Phase 1 dose escalation study of SL-172154 (SIRPα-Fc-CD40L) in platinum-resistant ovarian cancer

Authors: Nehal J. Lakhani, MD, PhD[1]; Daphne Stewart, MD[2]; Debra L. Richardson, MD[3]; Lauren E. Dockery, MD MS[3]; Linda Van Le, MD[4]; Justin Call, MD[5]; Fatima Rangwala, MD, PhD[6]; Anya Scholl[6]; Guanfang Wang, PhD[6]; Bo Ma, PhD[6]; Simon Metenou, PhD[6]; Lini Pandite, MD[6]; Erika Hamilton, MD[7]

[1]START Midwest, Grand Rapids, MI, United States; [2]USC Keck School of Medicine, Norris Comprehensive Cancer Center, Los Angeles, CA, [3]Stephenson Cancer Center and Sarah Cannon Research Institute/ University of Oklahoma, Oklahoma City, OK, United States; [4]University of North Carolina at Chapel Hill, Chapel Hill, NC, United States; [5]START Mountain Region, West Valley City, UT, United States; [6]Shattuck Labs, Durham, NC, United States; [7]Sarah Cannon Research Institute/Tennessee Oncology, Nashville, TN, United States

Trial Registration: NCT04406623

Abstract Number: 5544

Please contact Dr. Nehal Lakhani (Nehal.Lakhani@startmidwest.com) with any questions or comments on this poster.

Background

SIRPα-Fc-CD40L (SL-172154) is a hexameric, bi-functional fusion protein consisting of SIRPα linked to CD40L via an inert Fc linker.

SIRPα-Fc-CD40L protein competitively inhibits CD47 to enhance tumor cell phagocytosis and activates CD40 to increase antigen processing and cross-presentation by antigen presenting cells (APCs) to CD8 T cells, thus bridging innate and adaptive immunity.

Of all solid tumors, ovarian cancers have the highest expression of CD47 (TCGA data; [Wang, 2015; Brightwell, 2016]). Tumor-associated macrophages (TAMs) are abundant within the ovarian tumor microenvironment ([TME] Gupta, 2018) and are the target cells for SL-172154 therapy.

In this Phase 1 dose escalation study, SL-172154 was evaluated in patients with platinum-resistant ovarian, fallopian tube and primary peritoneal cancers (ovarian cancer).

SL-172154 relies upon combination regimens to provide prophagocytic signals to drive efficacy (Figure 1).

SL-172154: Novel CD47 Inhibitor + CD40 Agonist

Rationally Designed to Maximize the Benefits of CD47 Blockade

1	High Affinity and Avidity CD47 Binding	Inhibition of CD47/SIRP alpha interaction **potentiates phagocytosis when prophagocytic signals ("eat me") predominate over antiphagocytic signals on tumor cells.**
2	Inert Fc Domain	Designed to reduce **Fc-mediated effector function and the risk of associated cytopenias.**
3	Activation of CD40 Pathway	Enhances antigen cross presentation, **leads to T cell activation, bridging innate and adaptive immunity.**
4	Combination Opportunities	**Enhances** the **efficacy** of tumor targeted antibodies (mAbs), antibody drug conjugates (ADC) and chemotherapeutic agents.

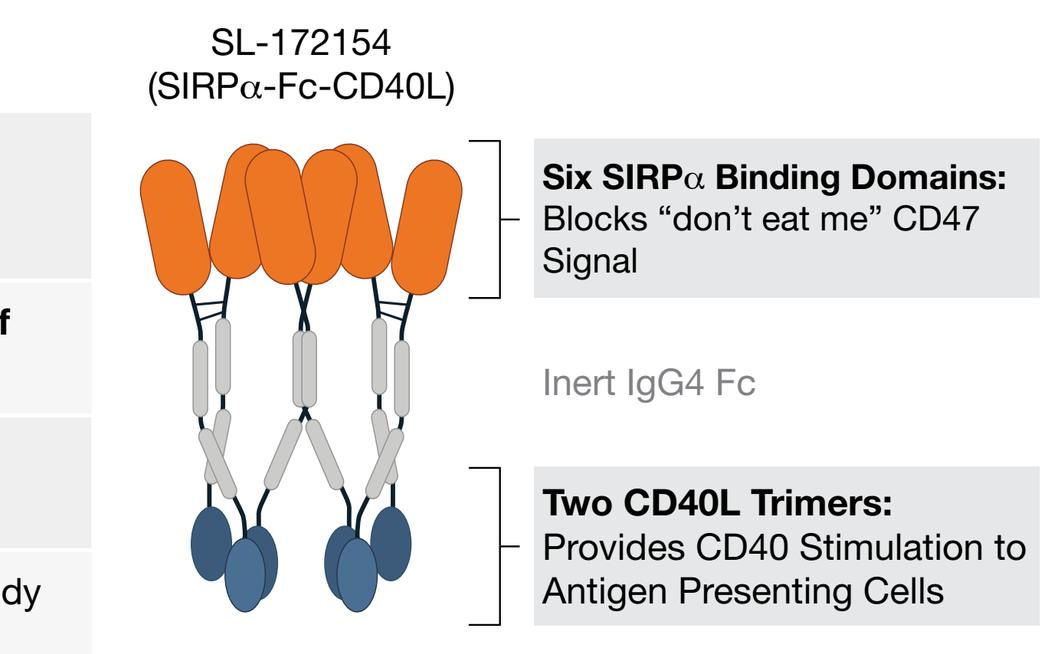

SL-172154
(SIRPα-Fc-CD40L)
Six SIRPα Binding Domains: Blocks "don't eat me" CD47 Signal
Inert IgG4 Fc
Two CD40L Trimers: Provides CD40 Stimulation to Antigen Presenting Cells

Figure 1: Pairing Rationale: SIRPα-Fc-CD40L

1. CD47 blockade disables the "don't eat me" checkpoint
2. Chemotherapy, ADC, or targeted mAbs provide the "eat me" signal on tumor cells to promote phagocytosis
3. CD40L activates antigen presenting cells, antigen presentation, and facilitates anti-tumor response

