LIGHT (TNFSF14) Co-stimulation Enhances Myeloid Cell Activation and Anti-Tumor Immunity in the Setting of PD-(L)1 and TIGIT Checkpoint Blockade

#585

Kyung Jin Yoo, Kellsey Johannes, Louis Gonzalez, Arpita Patel, Casey Shuptrine, Zachary Opheim, Karen Lenz, Kristen Campbell, Thuy-Ai Nguyen, Jayalakshmi Miriyala, Connor Smith, Fatima Rangwala, Suresh de Silva, Taylor H. Schreiber, and George Fromm

Shattuck Labs, Inc. Austin, TX & Durham, NC

Abstract

Background: Co-inhibition of TIGIT and PD-1/L1 improves response rates compared to monotherapy PD-1/L1 blockade in checkpoint naïve NSCLC with PD-L1 expression >50%. TIGIT mAbs with an effector competent Fc can induce myeloid cell activation, and some have also demonstrated effector T cell depletion, which carries a clinical liability of unknown significance. TIGIT antibody blockade translates to anti-tumor activity by enabling PVR signaling through CD226 (DNAM-1), which can be directly inhibited by PD-1. Further, DNAM-1 is downregulated on TILs in advanced and CPI resistant cancers. Broadening clinical responses from TIGIT blockade into PD-$L1_{low}$ or CPI resistant tumors, may be enhanced by immune co-stimulation that independently operates from PD-1/L1 inhibition.

Methods: Mouse and human TIGIT-Fc-LIGHT molecules were generated and assessed using Octet, MSD, and cell binding assays. Function was evaluated using *in vitro* / *in vivo* activation and anti-tumor efficacy experiments; including a preclinical model of human CPI acquired resistance, and a non-human primate toxicology study.

