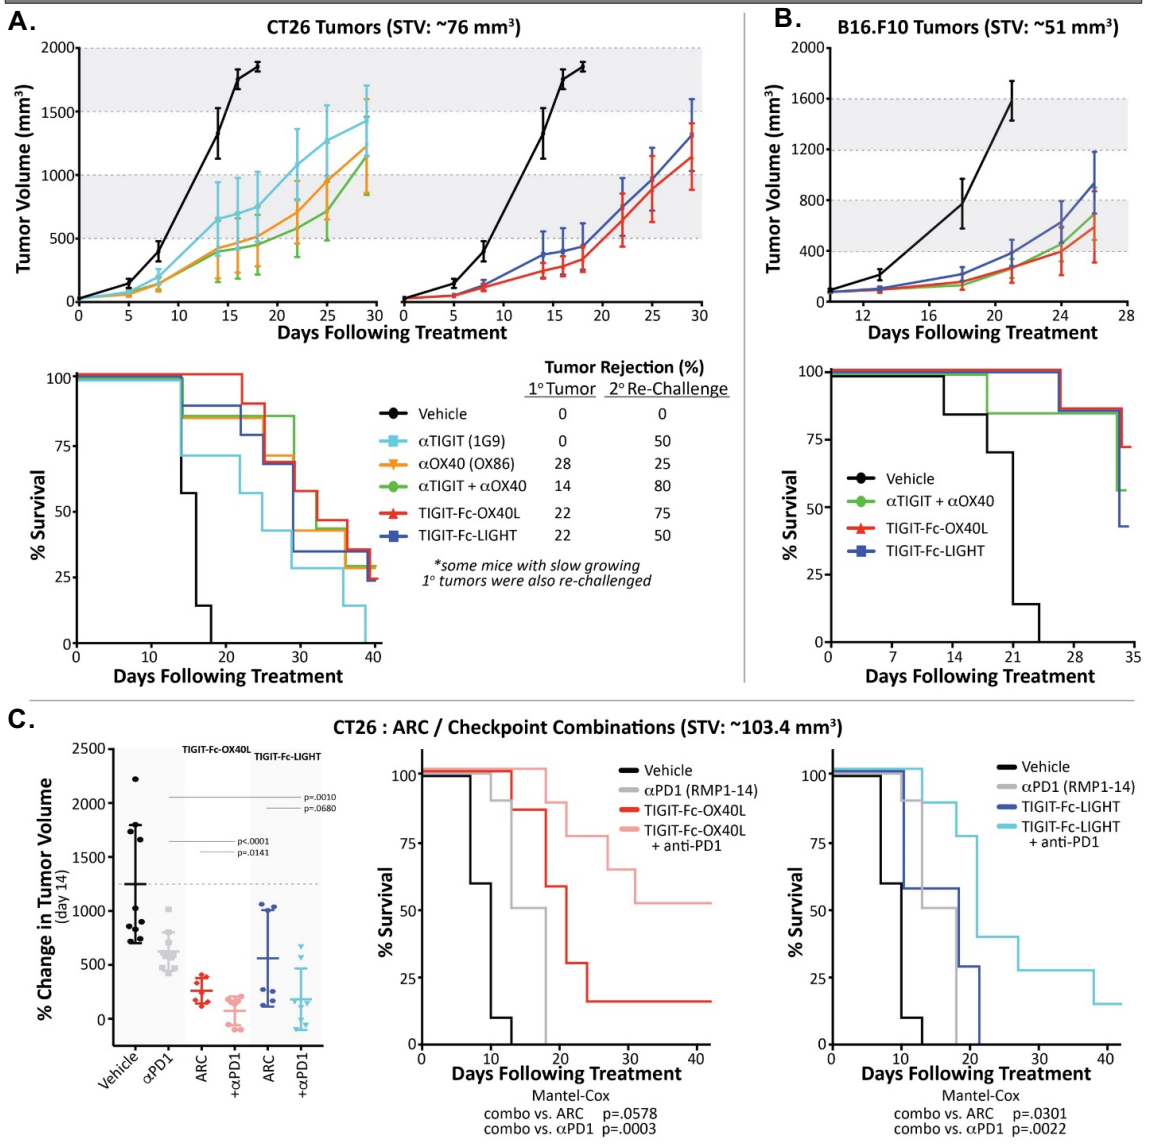

A.
CT26 Tumors (STV: ~76 mm³)
Tumor Volume (mm³)
Days Following Treatment
% Survival
Tumor Rejection (%)
1° Tumor 2° Re-Challenge
Vehicle 0 0
αTIGIT (1G9) 0 50
αOX40 (OX86) 28 25
αTIGIT + αOX40 14 80
TIGIT-Fc-OX40L 22 75
TIGIT-Fc-LIGHT 22 50
*some mice with slow growing 1° tumors were also re-challenged
B.
B16.F10 Tumors (STV: ~51 mm³)
Vehicle
αTIGIT + αOX40
TIGIT-Fc-OX40L
TIGIT-Fc-LIGHT
C.
CT26 : ARC / Checkpoint Combinations (STV: ~103.4 mm³)
% Change in Tumor Volume (day 14)
Vehicle
αPD1
ARC
+αPD1
αPD1 (RMP1-14)
TIGIT-Fc-OX40L + anti-PD1
TIGIT-Fc-LIGHT + anti-PD1
Mantel-Cox
combo vs. ARC p=.0578
combo vs. αPD1 p=.0003
combo vs. ARC p=.0301
combo vs. αPD1 p=.0022