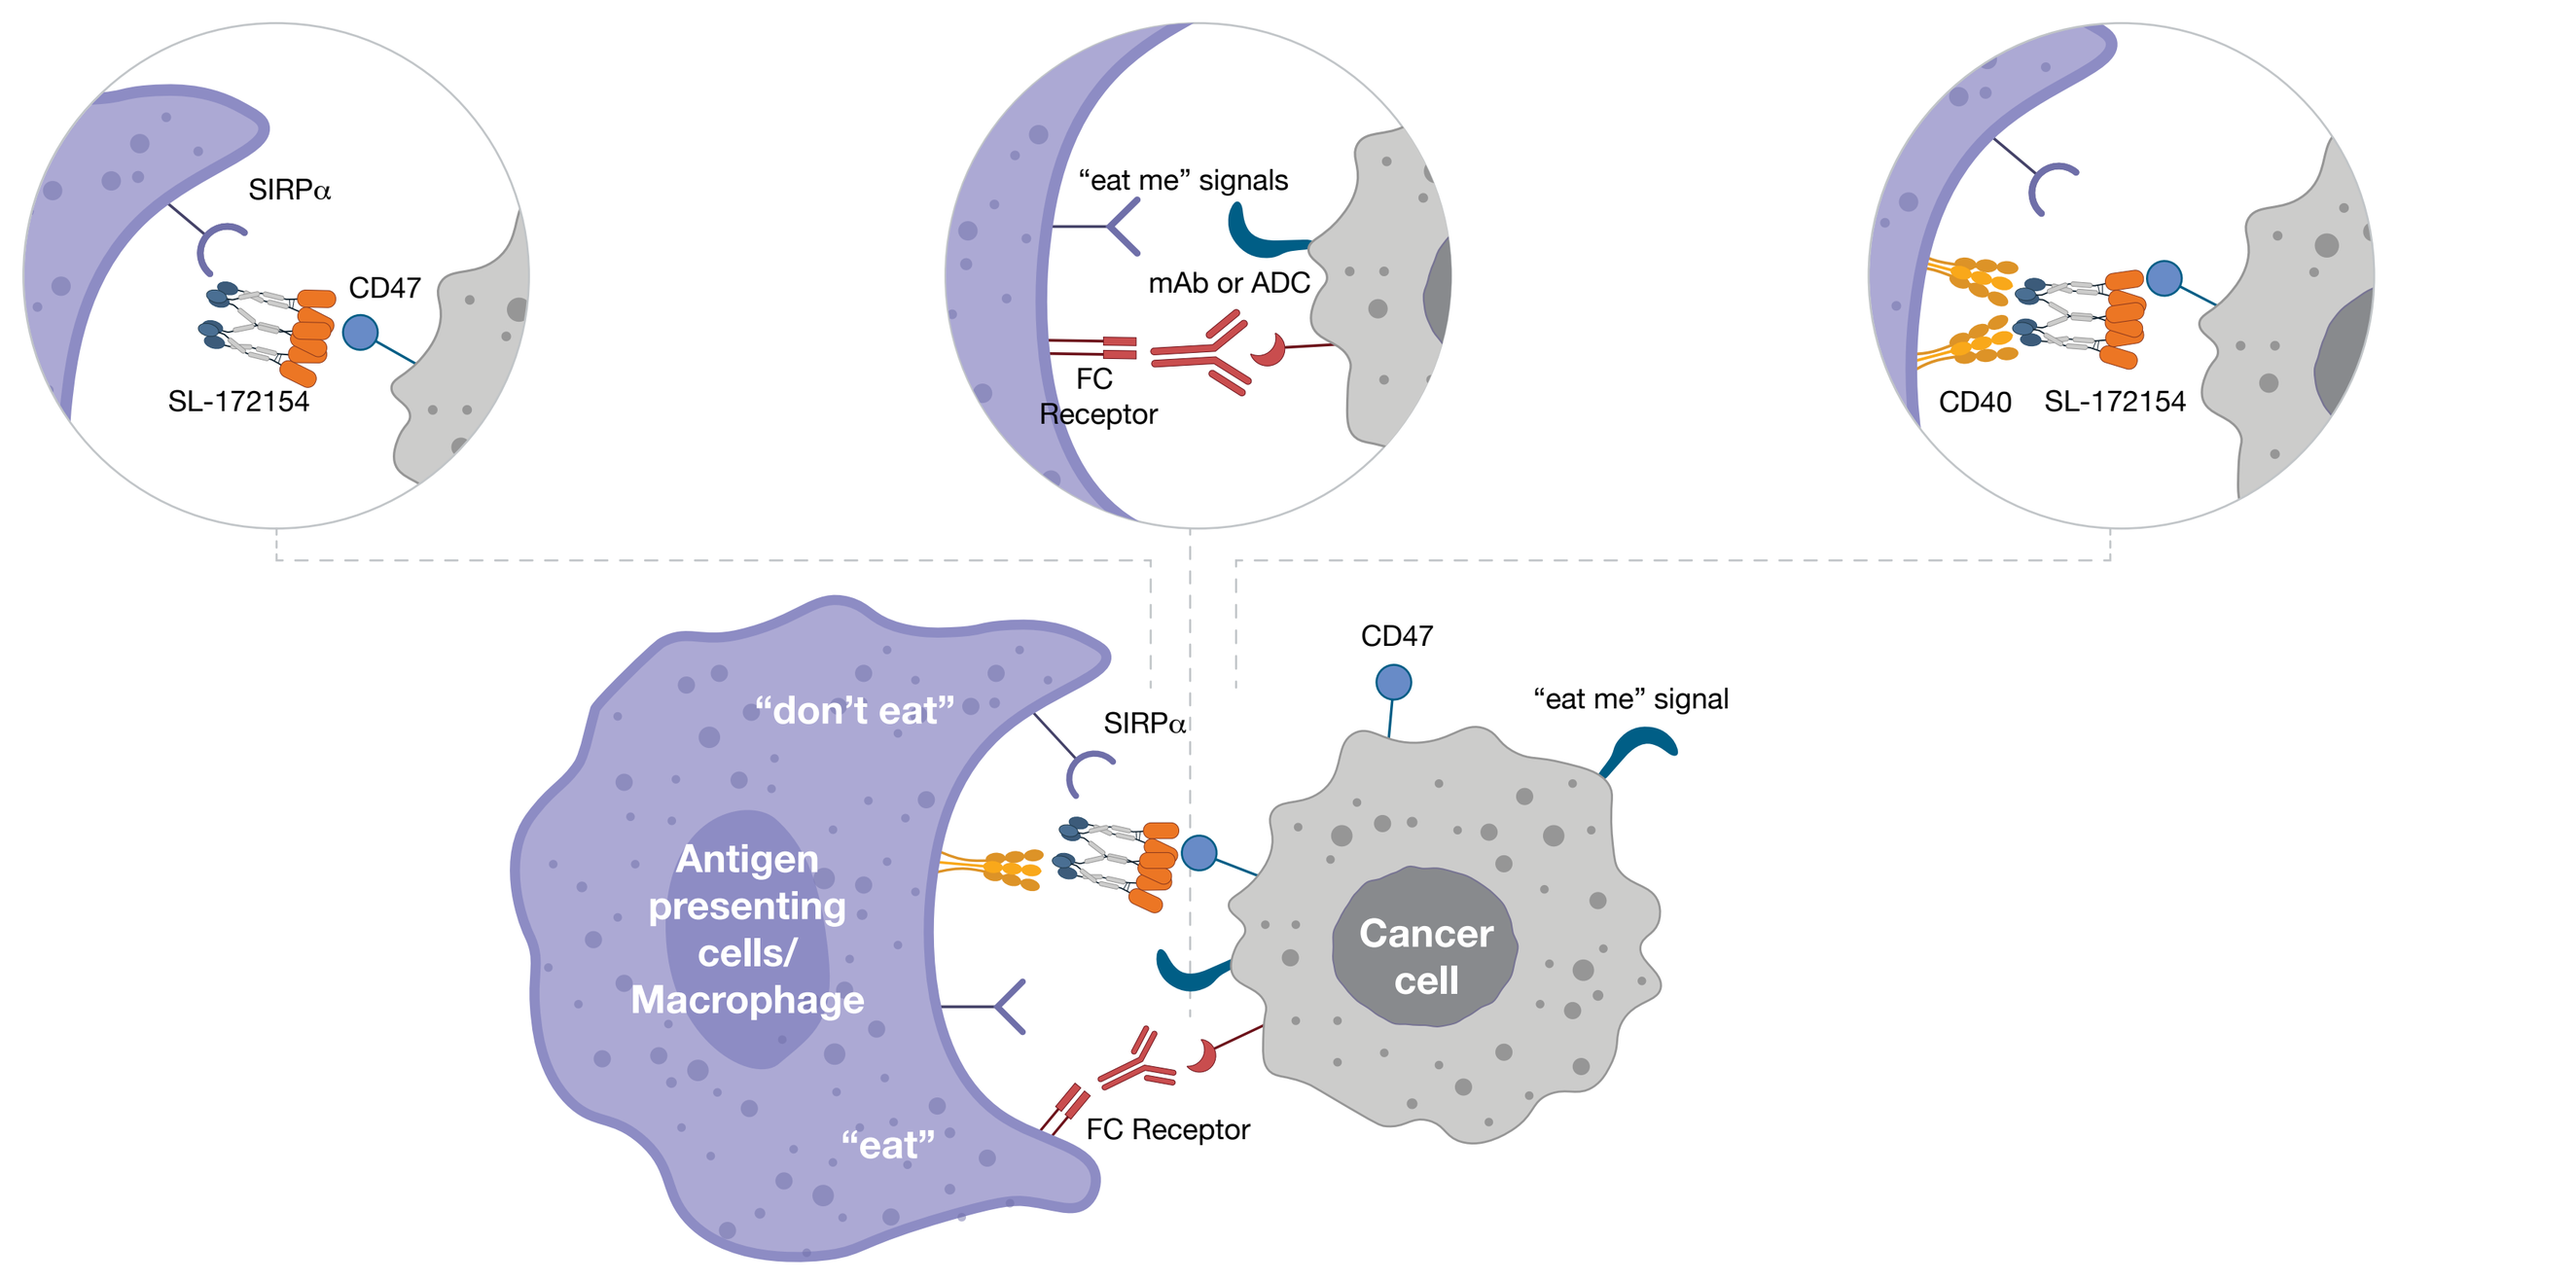

SIRPα
CD47
SL-172154
"eat me" signals
mAb or ADC
FC Receptor
CD40
"don't eat"
Antigen presenting cells/ Macrophage
"eat"
"eat me" signal
Cancer cell

Methods

Figure 2: Dose Escalation per Modified Toxicity Probability Interval (mTPI-2) Design (N=27)

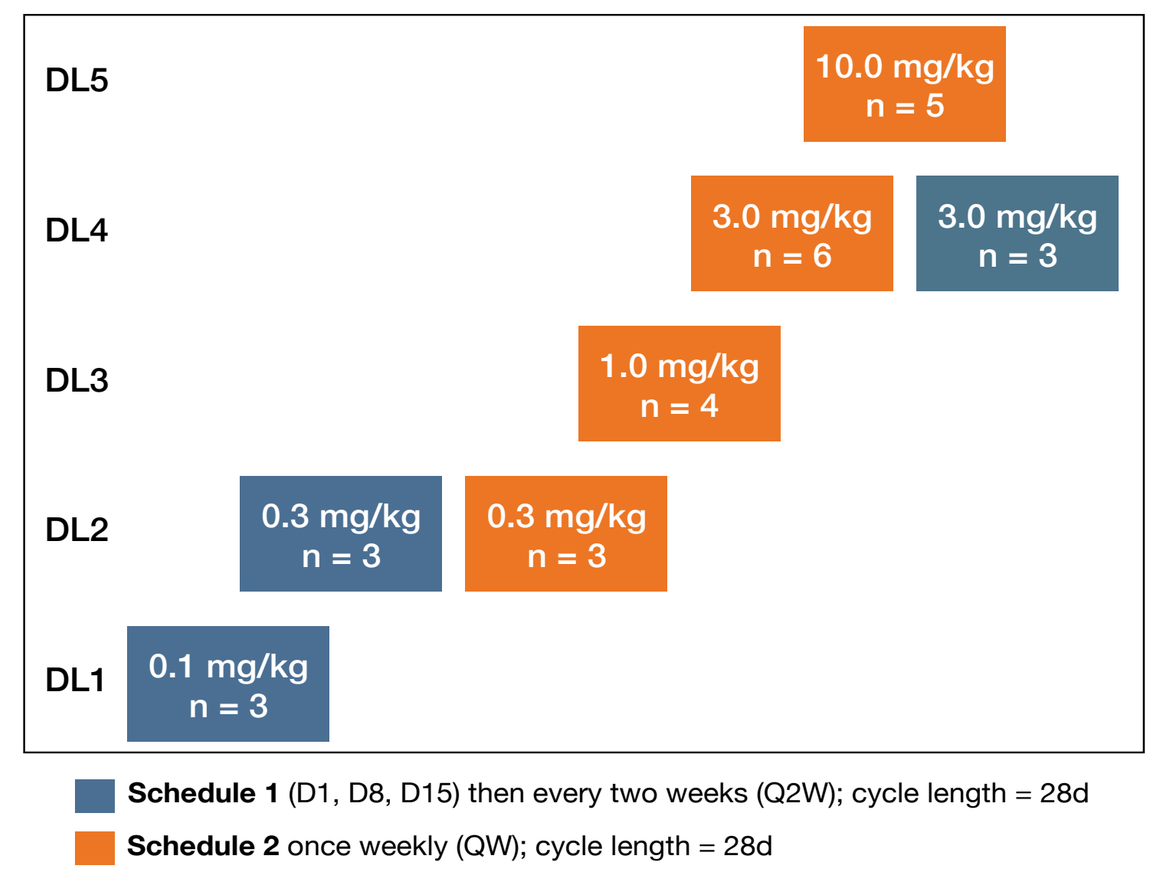

DL5
10.0 mg/kg n = 5
DL4
3.0 mg/kg n = 6
3.0 mg/kg n = 3
DL3
1.0 mg/kg n = 4
DL2
0.3 mg/kg n = 3
0.3 mg/kg n = 3
DL1
0.1 mg/kg n = 3
Schedule 1 (D1, D8, D15) then every two weeks (Q2W); cycle length = 28d
Schedule 2 once weekly (QW); cycle length = 28d

Key Study Objectives:

- Evaluate the safety and tolerability of SL-172154
- Identify a recommended Phase 2 dose (RP2D) for SL-172154
- Characterize pharmacokinetics (PK), pharmacodynamics (PD) and immunogenicity of SL-172154
- Assess preliminary anti-tumor activity

Enrollment:

- 27 subjects with platinum resistant ovarian cancers were treated with SL-172154, administered by IV infusion
- 9 subjects were treated on schedule 1
- 18 subjects were treated on schedule 2

Eligibility Criteria

Key Inclusion Criteria

- Histologically confirmed unresectable, locally advanced or metastatic ovarian cancer, primary peritoneal cancer or fallopian tube cancer
- Refractory to existing therapy(ies) and ineligible for further platinum therapy
- Age 18 years or older
- ECOG performance status of 0 or 1
- Measurable disease per RECIST v1.1

Key Exclusion Criteria

- Primary platinum refractory as defined by progressing during or within 1 month of upfront platinum therapy
- Prior treatment with an anti-CD47 or anti-SIRPα targeting agent or a CD40 agonist
- Documented history of autoimmune disease or active pneumonitis
- Concurrent use of systemic corticosteroids or other immunosuppressive medication

Results

Table 1. Study Population and Tumor Characteristics

A. Study Population

Total Number of Subjects	N=27
Median Age (range)	66 yrs (33-85)
Race	
White	21 (78%)
Black	3 (11%)
Other/not reported	3 (11%)
Baseline ECOG	
0	9 (33%)
1	18 (67%)
Median years from Initial Diagnosis (range)	4.9 (1-18)
Median number of prior systemic regimens for all intent (range)	4 (2-9)

B. Tumor Characteristics

Total Number of Subjects		N=27
		n (%)
Cancer type	Ovarian cancer	19 (70)
	Primary peritoneal cancer	4 (15)
	Fallopian tube cancer	4 (15)
FIGO stage	Stage III	11 (41)
	Stage IV	16 (59)
Grade	High grade	21 (78)
	Grade 3	1 (4)
	Unknown	5 (18)
WHO Histologic classification	Serous carcinoma	23 (85)
	Clear cell carcinoma	2 (7)
	Adenocarcinoma	2 (7)

FIGO = International Federation of Gynecology and Obstetrics; WHO = World Health Organization