Results: TIGIT-Fc-LIGHT was nominated using *in vitro* and genomic screening assays that identified TNF costimulatory receptors widely expressed on TILs, T stem cell memory (Tscm), and NK cells; relative to DNAM-1 expression. HVEM was prioritized, and its ligand TNFSF14 (LIGHT), directly activated myeloid cells through binding to a second receptor, LTβR. TIGIT-Fc-LIGHT simultaneously engaged TIGIT and LIGHT receptors at low nanomolar affinities (~3.5-6.5 nM), without the requirement for an effector competent Fc. HVEM signaling overlapped with DNAM-1, and TIGIT-Fc-LIGHT activated NFκB pathways, leading to increased tumor infiltration of antigen-specific CD8+ T and NK cells. Importantly, anti-tumor efficacy induced by monotherapy TIGIT-Fc-LIGHT was maintained in aggressive anti-PD-1 acquired resistant tumors, a model where combined PD-1 and TIGIT antibody blockade was inactive. Because HVEM lacks cytoplasmic domain homology to DNAM-1, HVEM signaling is unlikely to be regulated by PD-1. Indeed, anti-tumor activity of TIGIT-Fc-LIGHT was enhanced by, but not dependent upon, concurrent PD-1/L1 blockade. TIGIT-Fc-LIGHT also directly activated myeloid cells and increased the expression of CXCL10, ITGAX, and B2M, and stimulated cytokines, including IL-2, IFNγ, CCL2, CCL4, IP-10 and CCL17, which was consistent with findings in the serum of treated NHP, where TIGIT-Fc-LIGHT was well tolerated. In NHP, TIGIT-Fc-LIGHT also activated and induced the margination of HVEM+ T cells out of the peripheral circulation.

TNF Receptor Identification

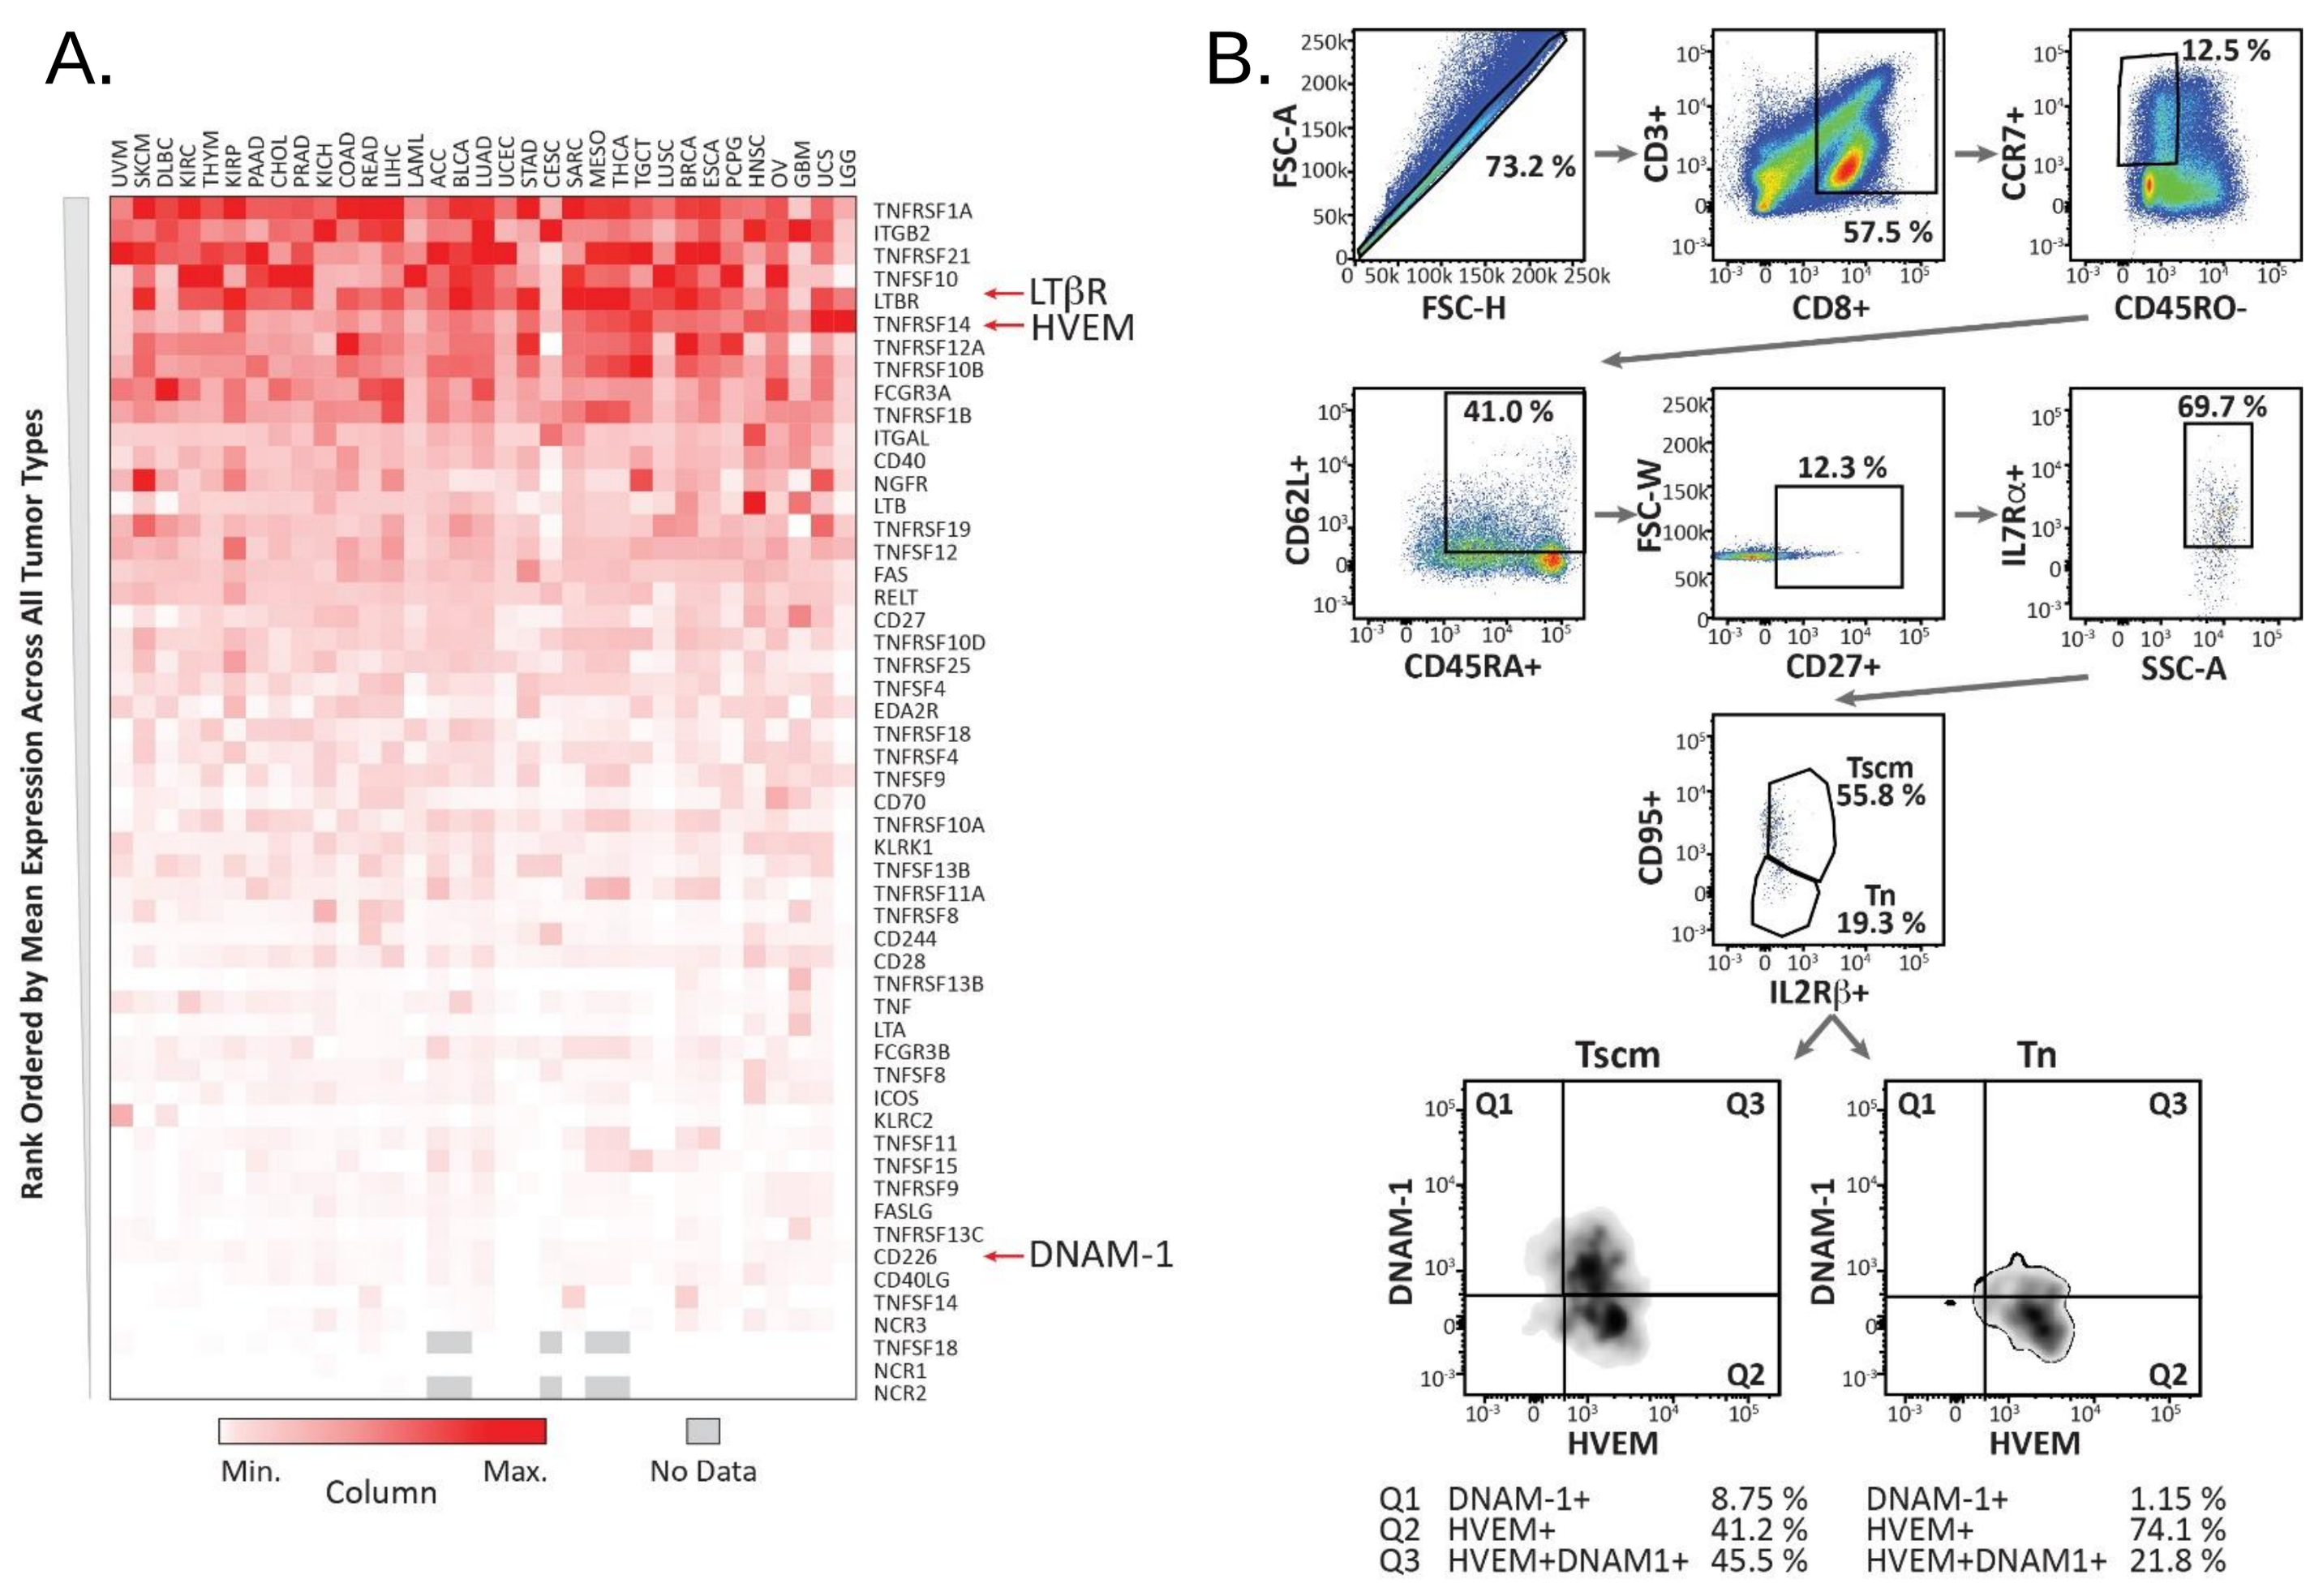
A.
LTβR
HVEM
DNAM-1
Rank Ordered by Mean Expression Across All Tumor Types
Min. Column Max.
No Data
B.
FSC-A
FSC-H
73.2 %
CD3+
CD8+
57.5 %
CCR7+
CD45RO-
12.5 %
CD62L+
CD45RA+
41.0 %
FSC-W
CD27+
12.3 %
IL7Rα+
SSC-A
69.7 %
CD95+
IL2Rβ+
Tscm 55.8 %
Tn 19.3 %
Tscm
Tn
DNAM-1
HVEM
Q1 DNAM-1+ 8.75 %
Q2 HVEM+ 41.2 %
Q3 HVEM+DNAM1+ 45.5 %
DNAM-1+ 1.15 %
HVEM+ 74.1 %
HVEM+DNAM1+ 21.8 %