Figure 4. Anti-tumor efficacy. ARCs (300 μg x3) and/or antibodies (100 μg x3) were given (IP) to mice bearing (A) CT26 or (B) B16.F10 tumors. Starting Tumor Volume ***(STV)*** at treatment, average tumor volume (top), and survival curves (bottom) are shown. In the CT26 experiment, mice with small or rejected tumors, were inoculated with a 2nd tumor on day 30 with **no retreatment** - to assess memory response to tumor re-challenge. (C) CT26 tumors were inoculated and allowed to reach a larger STV to assess combination efficacy of ARCs (300 μg x3) with anti-PD1 (RMP1-14; 100 μg x3). Percent change in tumor volume on day 14 of the time course (left) and survival curves (right) are shown. Statistics: unpaired t-tests and Mantel Cox tests.

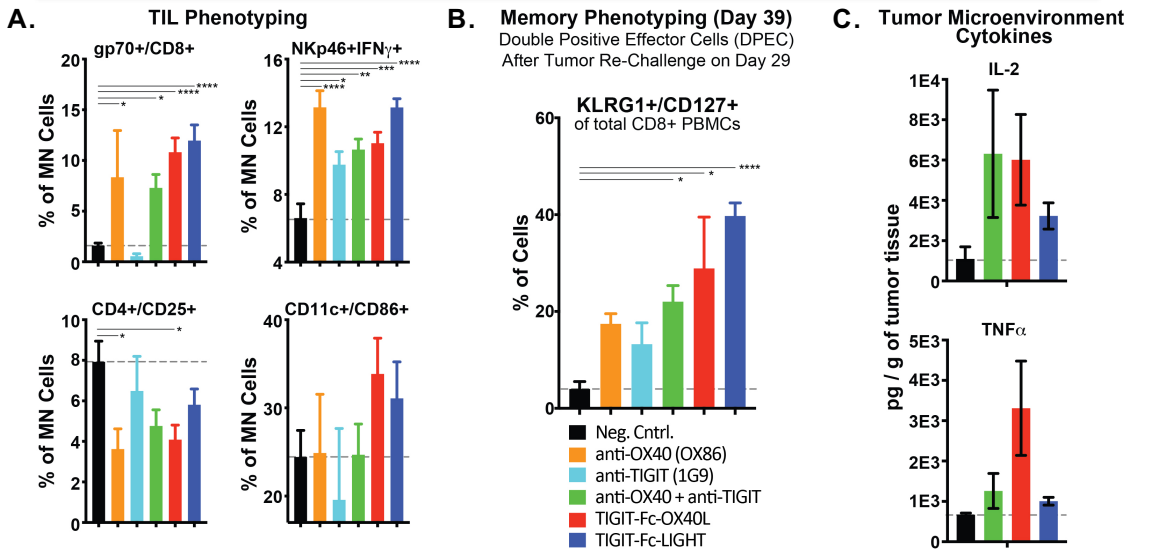

A.
TIL Phenotyping
gp70+/CD8+
NKp46+IFNγ+
CD4+/CD25+
CD11c+/CD86+
% of MN Cells
B.
Memory Phenotyping (Day 39)
Double Positive Effector Cells (DPEC)
After Tumor Re-Challenge on Day 29
KLRG1+/CD127+
of total CD8+ PBMCs
% of Cells
Neg. Cntrl.
anti-OX40 (OX86)
anti-TIGIT (1G9)
anti-OX40 + anti-TIGIT
TIGIT-Fc-OX40L
TIGIT-Fc-LIGHT
C.
Tumor Microenvironment Cytokines
IL-2
TNFα
pg / g of tumor tissue

Figure 5. Immune Phenotyping. (A) CT26 Tumors *(day 9)* and (B) PBMCs *(day 39)* were collected from CT26 bearing mice treated with ARC and/or antibodies as above, and phenotyped using flow cytometry. MN = mononuclear (C) Cytokines from the tumor microenvironment of treated animals were assessed using Luminex (using dissociated tumor supernatant). *p<.05, **p<.01, ***p<.001,****p<.0001.

Conclusions

The addition of **OX40L** or **LIGHT** as co-stimulatory molecules dramatically improved the efficacy of immune checkpoint blockade directed to TIGIT both alone and in combination with PD-1 or CTLA-4 blockade.

Pharmacodynamic evidence from NHP suggests that hexamerized OX40L and LIGHT activate OX40 and LTβR uniquely when compared to existing data from agonist monoclonal antibodies to the same targets, and may uniquely reverse immune exhaustion.

PhDPosters.com