Safety

Figure 3: All Causality AEs in ≥3 Subjects

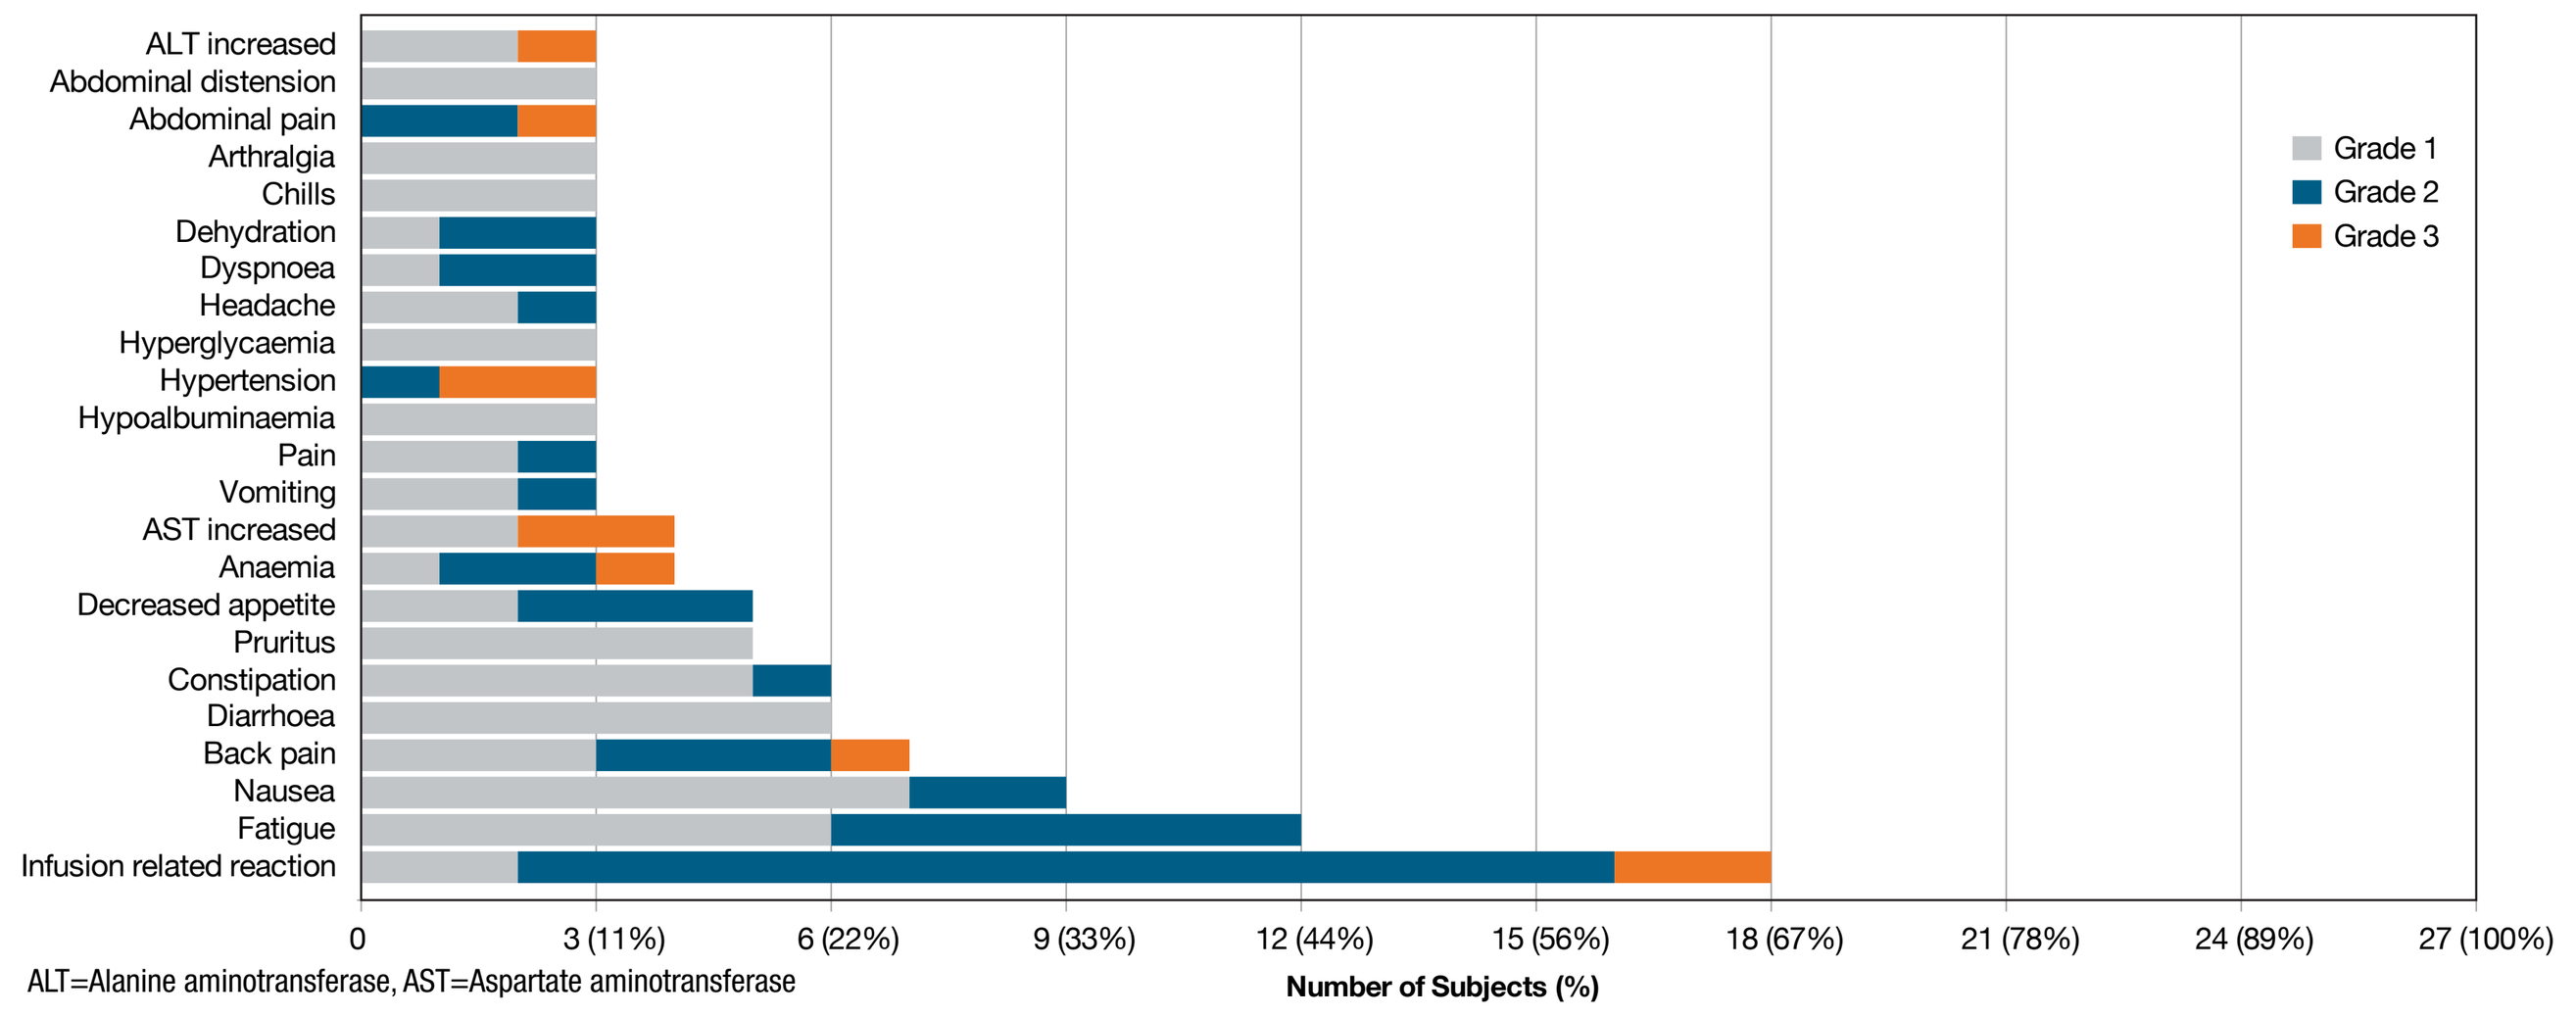

ALT increased
Abdominal distension
Abdominal pain
Arthralgia
Chills
Dehydration
Dyspnoea
Headache
Hyperglycaemia
Hypertension
Hypoalbuminaemia
Pain
Vomiting
AST increased
Anaemia
Decreased appetite
Pruritus
Constipation
Diarrhoea
Back pain
Nausea
Fatigue
Infusion related reaction
0
3 (11%)
6 (22%)
9 (33%)
12 (44%)
15 (56%)
18 (67%)
21 (78%)
24 (89%)
27 (100%)
Number of Subjects (%)
Grade 1
Grade 2
Grade 3

ALT=Alanine aminotransferase, AST=Aspartate aminotransferase

Table 2: Drug-related AEs (Any Grade) >10% Subjects

Total Number of Subjects (n=27)	Any grade
Any drug-related AEs	**24 (89%)**
Infusion Related Reactions	18 (67%)
Fatigue	9 (33%)
Nausea	6 (22%)
AST increased	4 (15%)
Decreased appetite	4 (15%)
Chills	3 (11%)
Dyspnoea	3 (11%)
ALT increased	3 (11%)

- A dose dependency in the overall AE profile was noted
- One DLT at 10.0 mg/kg of Grade (G) 3 ALT increase
- G3/4 drug-related AEs (>1 subject): AST increased (G3) and lymphopenia (G4), all were fully resolved with no dose modifications. Treatment was delayed for one subject for G3 ALT increase. These events occurred at doses of 3.0 mg/kg or 10.0 mg/kg.
- No fatal AEs
- No AEs that led to drug discontinuation

- SAEs: 7 events reported in 6 (22%) subjects: embolism (2), sepsis (2), large intestine infection, lower GI hemorrhage and small intestinal obstruction. None were considered related to SL-172154.

Infusion Related Reactions (IRR): 45 drug-related IRRs were reported from 18 subjects (67%).

- All drug-related IRRs were G1/2 events except one G3 IRR. Infusion was interrupted for G3 IRR and the subject was treated with steroids. The event resolved the same day.
- Severity and frequency of IRRs events were dose-dependent and correlated with rate of infusion
- At ≤ 3.0 mg/kg, IRRs were typically limited to Cycle 1 Day 1 (C1D1), C1D8. At 10.0 mg/kg IRRs were recurrent, occurring beyond C1D1, C1D8
- All IRRs occurred either during the infusion or within 2 hrs after the end of infusion, except one event which occurred between 2 and 24hrs after the end of infusion
- Most IRRs did not prevent completion of infusion, except 2 events
- Common symptoms of the IRR events variably included fever, chills, rigors, back pain, rash, hyper/ hypotension, tachycardia, nausea, and visual symptoms
- No IRRs led to discontinuation of SL-172154
- No evidence for cytokine release syndrome (no appreciable increases in IL-6 and TNFα)

Pharmacokinetics

Figure 4A: C1D1 concentration profile **Figure 4B**: Dose Normalized C_{max} **Figure 4C**: Dose Normalized AUC_{last}