Figure 1. Nomination of HVEM and LTβR as TNF-receptors highly expressed in tumors and on a range of immune cells. (A) The expression of 53 immune co-stimulatory genes was rank ordered across all TCGA cancers, identifying HVEM and LTBR in the top 10. (B) HVEM expression was characterized in relation to DNAM-1 expression on T stem cell memory (Tscm) cells induced for 9 days with CD3/CD28, IL-2, and a GSK3β inhibitor.

Generation of TIGIT-Fc-LIGHT

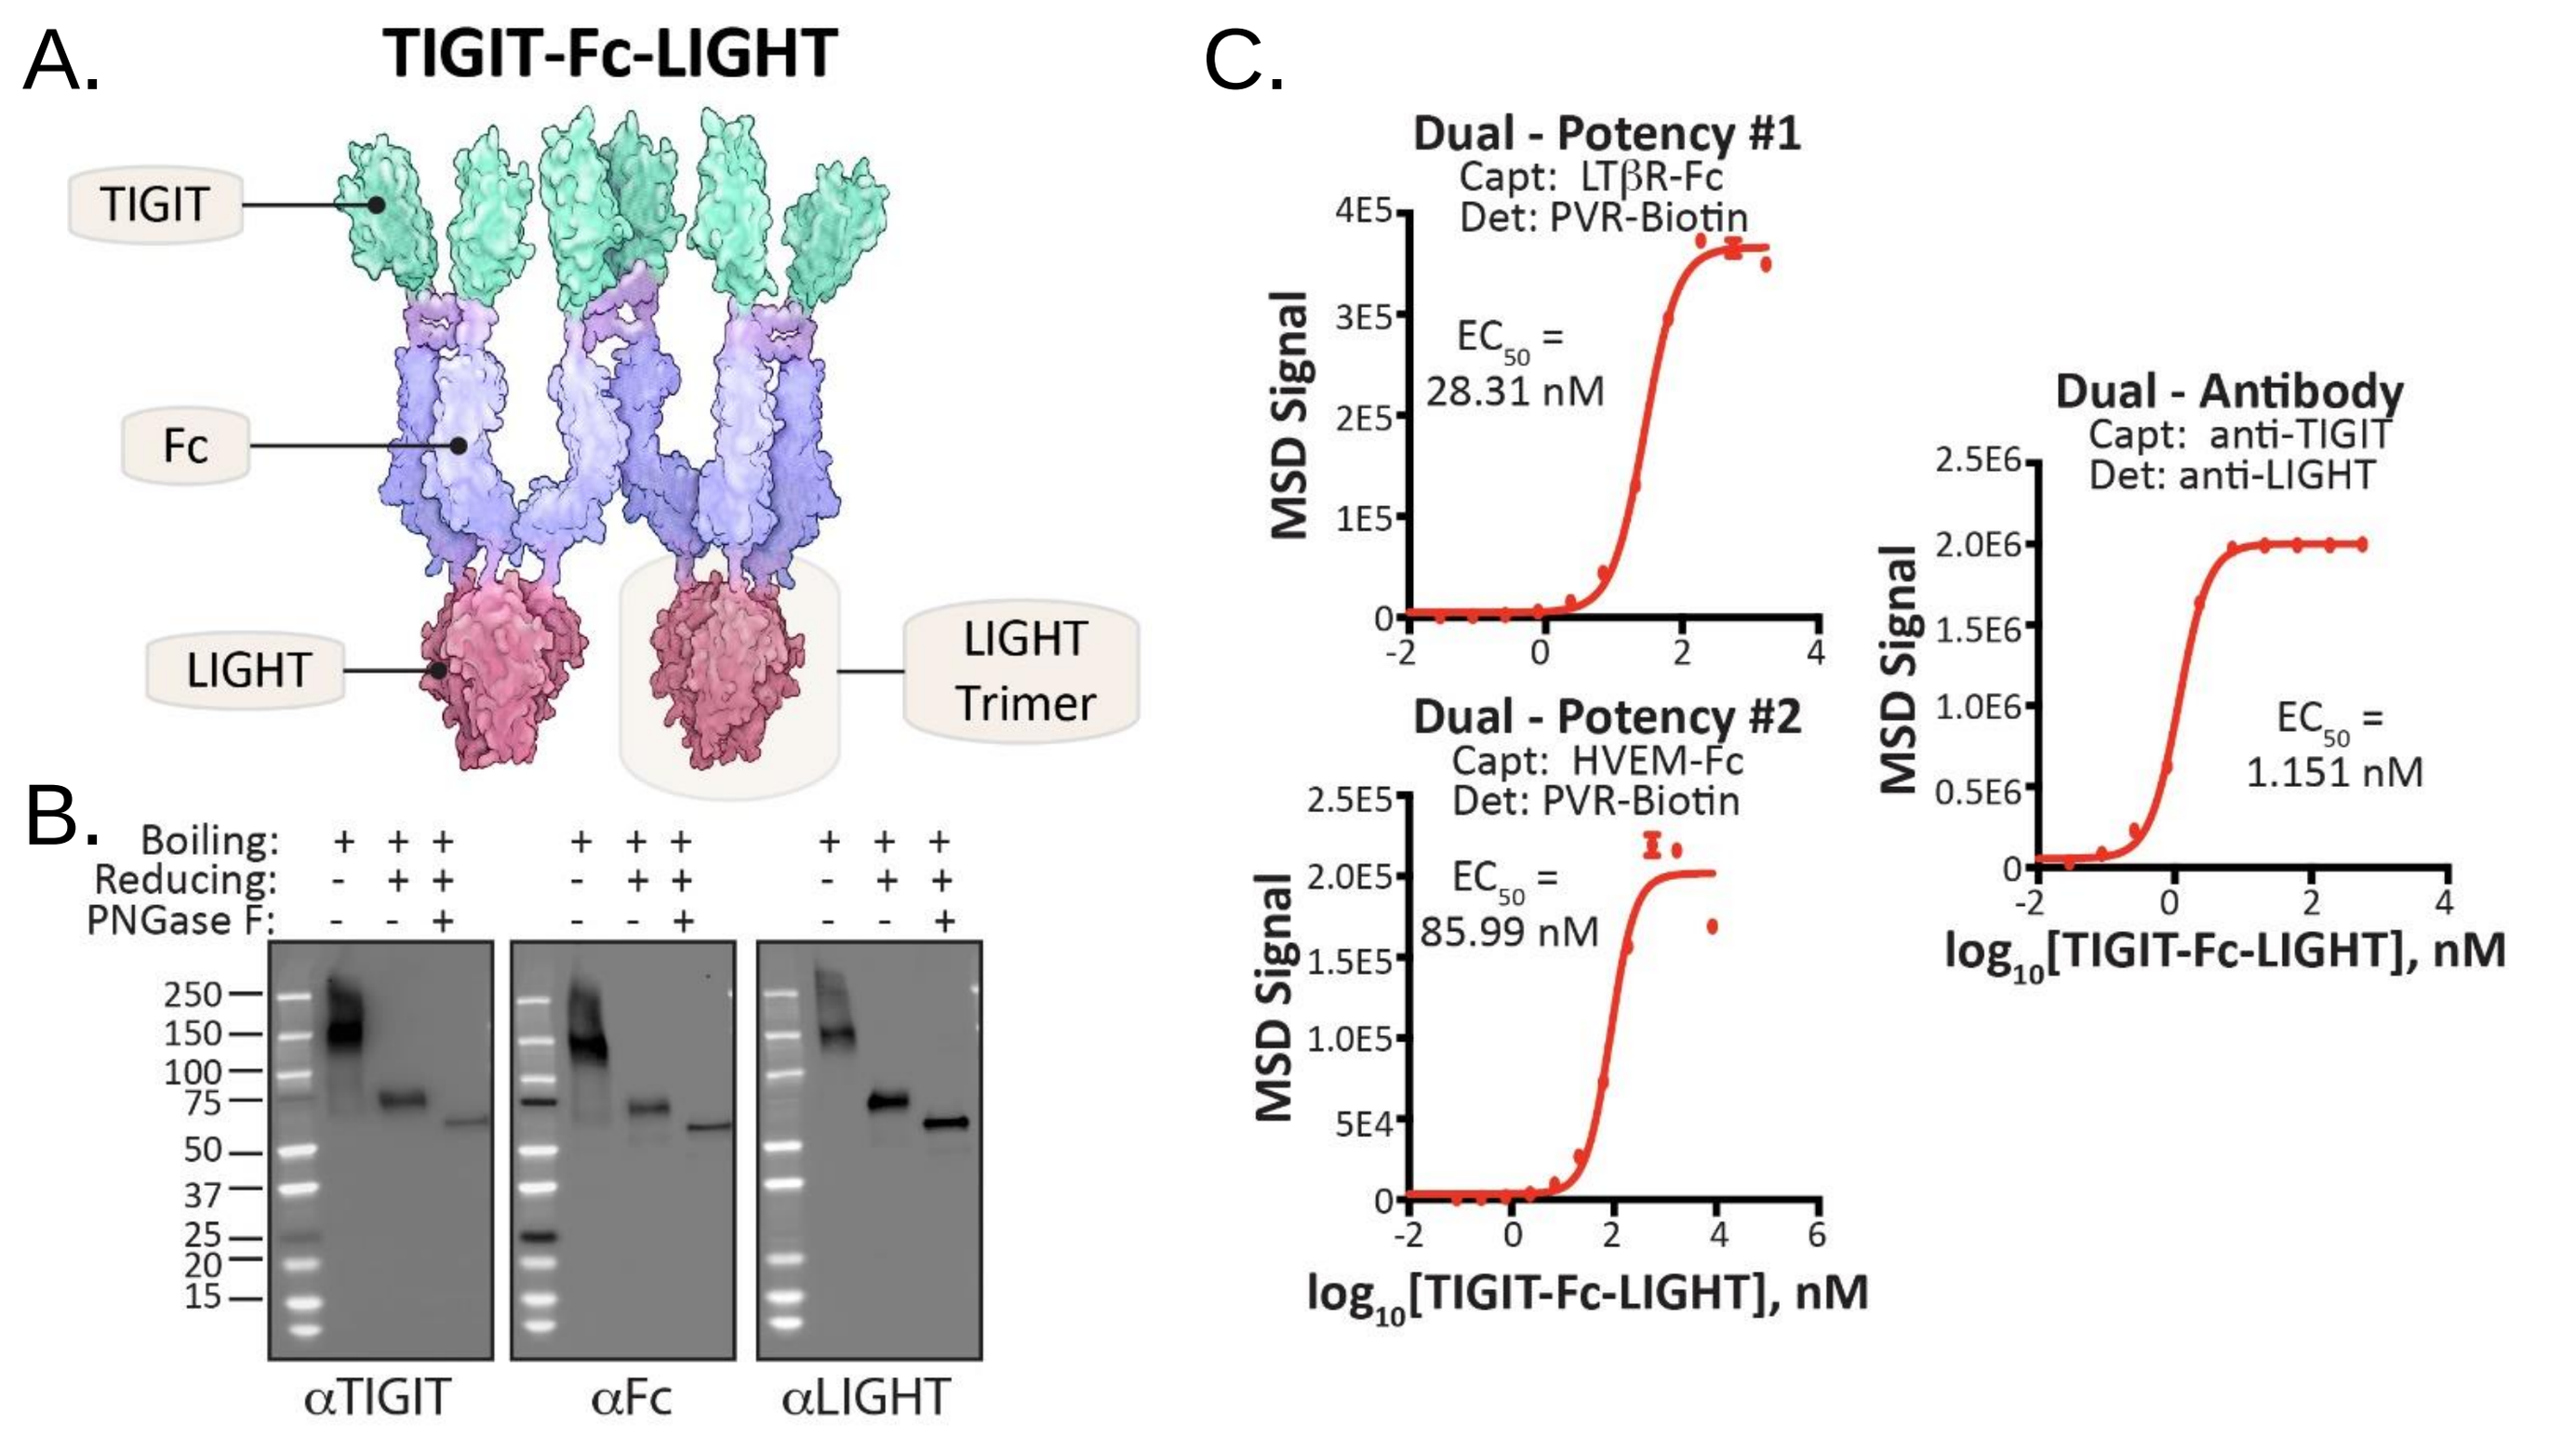
A.
TIGIT-Fc-LIGHT
TIGIT
Fc
LIGHT
LIGHT Trimer
B.
Boiling:
Reducing:
PNGase F:
αTIGIT
αFc
αLIGHT
C.
Dual - Potency #1
Capt: LTβR-Fc
Det: PVR-Biotin
EC50 = 28.31 nM
Dual - Potency #2
Capt: HVEM-Fc
Det: PVR-Biotin
EC50 = 85.99 nM
Dual - Antibody
Capt: anti-TIGIT
Det: anti-LIGHT
EC50 = 1.151 nM
MSD Signal
log10[TIGIT-Fc-LIGHT], nM

Figure 2. TIGIT-Fc-LIGHT characterization and receptor binding. (A) Hexameric TIGIT-Fc-LIGHT structure consisting of six TIGIT extracellular domains linked to two sets of active LIGHT trimers. (B) Each domain of TIGIT-Fc-LIGHT was detected using Western blot under native, reduced (BME), and reduced/deglycosylated conditions. (C) MSD demonstrated simultaneous binding of TIGIT-Fc-LIGHT to checkpoint targets (PVR) and immune co-stimulatory receptors found on myeloid, CD8+ T, and NK cells (LTβR and HVEM).