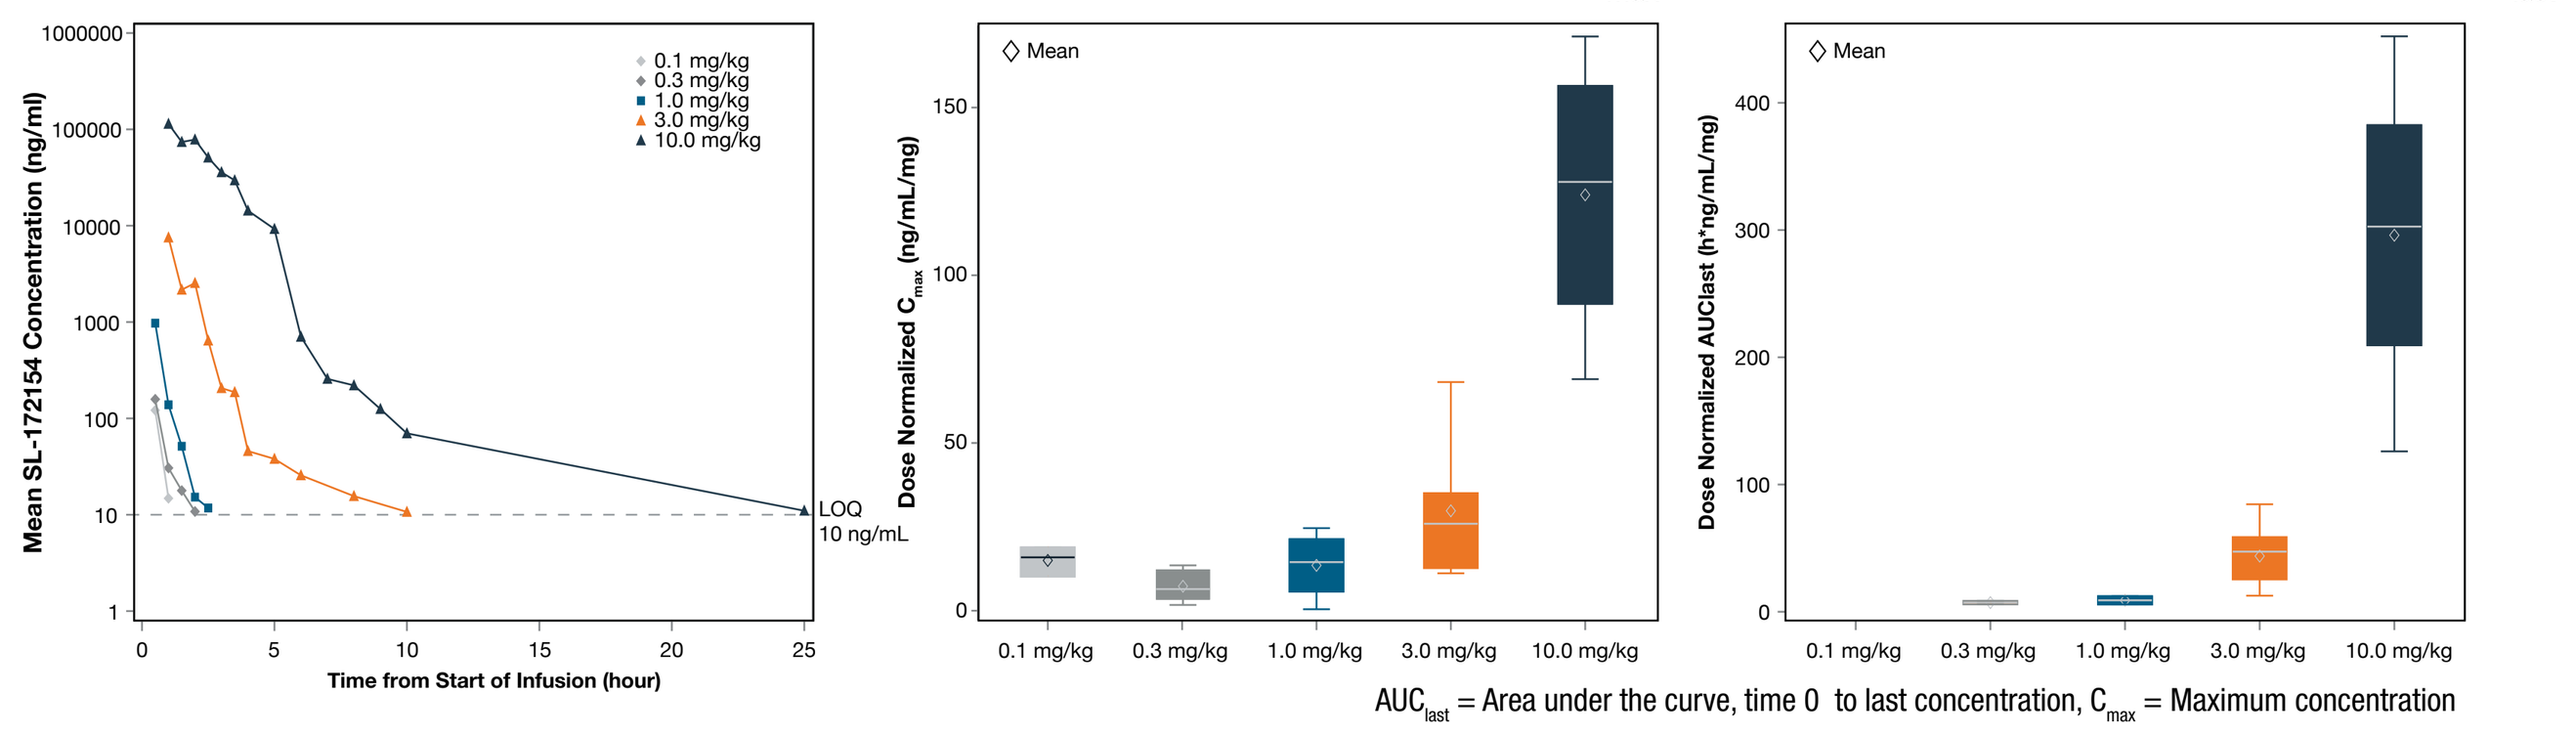

Mean SL-172154 Concentration (ng/ml)
Time from Start of Infusion (hour)
0.1 mg/kg
0.3 mg/kg
1.0 mg/kg
3.0 mg/kg
10.0 mg/kg
LOQ 10 ng/mL
Mean
Dose Normalized Cmax (ng/mL/mg)
Dose Normalized AUClast (h*ng/mL/mg)

AUC_{last} = Area under the curve, time 0 to last concentration, C_{max} = Maximum concentration

- SL-172154 exhibits greater than dose proportional pharmacokinetics at or above 3mg/kg.
- Nonlinear increase in C_{max} and AUC is largely due to target mediated drug disposition (binding to CD47 and CD40 receptors), and impacted by CD40 mediated pharmacodynamic effects such as rapid cell egress, receptor mediated endocytosis etc.

Immunogenicity

Low-titer anti-drug antibody (ADA) responses were detected in 8/25 evaluable subjects. No evidence of accelerated drug clearance, IRRs, or reductions in pharmacodynamic activity were detected in association with ADA.

Pharmacodynamics

Figure 5. CD47 Receptor Saturation on CD4 T Cells

Dose dependent increase in CD47 receptor saturation on CD4 T cells, approaching 100% by ≥ 3.0mg/kg dose.

Egress of CD4 T cells from the circulation occurs as a secondary pharmacodynamic effect of CD40 agonism, and likely contributes to the observed variability in CD47 receptor saturation.

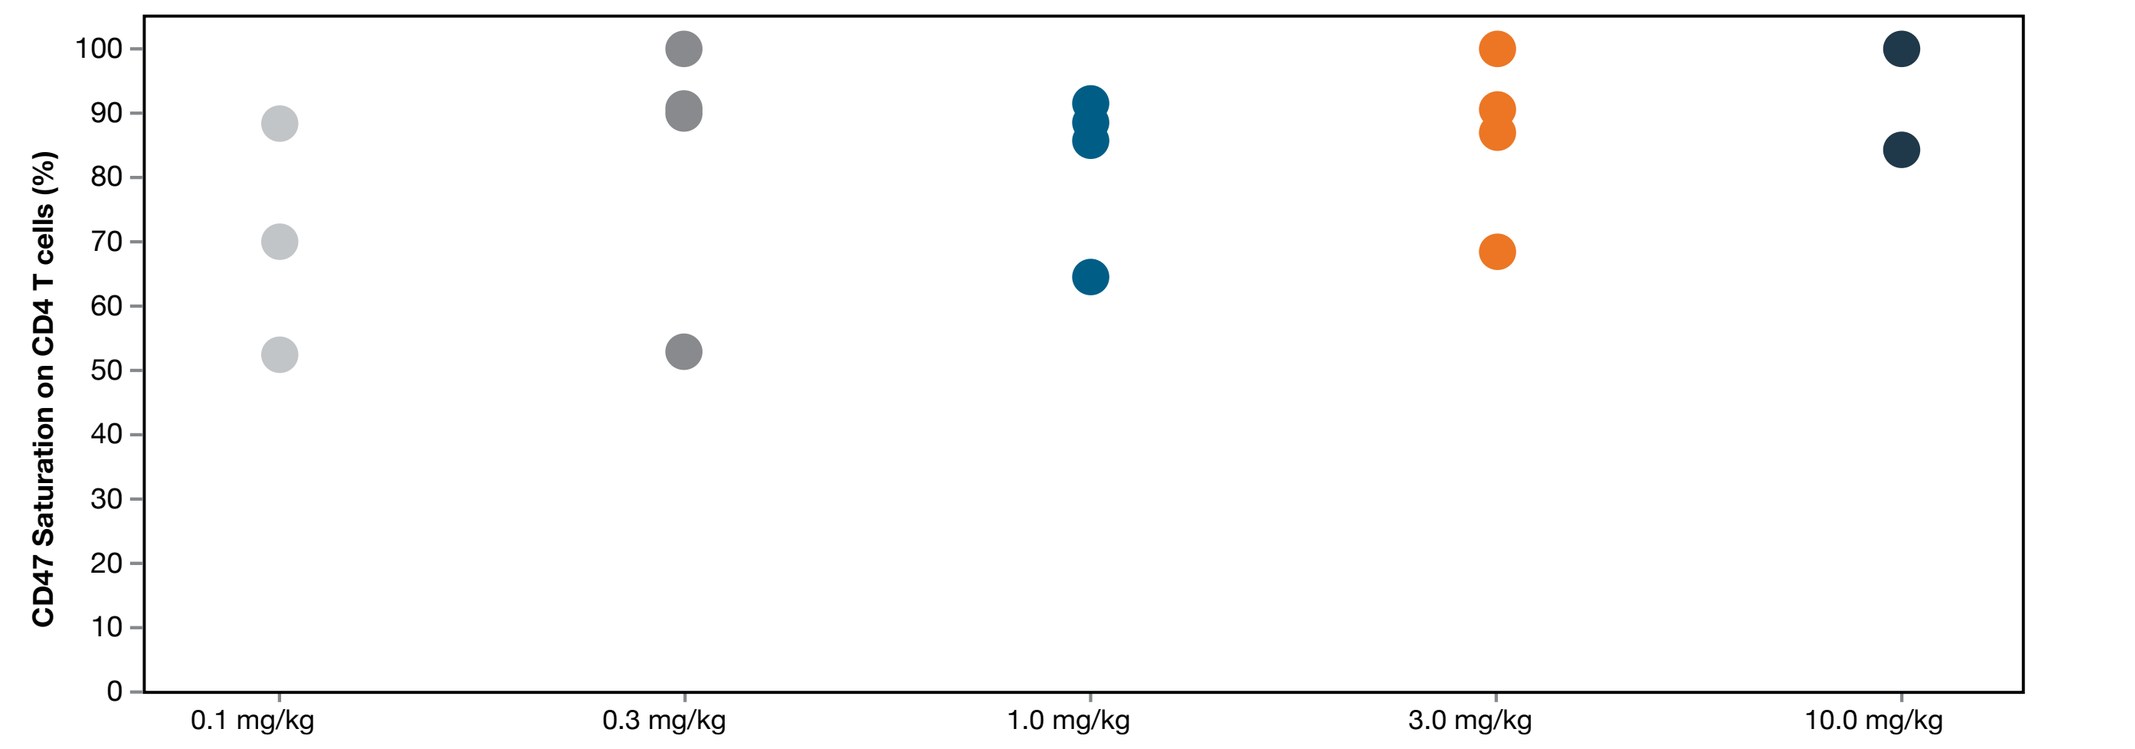

CD47 Saturation on CD4 T cells (%)
0.1 mg/kg
0.3 mg/kg
1.0 mg/kg
3.0 mg/kg
10.0 mg/kg

Figure 6. Dose-Dependent & Persistent Cytokine Responses: IL-12, CXCL-10, CCL-22, CXCL-8 and IL-10

A Dose dependent increase in interleukin 12 (IL-12) responses after the first infusion

B Mean IL-12 concentrations over time. Cyclical increases in IL-12, a mediator of TH1 proinflammatory responses is observed in study subjects.

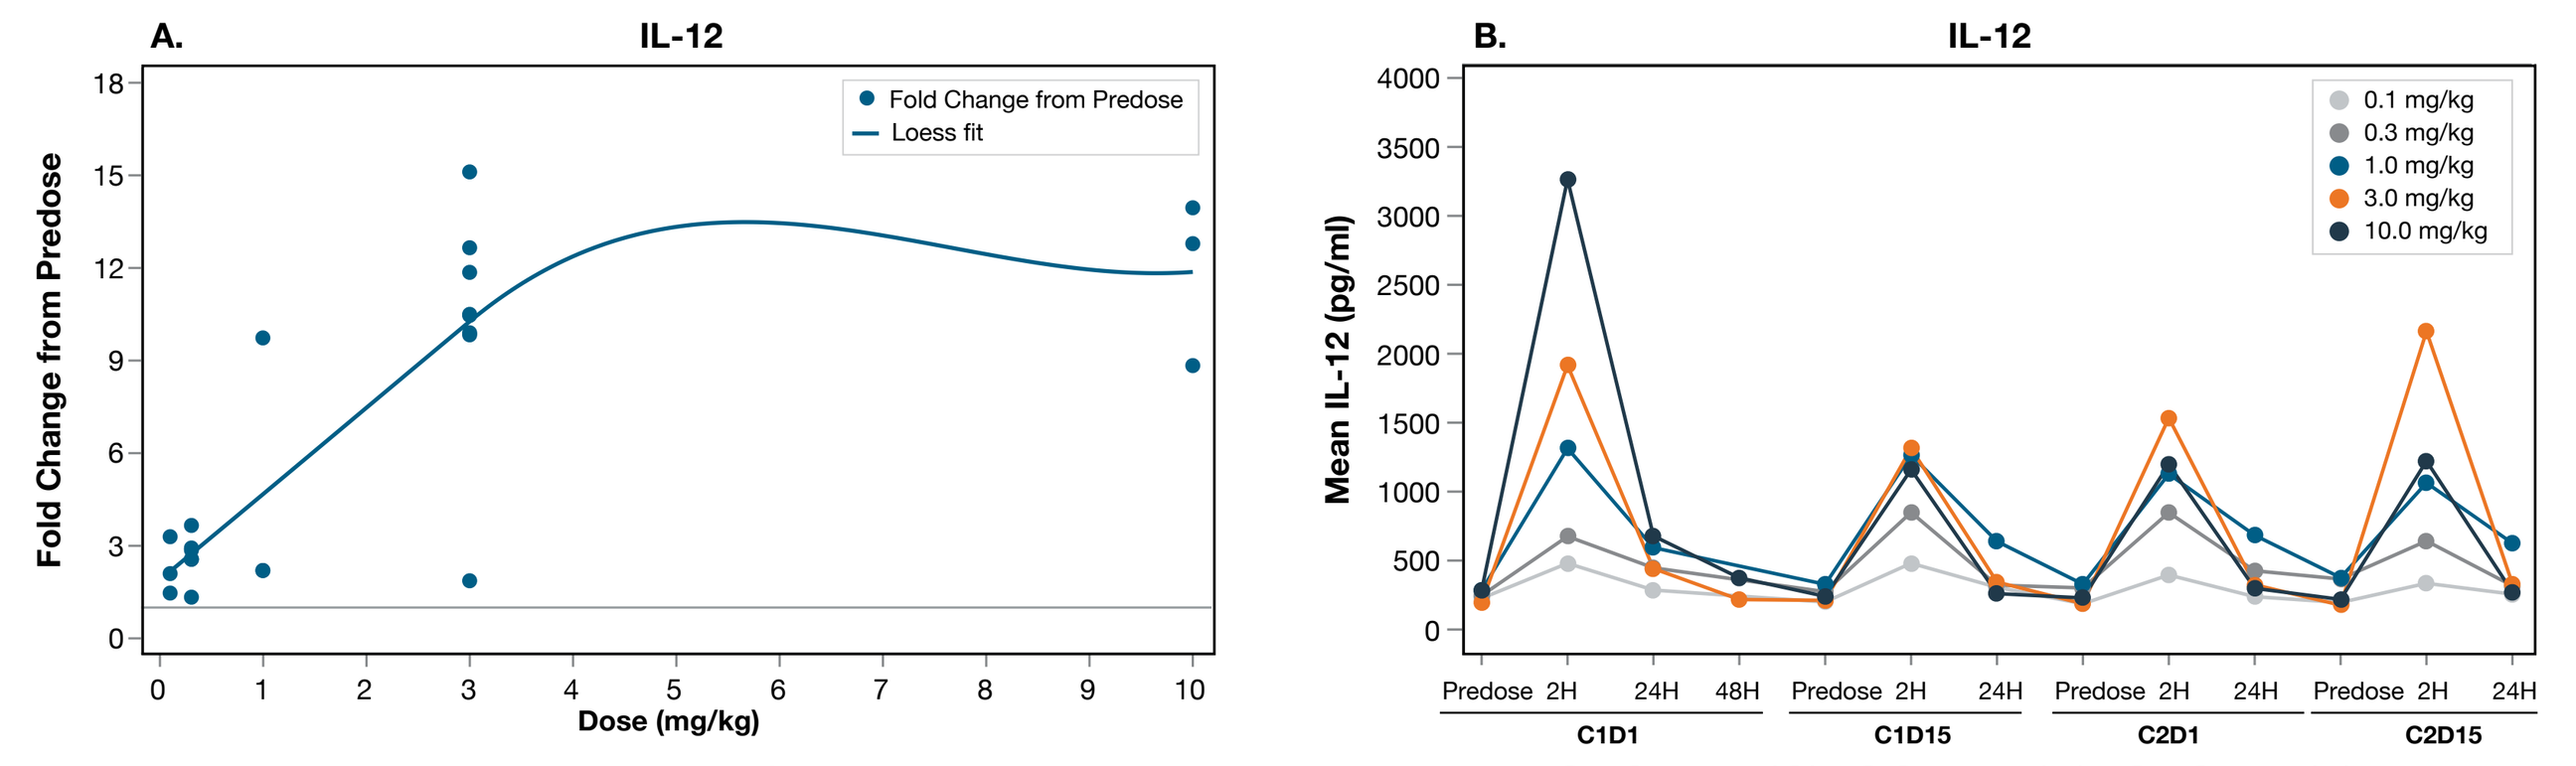

A.
IL-12
Fold Change from Predose
Loess fit
Dose (mg/kg)
B.
IL-12
Mean IL-12 (pg/ml)
0.1 mg/kg
0.3 mg/kg
1.0 mg/kg
3.0 mg/kg
10.0 mg/kg
Predose 2H 24H 48H
C1D1
C1D15
C2D1
C2D15

Dose-dependent increases of other on-target cytokines including CXCL-10 (C), CCL-22 (D), CXCL-8 (E), and IL-10 (F) are observed following SL-172154 administration. Similar to IL-12, these cytokine responses are persistent with each infusion.