Broad Immune Activation

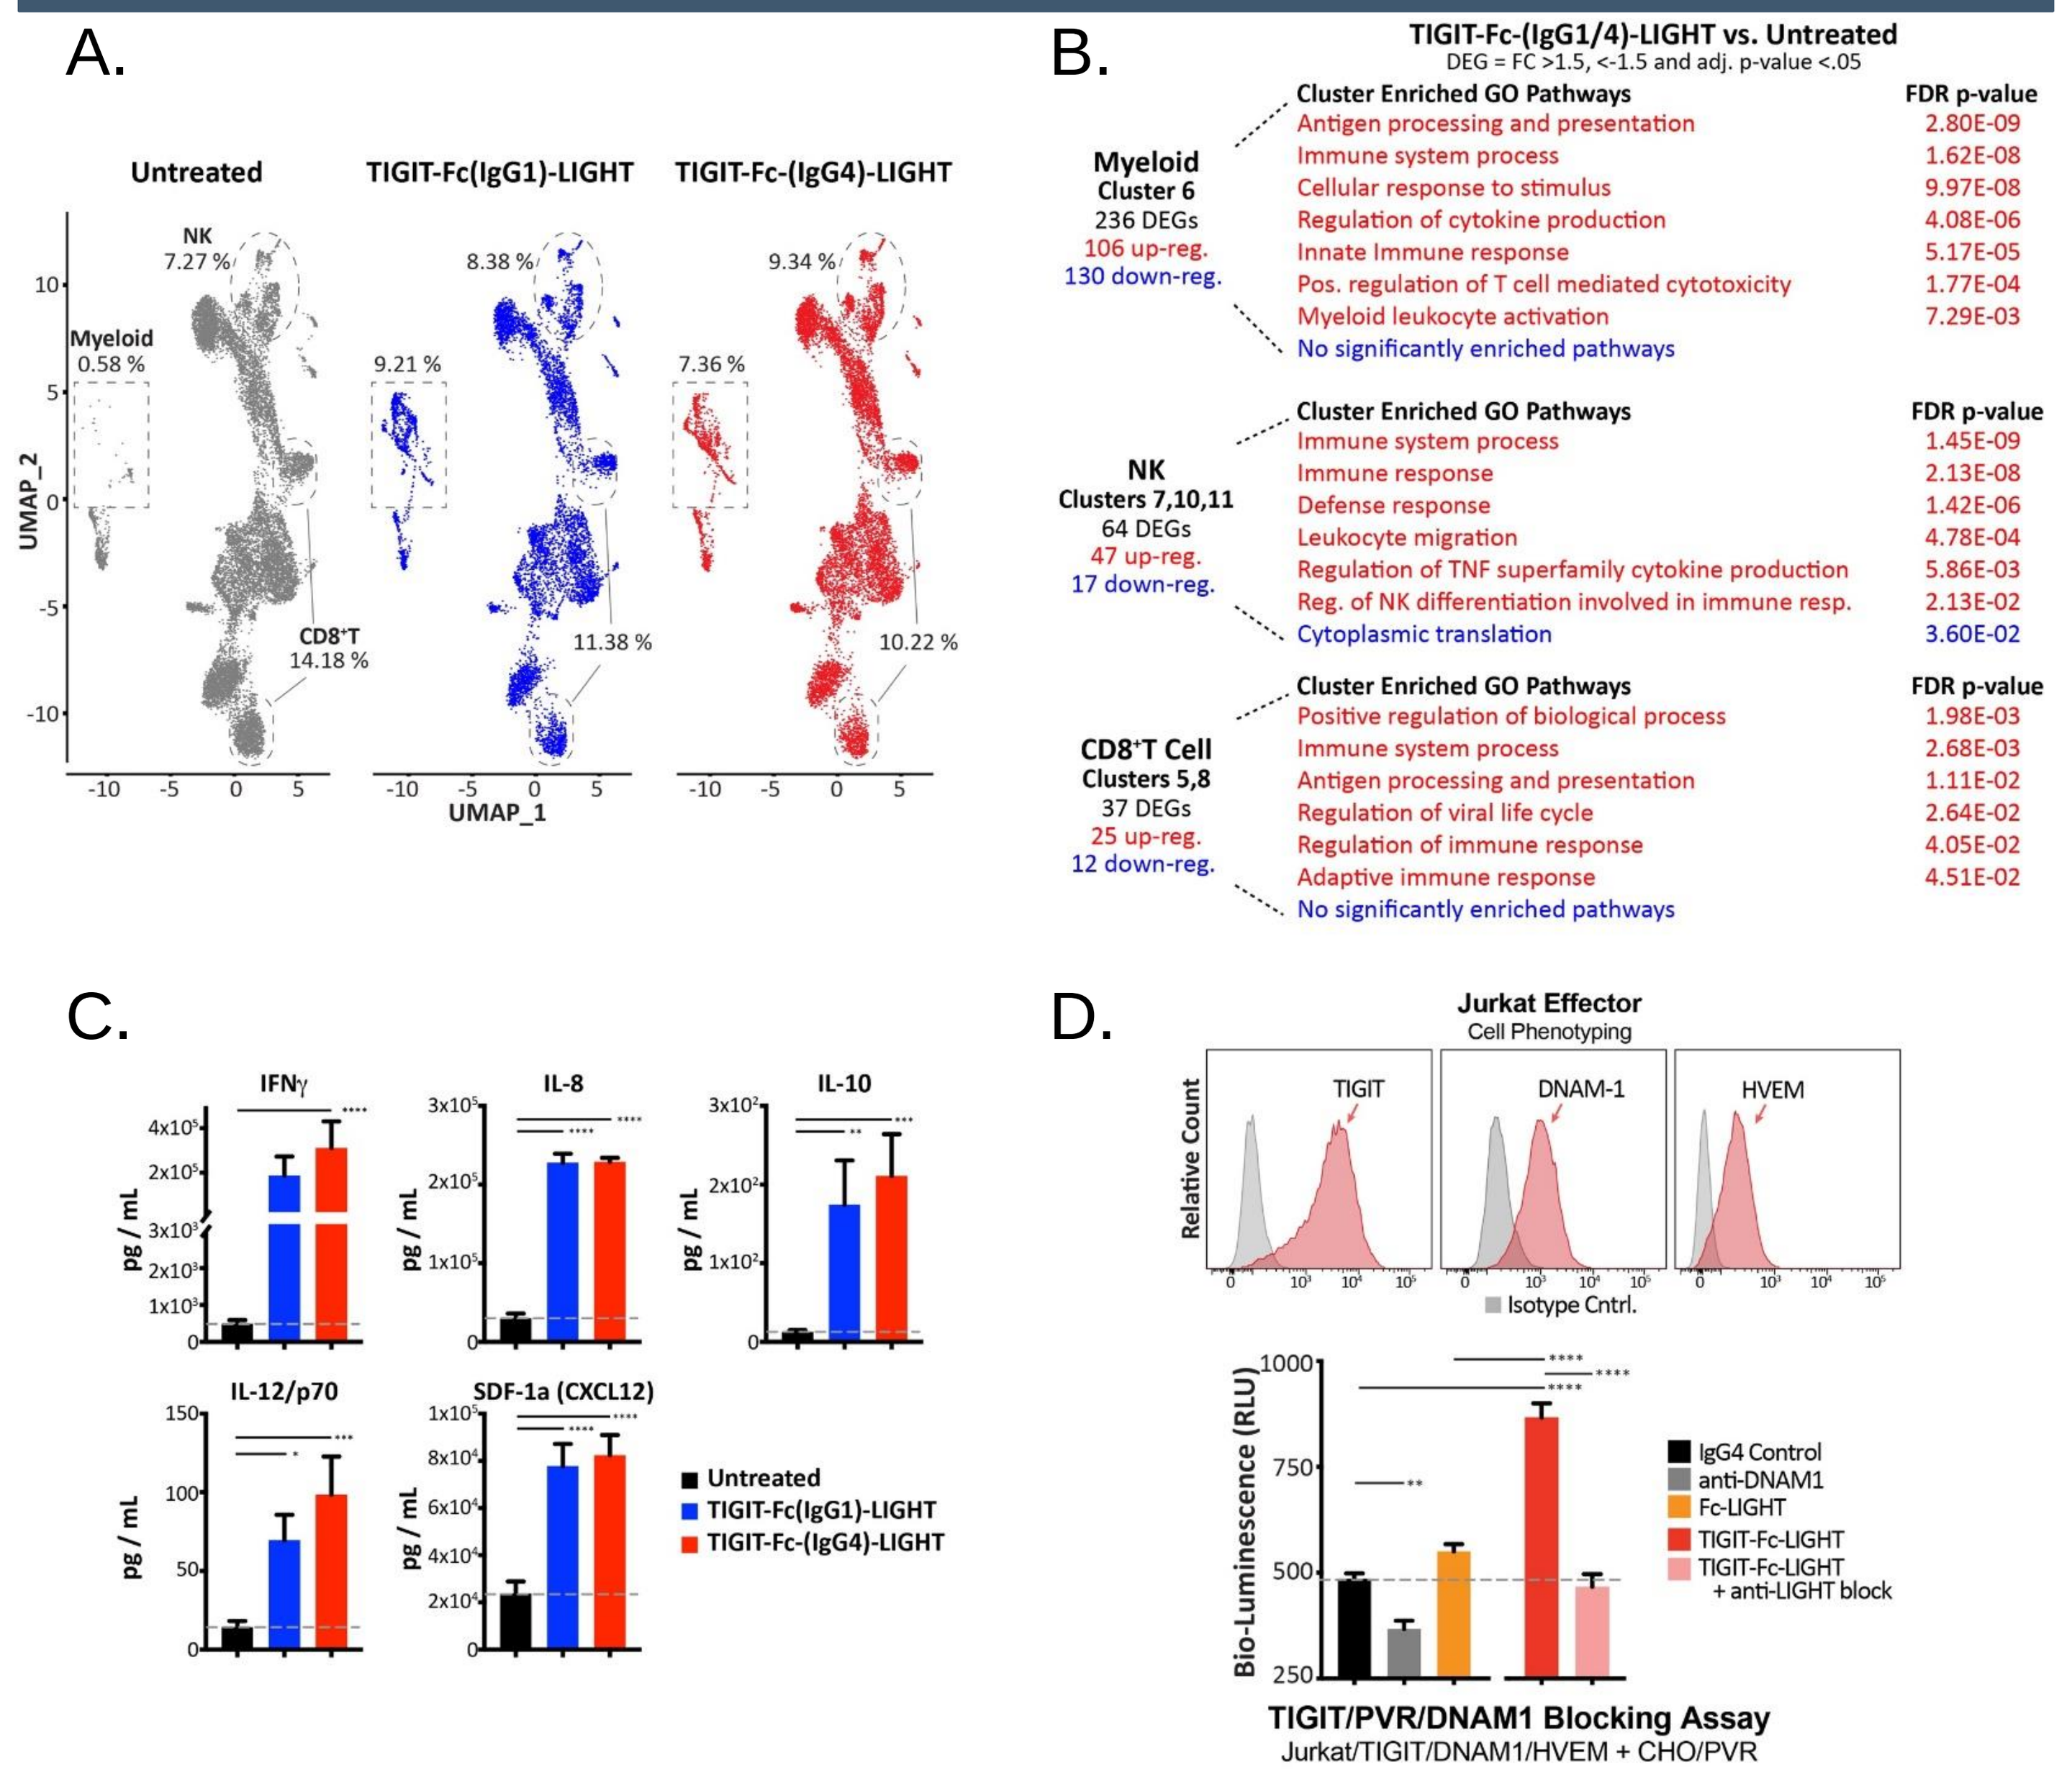
A.
Untreated
TIGIT-Fc(IgG1)-LIGHT
TIGIT-Fc-(IgG4)-LIGHT
NK
Myeloid
CD8+T
UMAP_1
UMAP_2
B.
TIGIT-Fc-(IgG1/4)-LIGHT vs. Untreated
DEG = FC >1.5, <-1.5 and adj. p-value <.05
Myeloid Cluster 6 236 DEGs 106 up-reg. 130 down-reg.
Cluster Enriched GO Pathways FDR p-value
Antigen processing and presentation 2.80E-09
Immune system process 1.62E-08
Cellular response to stimulus 9.97E-08
Regulation of cytokine production 4.08E-06
Innate Immune response 5.17E-05
Pos. regulation of T cell mediated cytotoxicity 1.77E-04
Myeloid leukocyte activation 7.29E-03
No significantly enriched pathways
NK Clusters 7,10,11 64 DEGs 47 up-reg. 17 down-reg.
Cluster Enriched GO Pathways FDR p-value
Immune system process 1.45E-09
Immune response 2.13E-08
Defense response 1.42E-06
Leukocyte migration 4.78E-04