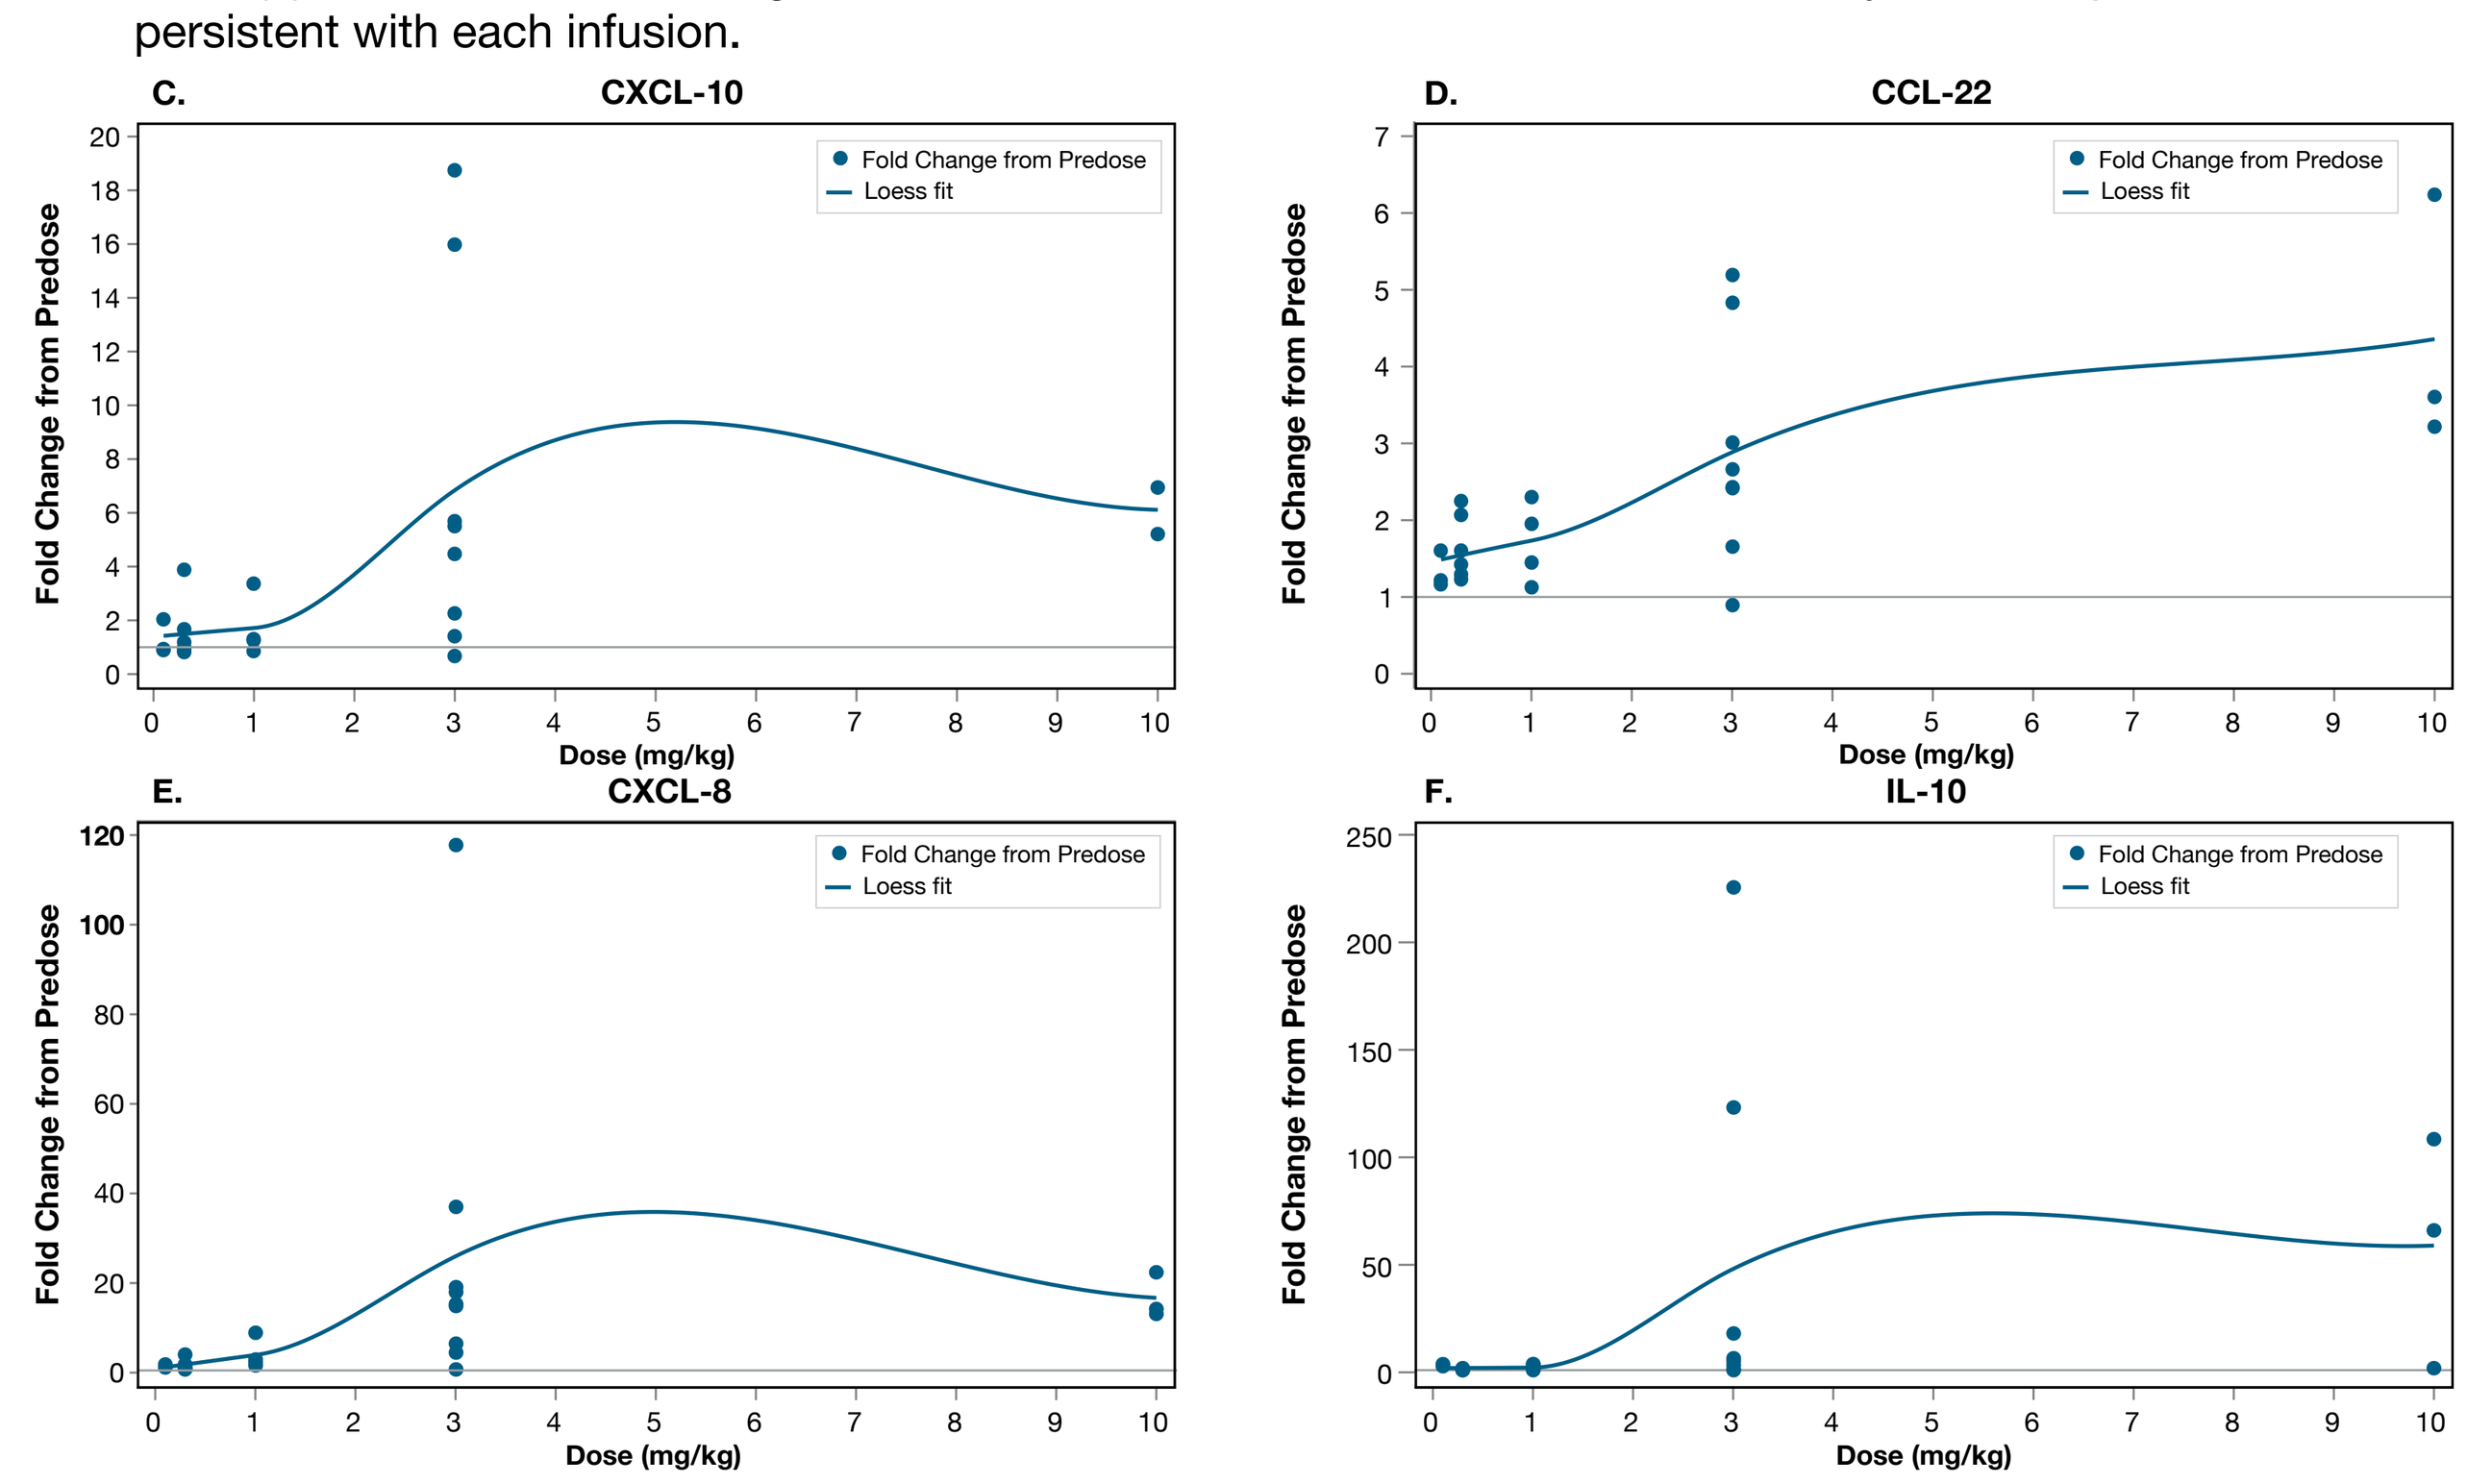

C.
CXCL-10
D.
CCL-22
E.
CXCL-8
F.
IL-10
Fold Change from Predose
Loess fit
Dose (mg/kg)

Figure 7. SL-172154 Stimulates Dose-Dependent Margination of B cells, Monocytes and CD4 T cells

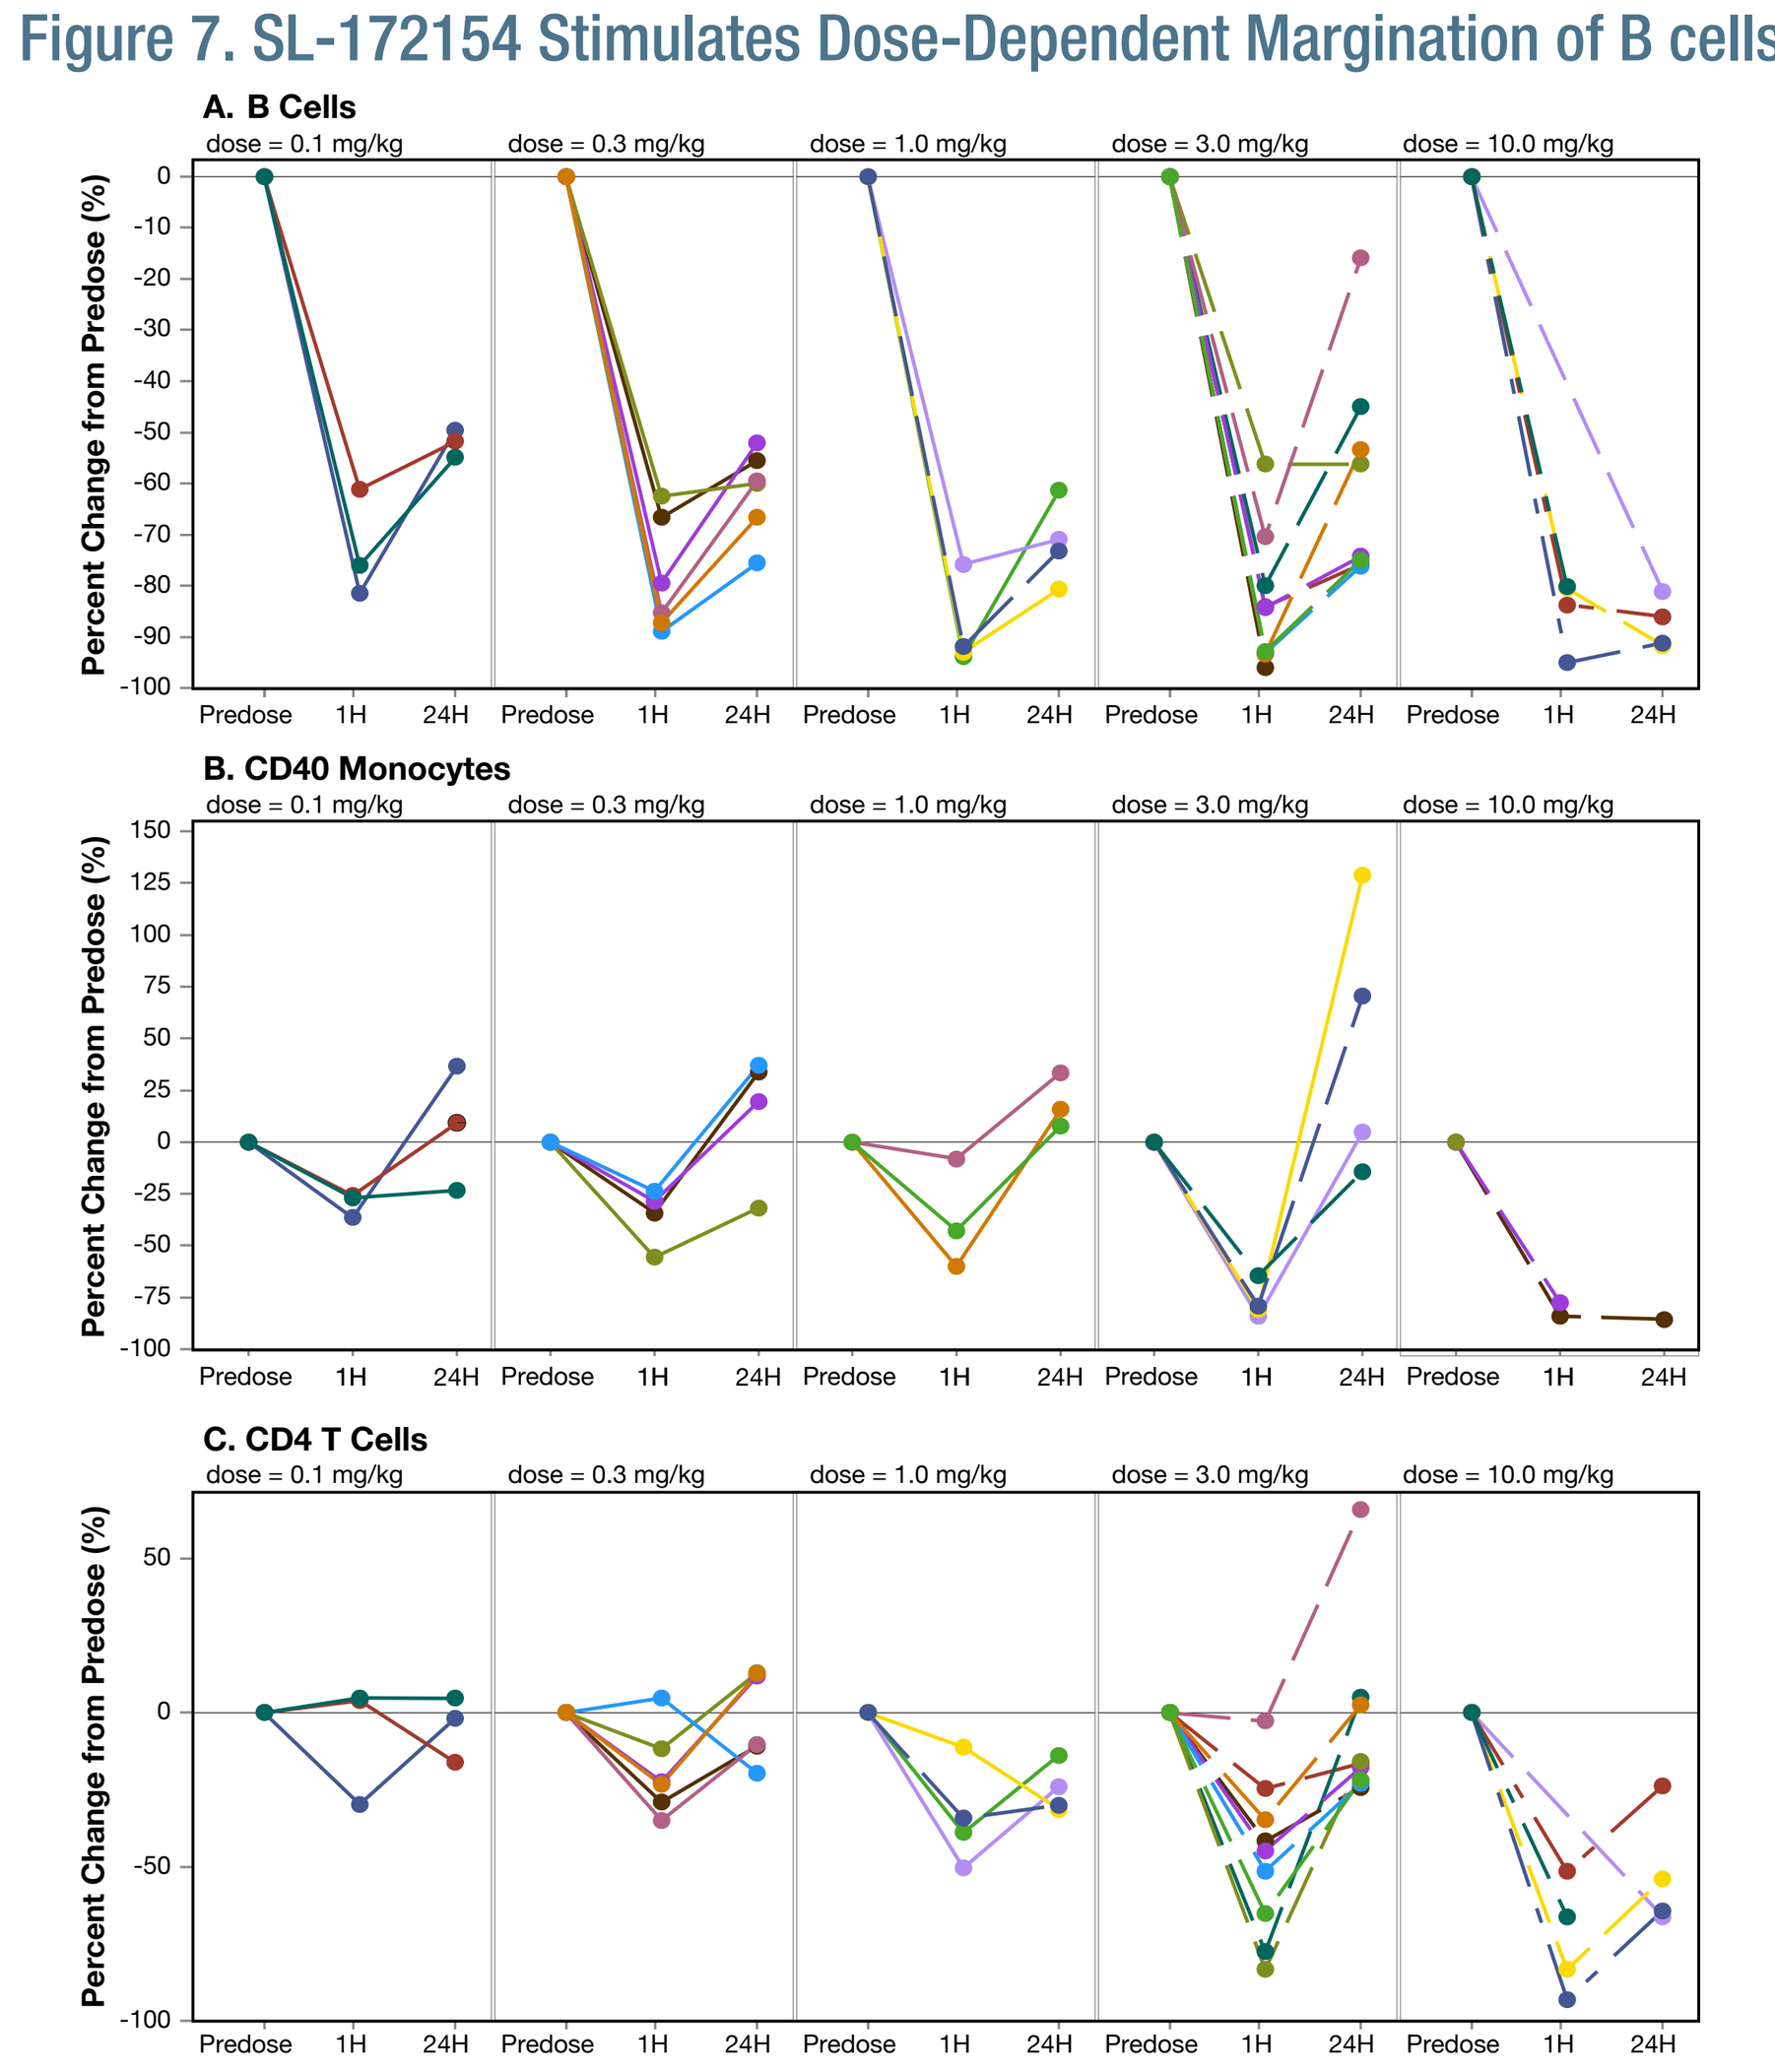

A. B Cells
B. CD40 Monocytes
C. CD4 T Cells
dose = 0.1 mg/kg
dose = 0.3 mg/kg
dose = 1.0 mg/kg
dose = 3.0 mg/kg
dose = 10.0 mg/kg
Percent Change from Predose (%)
Predose 1H 24H

B cells (A), CD40 expressing monocytes (B) and CD4 T cells (C) rapidly marginated from the circulation post infusion in a dose-dependent manner. Figure shows the percentage change from pre-dose at 1 hour and 24 hour post infusion on C1D1. Maximal margination is observed at 3 mg/kg and 10 mg/kg doses.

Figure 8. PK/PD Model Simulation of B cell Margination and IL-12 Stimulation Demonstrates Optimal Pharmacodynamic Activity at 3 mg/kg

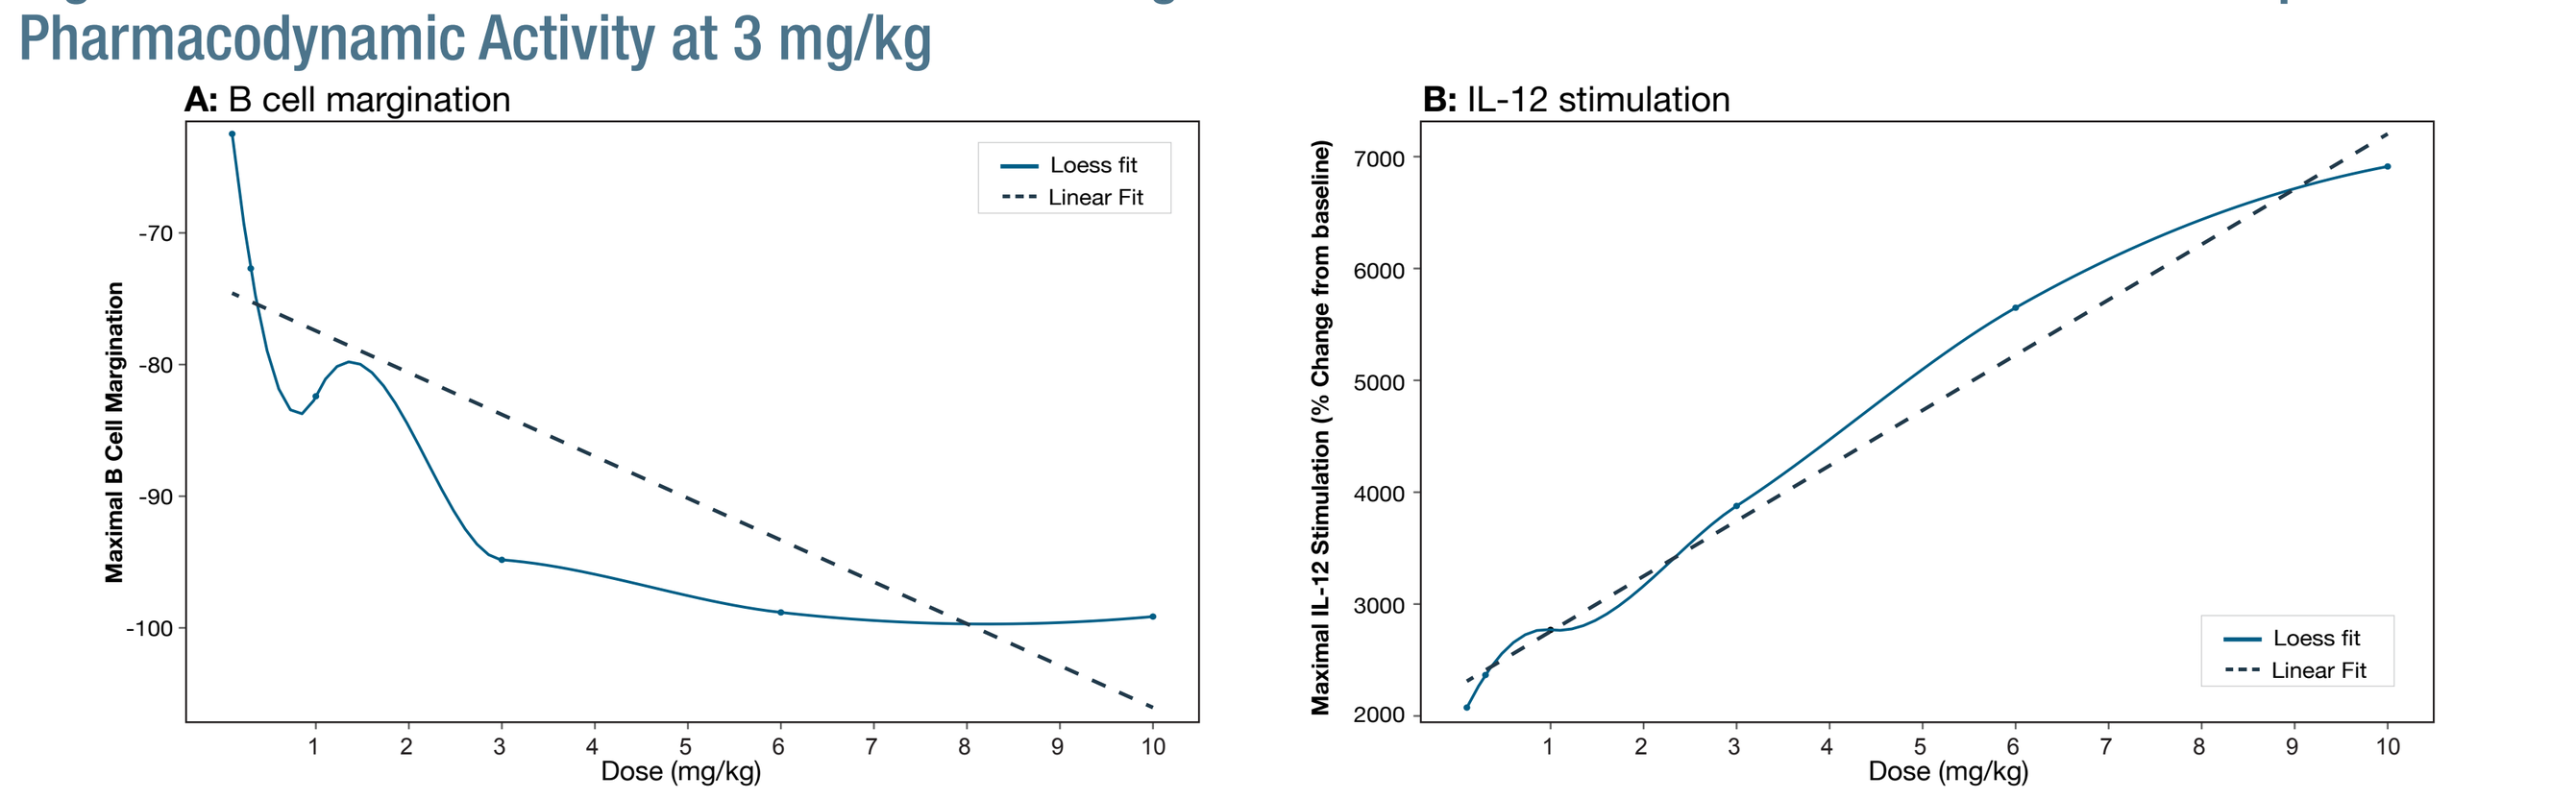

A: B cell margination
Maximal B Cell Margination
Loess fit
Linear Fit
Dose (mg/kg)
B: IL-12 stimulation
Maximal IL-12 Stimulation (% Change from baseline)

The PK/PD modeling using B cell margination (A) and IL-12 production (B) shows near maximal egress of B cells is reached at 3.0 mg/kg and doesn't change at 10.0 mg/kg. Likewise, the production of IL-12 is dose-dependent with a trend towards a plateau at >3.0 mg/kg.