Regulation of TNF superfamily cytokine production 5.86E-03
Reg. of NK differentiation involved in immune resp. 2.13E-02
Cytoplasmic translation 3.60E-02
CD8+T Cell Clusters 5,8 37 DEGs 25 up-reg. 12 down-reg.
Cluster Enriched GO Pathways FDR p-value
Positive regulation of biological process 1.98E-03
Immune system process 2.68E-03
Antigen processing and presentation 1.11E-02
Regulation of viral life cycle 2.64E-02
Regulation of immune response 4.05E-02
Adaptive immune response 4.51E-02
No significantly enriched pathways
C.
IFNγ
IL-8
IL-10
IL-12/p70
SDF-1a (CXCL12)
pg / mL
Untreated
TIGIT-Fc(IgG1)-LIGHT
TIGIT-Fc-(IgG4)-LIGHT
D.
Jurkat Effector
Cell Phenotyping
TIGIT
DNAM-1
HVEM
Relative Count
Isotype Cntrl.
Bio-Luminescence (RLU)
IgG4 Control
anti-DNAM1
Fc-LIGHT
TIGIT-Fc-LIGHT
TIGIT-Fc-LIGHT + anti-LIGHT block
TIGIT/PVR/DNAM1 Blocking Assay
Jurkat/TIGIT/DNAM1/HVEM + CHO/PVR

Figure 3. TIGIT-Fc-LIGHT stimulated myeloid, CD8+ T, and NK cells with both FcγR competent and inert Fc domains. (A) Single cell RNA-seq was performed on human PBMC cultured with IgG1 or IgG4 variants of TIGIT-Fc-LIGHT for 2 days in AIMV media. UMAP was used to visualize cell populations that corresponded to myeloid, CD8+ T, or NK cells (identified elsewhere using SingleR and ImmuneExp). (B) PANTHER was used to identify pathways associated with TIGIT-Fc-LIGHT induced DEGs. (C) After 2 days, cytokines were assessed in the culture supernatant using MSD. (D) Jurkat effector cells from a commercially available PVR:DNAM1 reporter assay were found to also express human HVEM. TIGIT-Fc-LIGHT bypassed the need for DNAM-1 co-stimulation and directly activated downstream signaling via HVEM.

TIGIT-Fc-LIGHT Anti-Tumor Activity

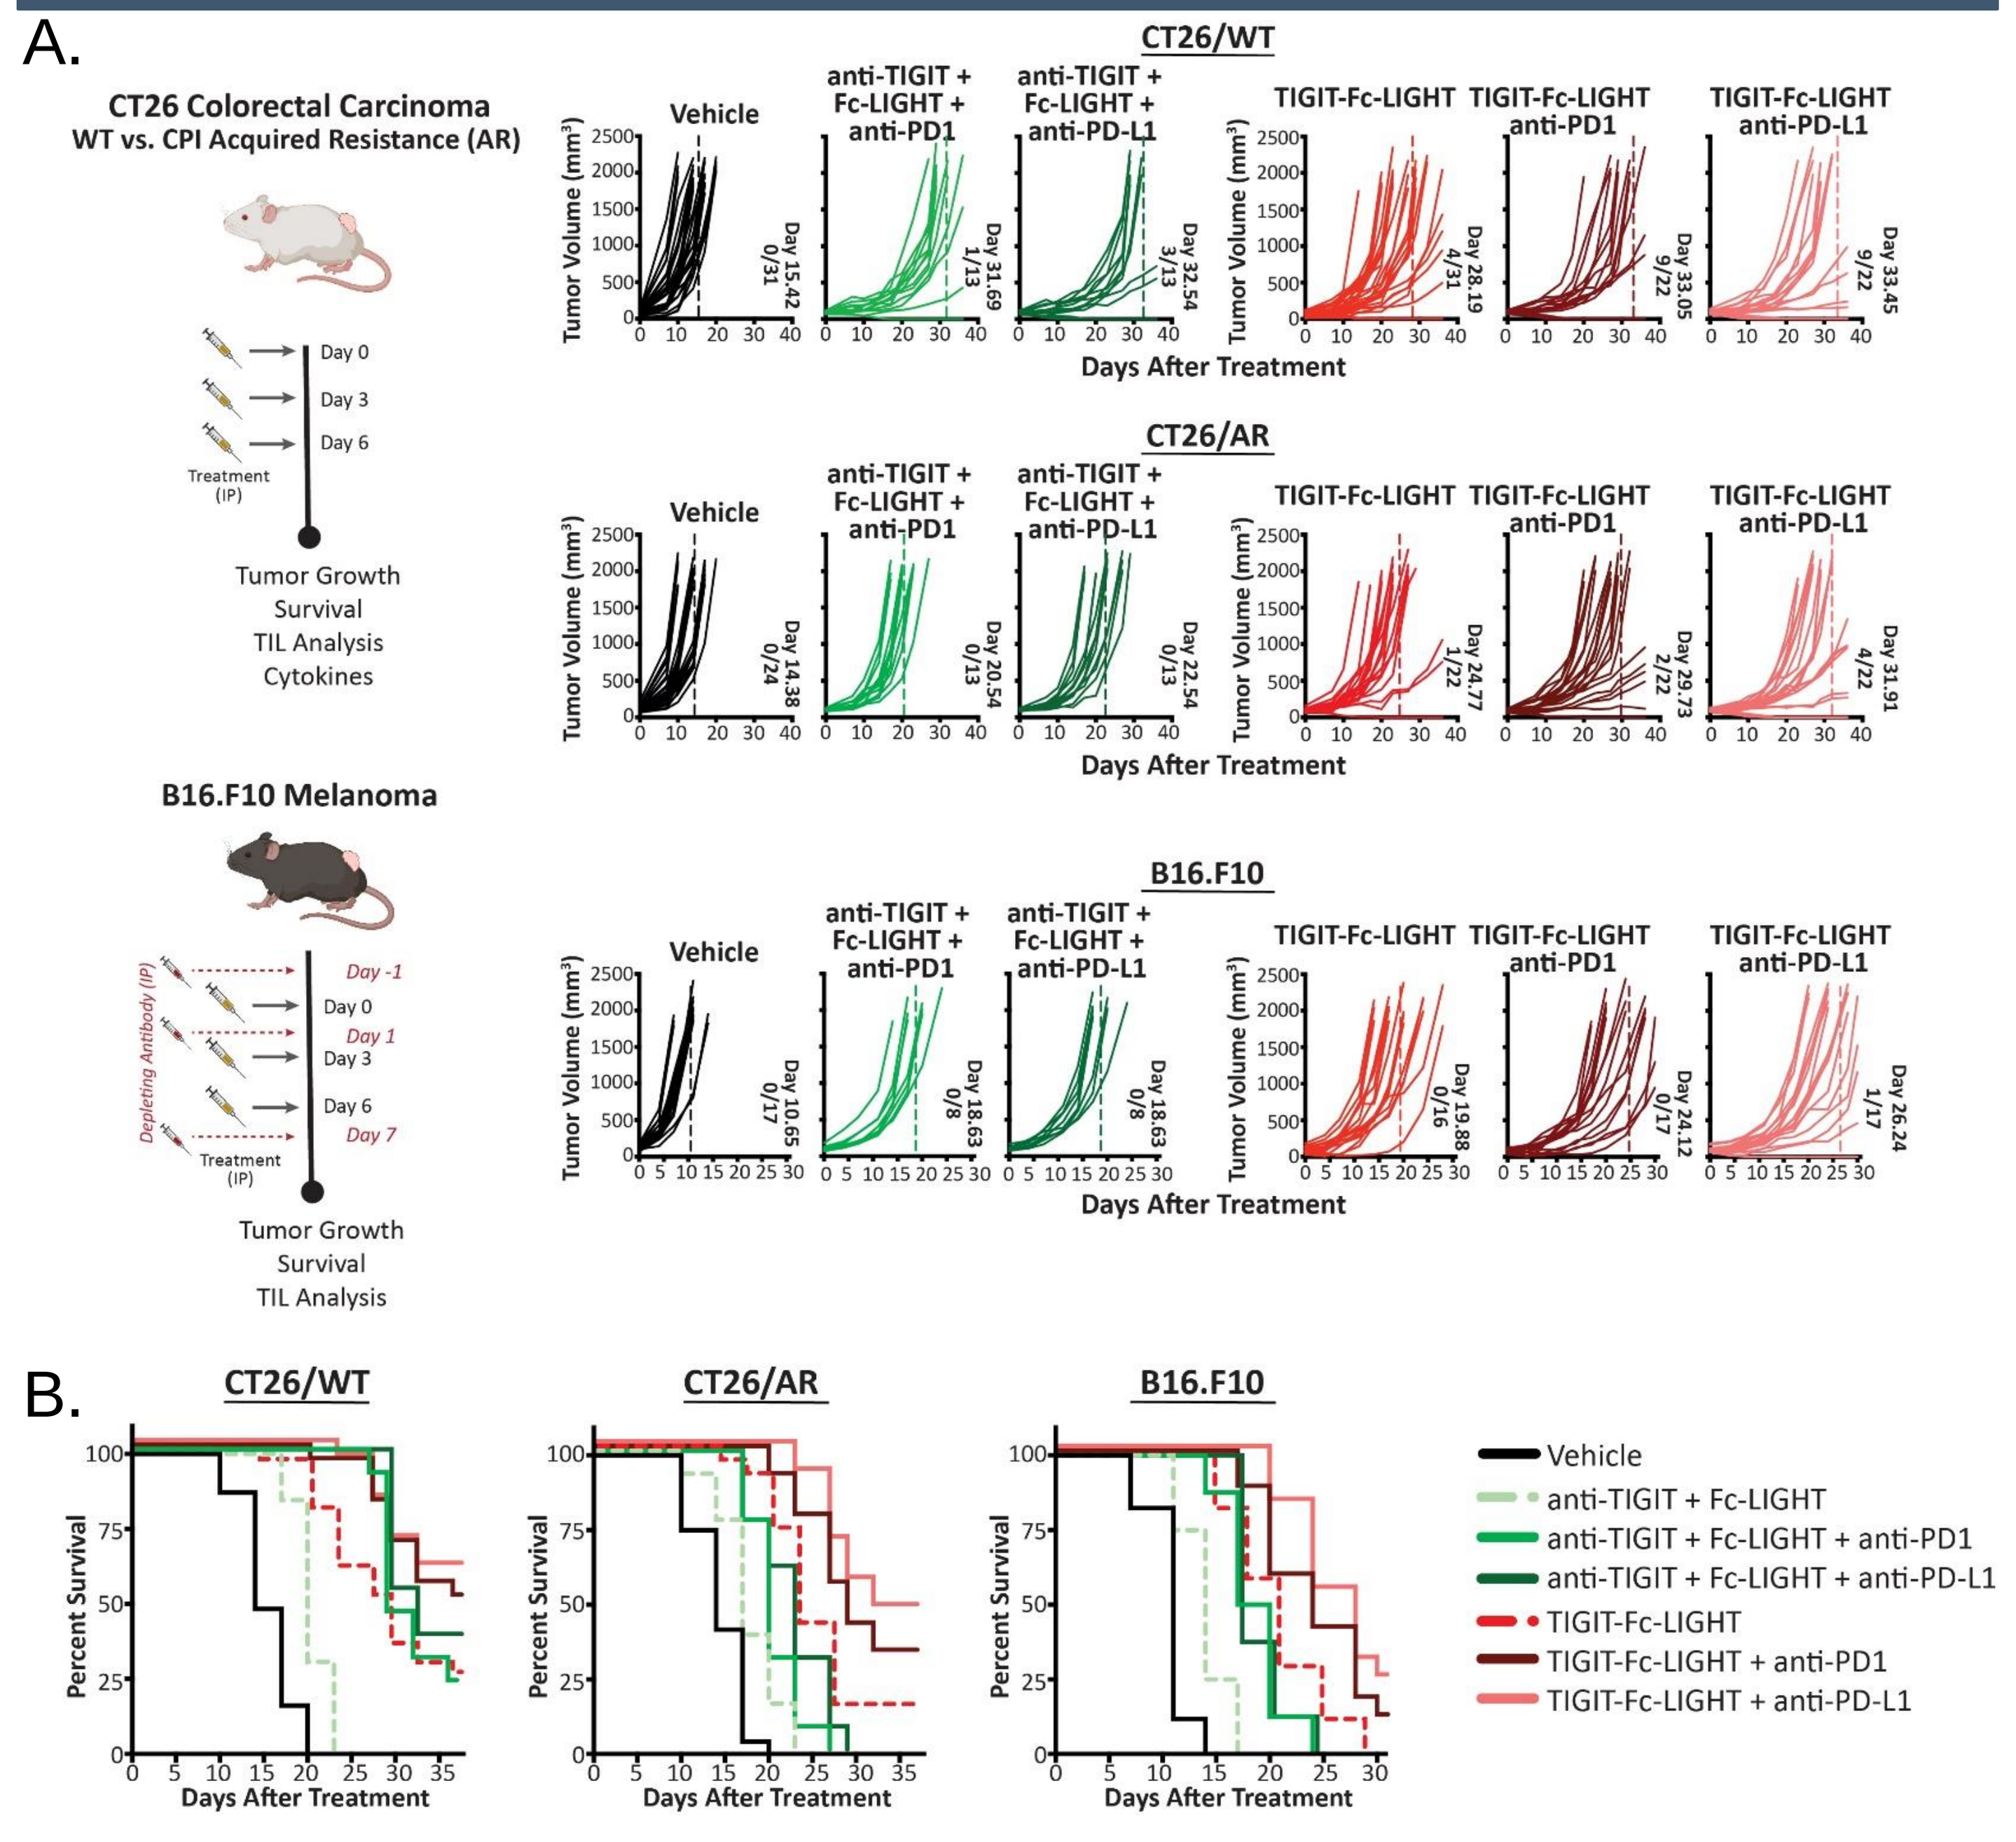
A.
CT26 Colorectal Carcinoma
WT vs. CPI Acquired Resistance (AR)
Day 0
Day 3
Day 6
Treatment (IP)
Tumor Growth
Survival
TIL Analysis
Cytokines
B16.F10 Melanoma
Depleting Antibody (IP)
Day -1
Day 1
Day 7
Tumor Growth
Survival
TIL Analysis
CT26/WT
CT26/AR
B16.F10
Vehicle
anti-TIGIT + Fc-LIGHT + anti-PD1
anti-TIGIT + Fc-LIGHT + anti-PD-L1
TIGIT-Fc-LIGHT
TIGIT-Fc-LIGHT anti-PD1
TIGIT-Fc-LIGHT anti-PD-L1
Tumor Volume (mm³)
Days After Treatment
B.
Percent Survival
Days After Treatment
Vehicle
anti-TIGIT + Fc-LIGHT
anti-TIGIT + Fc-LIGHT + anti-PD1
anti-TIGIT + Fc-LIGHT + anti-PD-L1
TIGIT-Fc-LIGHT
TIGIT-Fc-LIGHT + anti-PD1
TIGIT-Fc-LIGHT + anti-PD-L1

Figure 4. Anti-tumor activity of the murine TIGIT-Fc-LIGHT surrogate. (A) BALB/c mice were implanted with wild-type (WT) CT26 tumors, or tumors that were conditioned *in vivo* to develop anti-PD1 acquired resistance (CT26/AR), and C57BL/6 mice were implanted with B16.F10 melanoma tumors. When tumors established, mice were treated with vehicle (PBS), anti-TIGIT (1G9), Fc-LIGHT, anti-PD1 (RMP1-14), anti-PD-L1 (10F.9G2), or mTIGIT-Fc-LIGHT (controls at 100 μg and TIGIT-Fc-LIGHT at 200 μg per dose). Shown are the individual animal tumor growth curves, the average day in which each group reached tumor burden, and the number of mice that completely rejected the tumor in response to treatment. (B) Kaplan-Meir survival curves for various treatment groups in the CT26/WT, CT26/AR, and B16.F10 preclinical tumor models.

Enhanced Lymphocyte Infiltration into Tumors

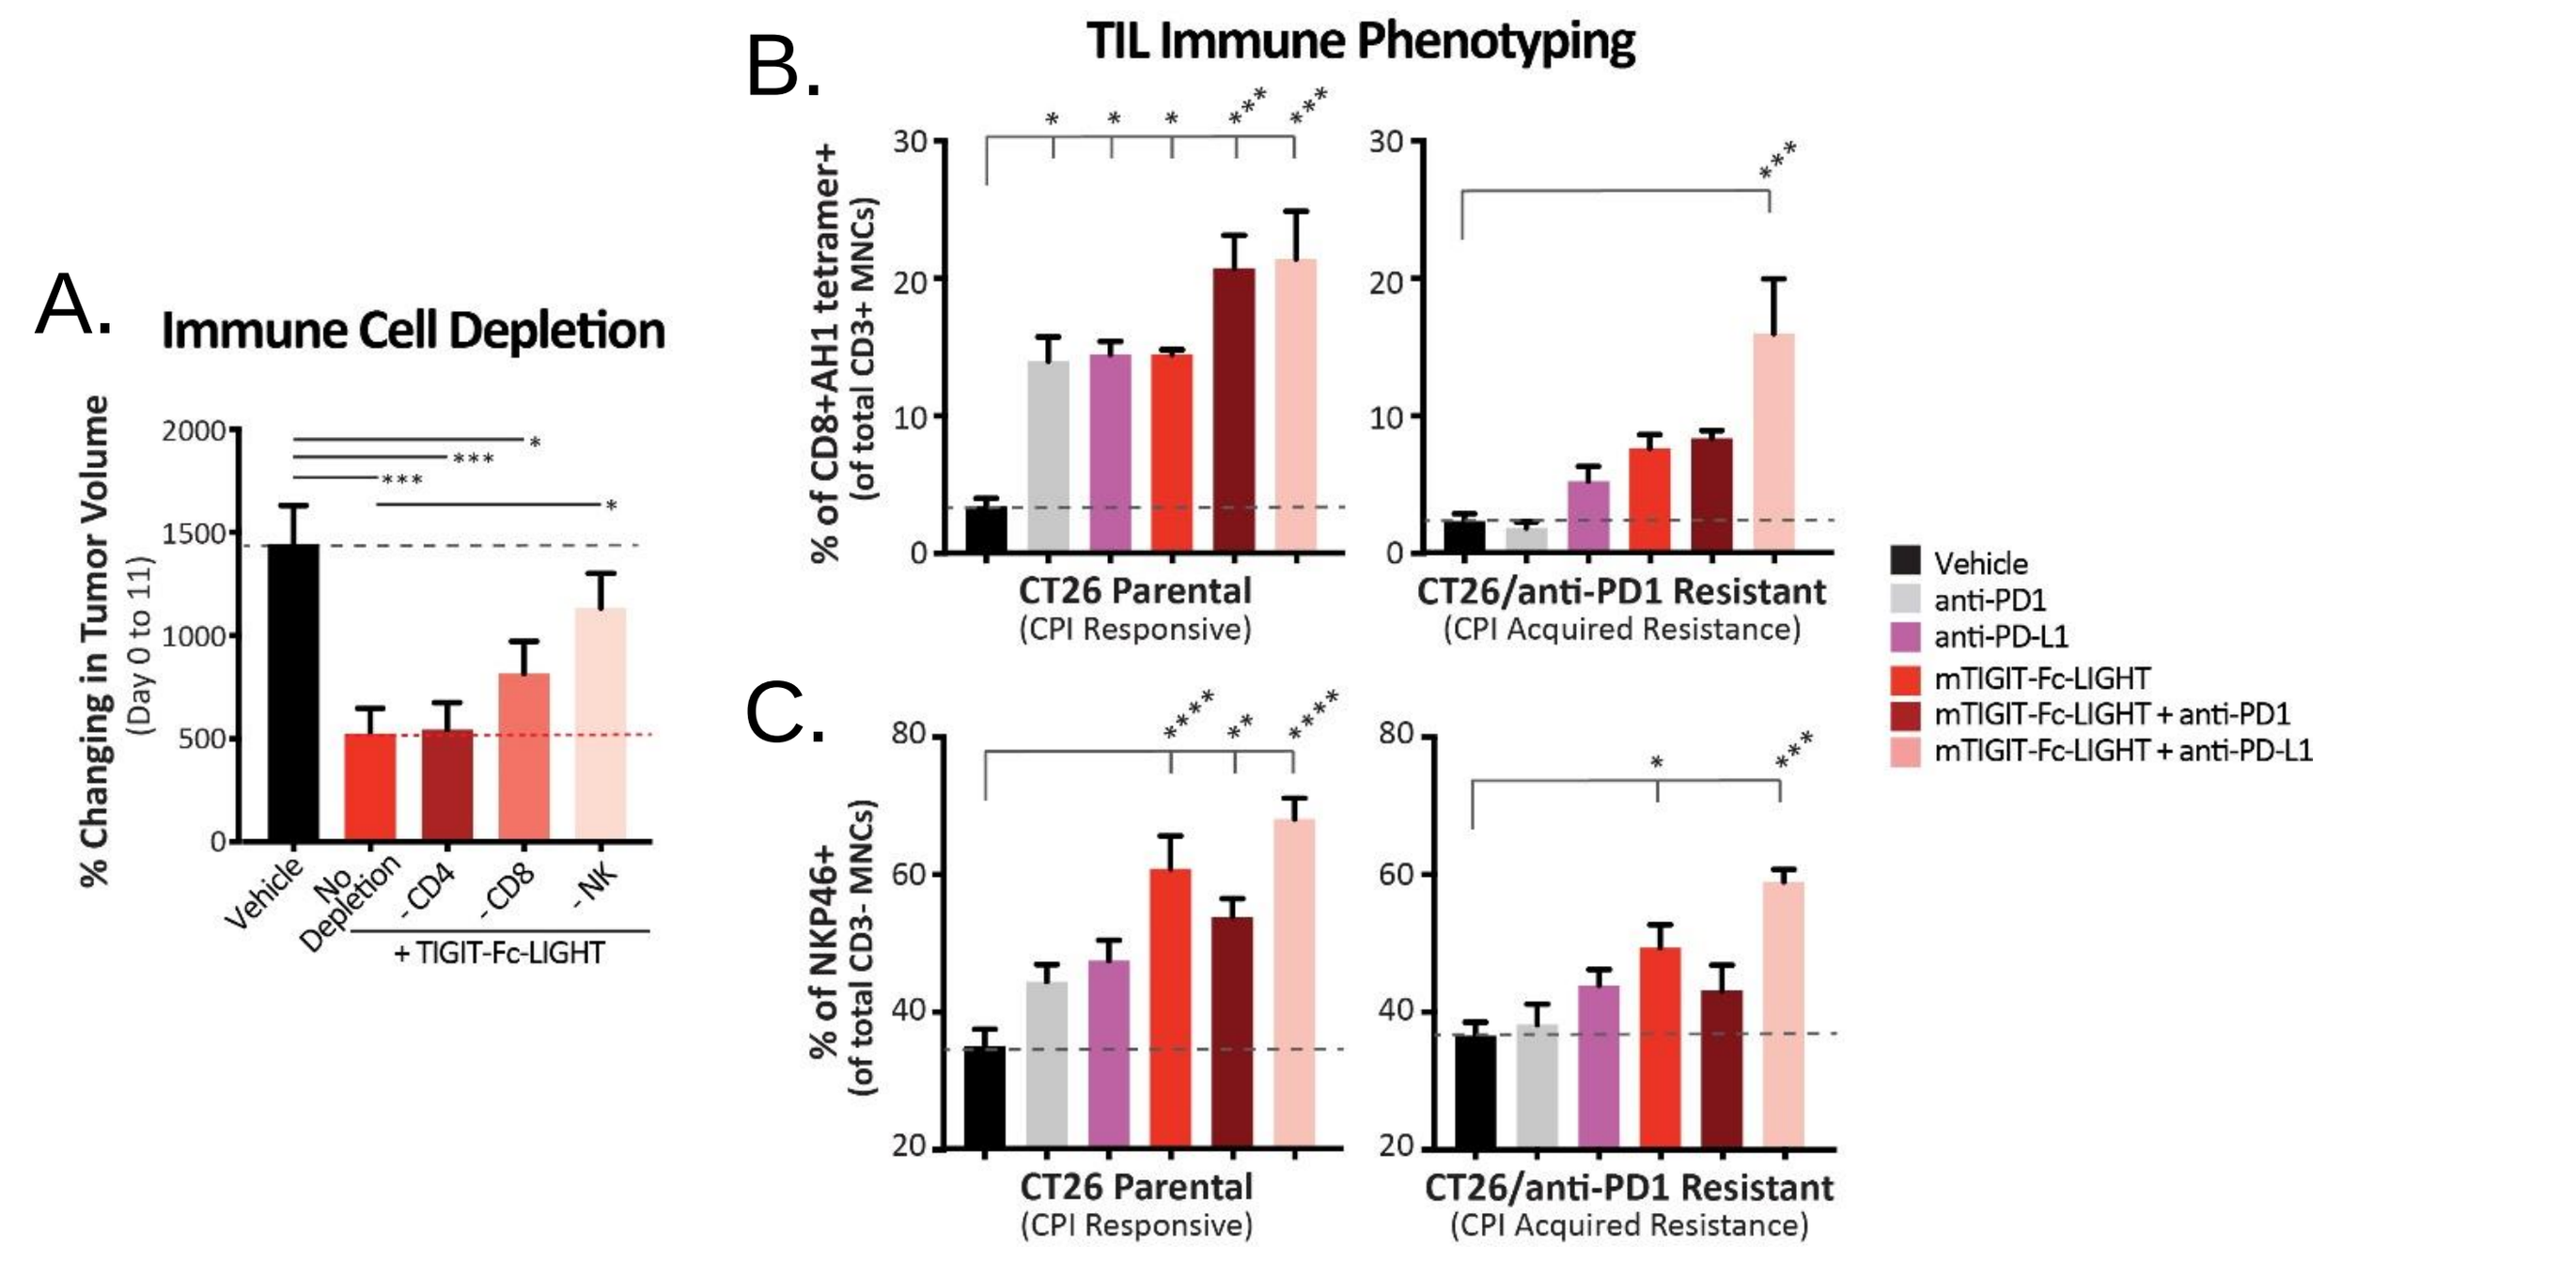
A.
Immune Cell Depletion
% Changing in Tumor Volume (Day 0 to 11)
Vehicle
No Depletion
-CD4
-CD8
-NK
+ TIGIT-Fc-LIGHT
B.
TIL Immune Phenotyping
% of CD8+AH1 tetramer+ (of total CD3+ MNCs)
CT26 Parental (CPI Responsive)
CT26/anti-PD1 Resistant (CPI Acquired Resistance)
C.
% of NKP46+ (of total CD3- MNCs)
Vehicle
anti-PD1
anti-PD-L1
mTIGIT-Fc-LIGHT
mTIGIT-Fc-LIGHT + anti-PD1
mTIGIT-Fc-LIGHT + anti-PD-L1

Figure 5. TIGIT-Fc-LIGHT *in vivo* activity. (A) The impact of CD4+ T, CD8+ T, or NK cells on TIGIT-Fc-LIGHT was evaluated in the B16.F10 model, through immune cell depletion studies. (B) Tumors were isolated from a cohort of treated animals 9 days after the initial treatment. Tumors were dissociated and assessed by flow cytometry for populations of antigen-specific CD8+ T cells (AH1 tetramer+) and (C) NKP46+ NK cells.

Translation of PD Activity to NHP

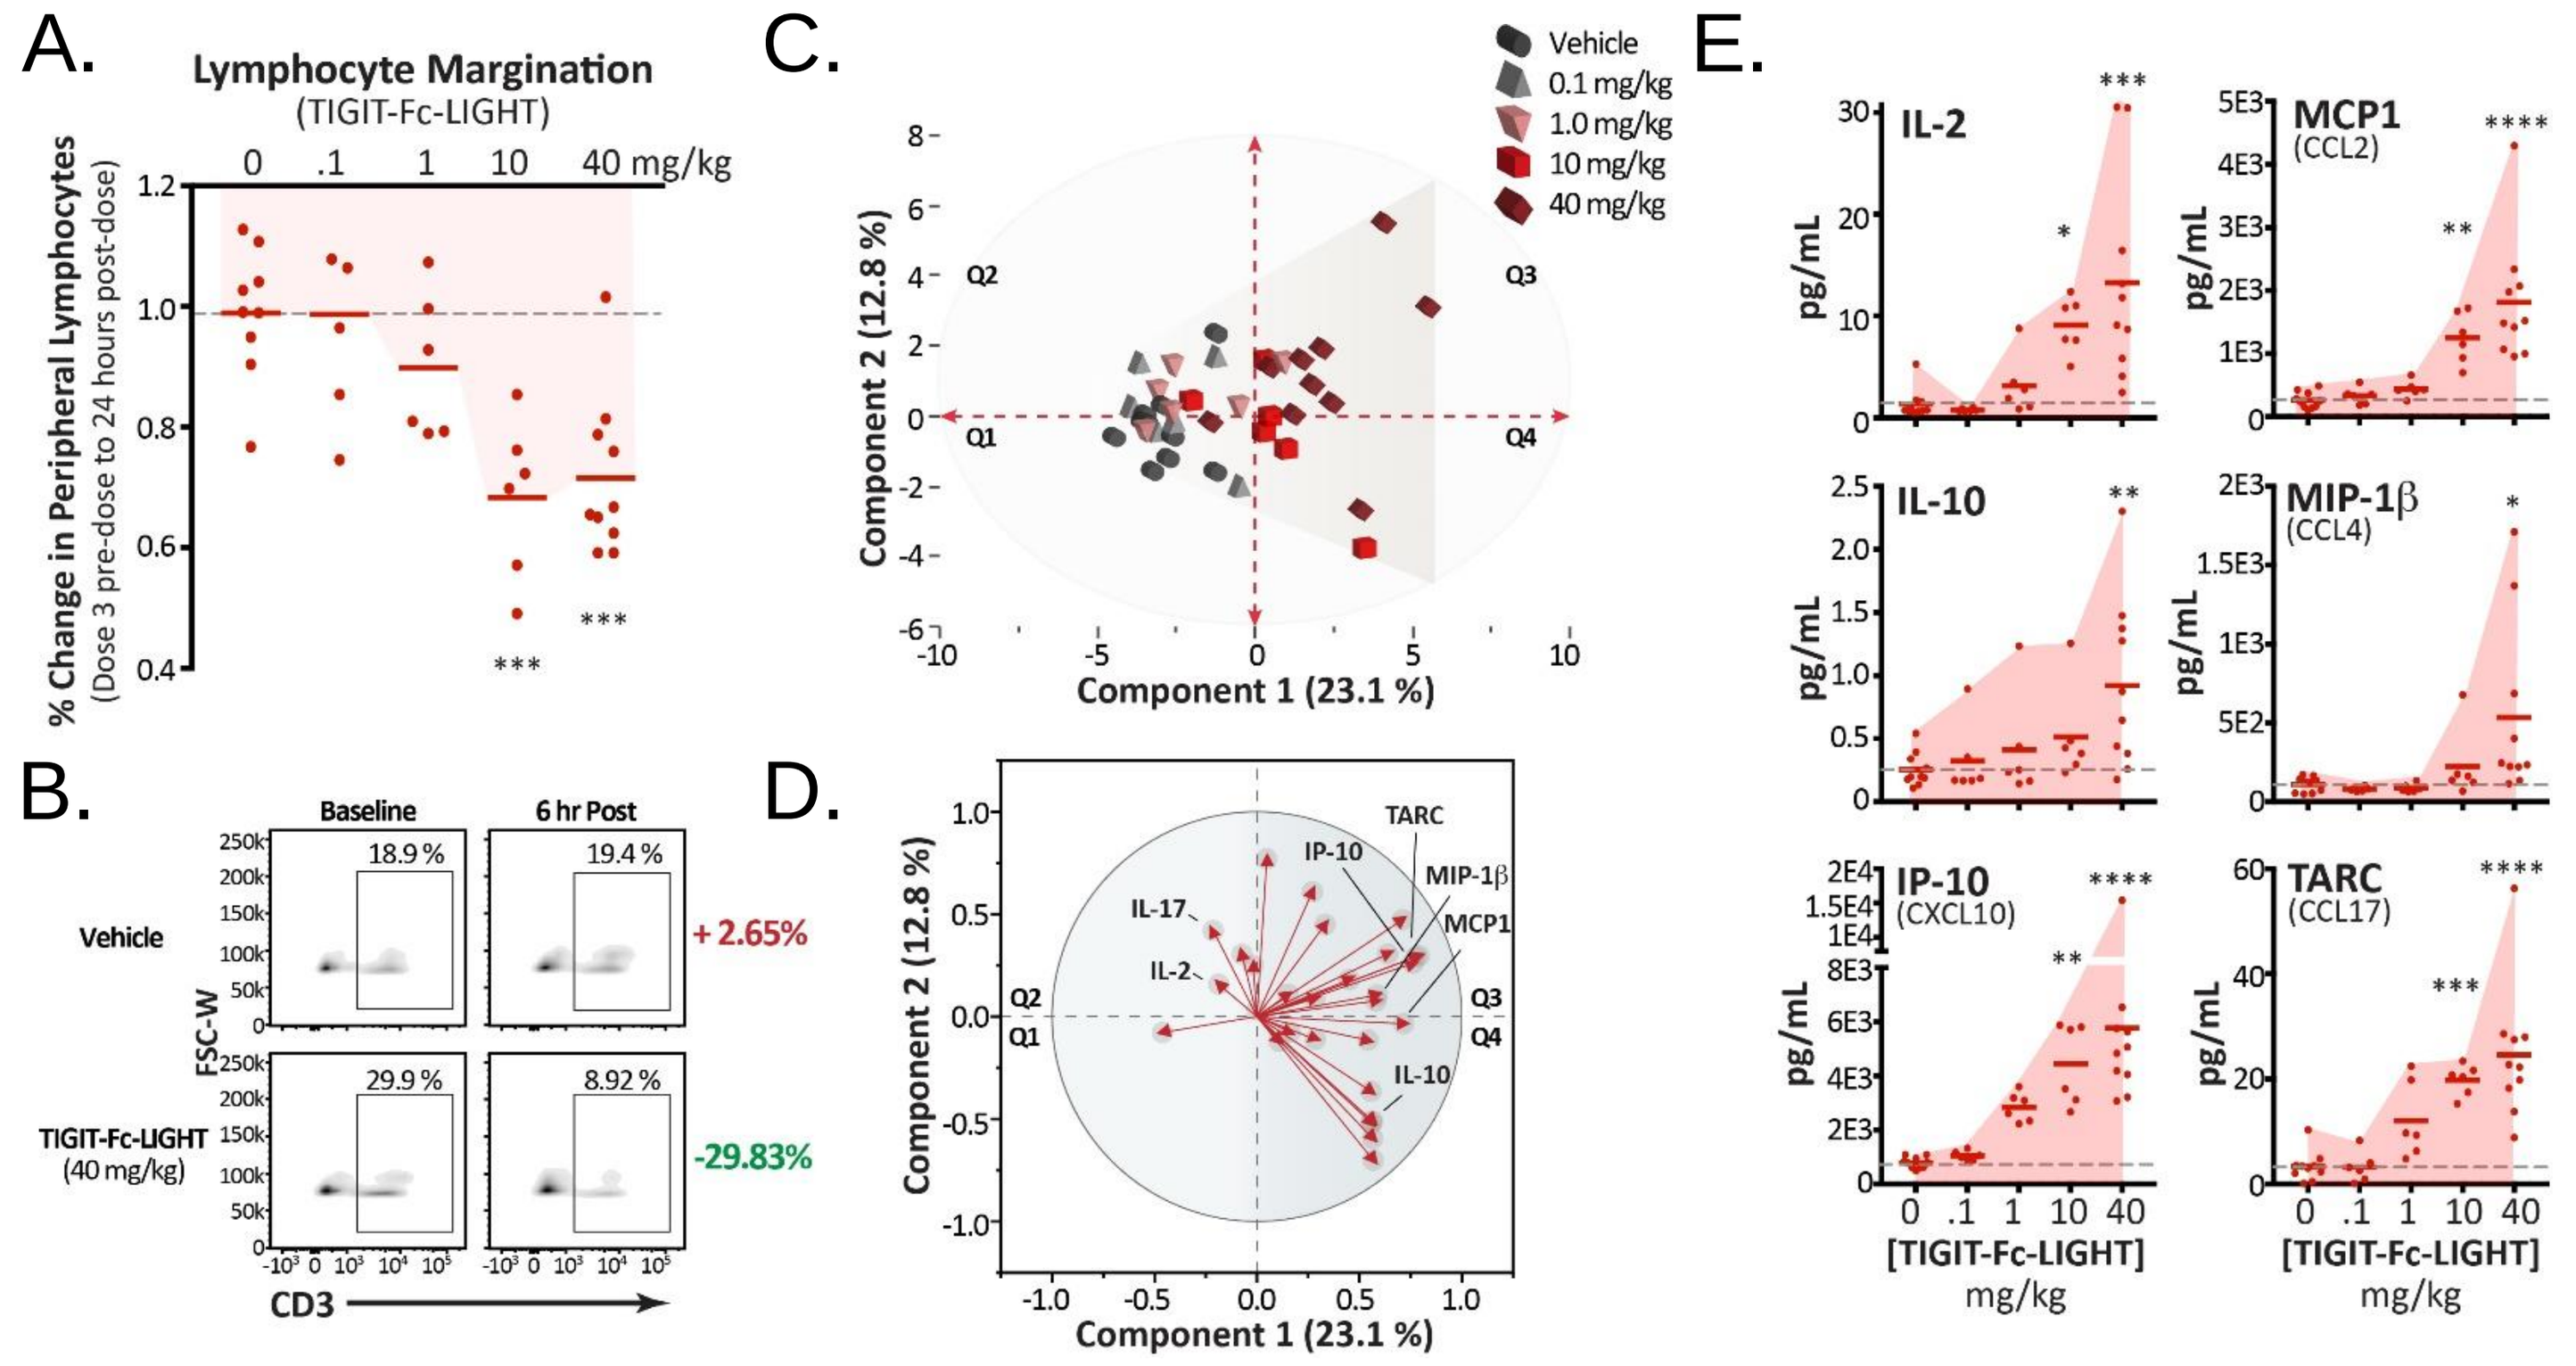
A.
Lymphocyte Margination
(TIGIT-Fc-LIGHT)
0 .1 1 10 40 mg/kg
% Change in Peripheral Lymphocytes
(Dose 3 pre-dose to 24 hours post-dose)
B.
Baseline
6 hr Post
Vehicle
TIGIT-Fc-LIGHT (40 mg/kg)
+2.65%
-29.83%
FSC-W
CD3
C.
Vehicle
0.1 mg/kg
1.0 mg/kg
10 mg/kg
40 mg/kg
Component 1 (23.1 %)
Component 2 (12.8 %)
D.
TARC
IP-10
MIP-1β
MCP1
IL-17
IL-2
IL-10
E.
IL-2
MCP1 (CCL2)
IL-10
MIP-1β (CCL4)
IP-10 (CXCL10)
TARC (CCL17)
pg/mL
[TIGIT-Fc-LIGHT] mg/kg

Figure 6. TIGIT-Fc-LIGHT pharmacodynamic (PD) activity in cynomolgus macaque. Animals were given 4 weekly IV infusions (days 1, 8, 15, 22) of vehicle or 0.1, 1.0, 10, or 40 mg/kg of TIGIT-Fc-LIGHT. (A) Complete blood counts revealed a dose dependent decrease or margination of lymphocytes, that included (B) HVEM+ T cells. (C) PCA distribution of animals based on the 2-hour post-dose cytokine signature, indicated a dose trend where (D) pro-inflammatory cytokines dominated the clustering of samples in quadrants Q3 and Q4, and adaptive immune cytokines (e.g. IL-2 and IL-17) dominated Q2. (E) The maximum cytokine response throughout the study shown at various cytokines was assessed using MSD.

Conclusions

TIGIT-Fc-LIGHT was designed to overcome the limitations of TIGIT blocking antibodies through:

- Preserved co-stimulation in advanced tumors (through HVEM)
- Direct myeloid cell activation (via LTβR)
- Co-stimulation is not dependent on PD-1 blockade (as it is for DNAM-1)
- Blockade of all known TIGIT ligands (PVR, PVRL2, PVRL3, and Nectin-4)
- No risk of depleting effector lymphocytes (no dependence on Fc composition)
- Translation of on-target pharmacodynamic activity into NHP

Pre-clinical data indicate that these goals were achieved, and further development is warranted.