Figure 9. SL-172154 Induces Polarization to M1 Macrophages and Infiltration of Cytotoxic T cells in the Tumor Nest

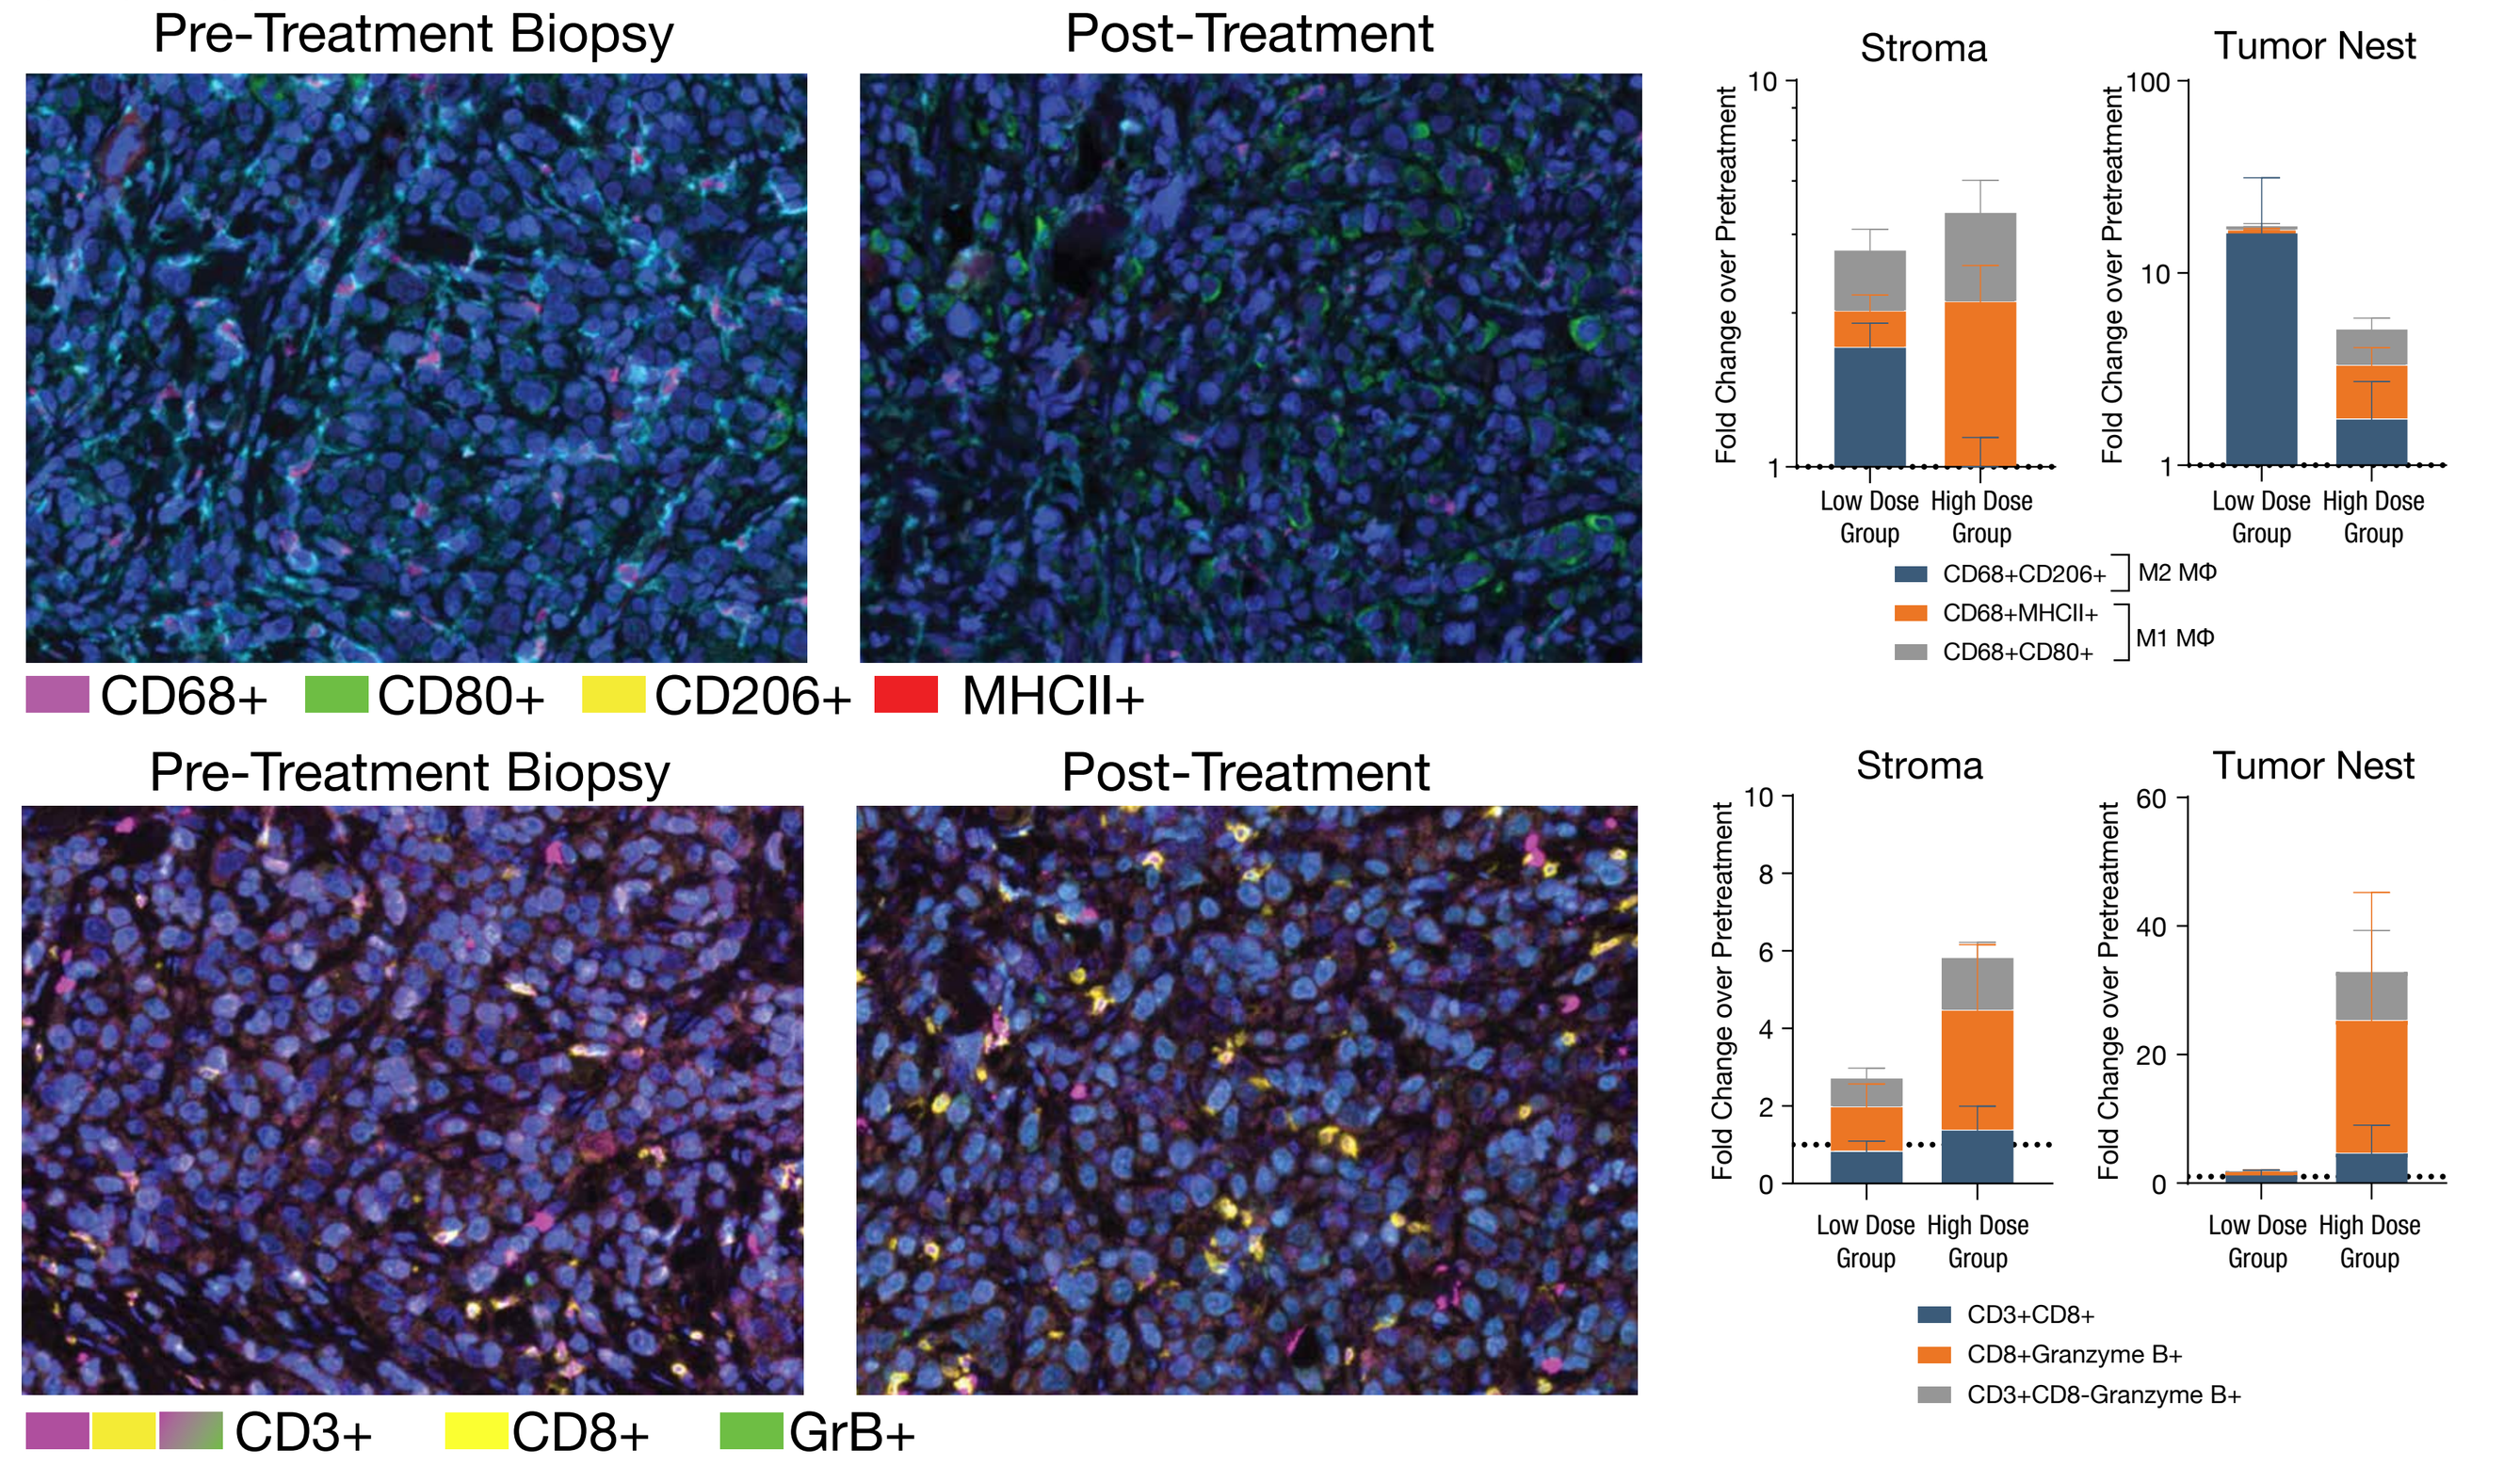

Pre-Treatment Biopsy
Post-Treatment
Stroma
Tumor Nest
Fold Change over Pretreatment
Low Dose Group
High Dose Group
CD68+CD206+
M2 MΦ
CD68+MHCII+
CD68+CD80+
M1 MΦ
CD68+
CD80+
CD206+
MHCII+
CD3+CD8+
CD8+Granzyme B+
CD3+CD8-Granzyme B+
CD3+
CD8+
GrB+

SL-172154 at high dose levels (3.0 mg/kg [n=4] and 10.0 mg/kg [n=1]) induced a shift in macrophages from an M2 to an M1 dominant phenotype both in the stroma and in the tumor nest. In contrast, at low dose levels (0.1 mg/kg [n=1], 0.3 and 1.0 mg/kg [n=3 each]) the M2 dominant phenotype persisted before and after treatment. The frequencies of granzyme B producing-T cells similarly increased in the tumor nest and tumor stroma in subjects in the high dose group compared to those in low dose group.

Antitumor Activity

Best response among 27 efficacy evaluable subjects with post-baseline scan was **Stable Disease (SD) in 6 (22%) subjects.** Median duration of SD was 138 days (range: 50 to 252 days).

Conclusions

- SL-172154 was overall well-tolerated in patients with heavily pretreated platinum resistant ovarian cancer. IRRs were the most common drug related AE and were readily manageable. A DLT of ALT increase was reported in one subject at 10.0 mg/kg dose.
- PK parameters for SL-172154 suggest greater than dose proportional increase in exposure at ≥ 3.0 mg/kg dose and target-mediated drug disposition via receptor binding.
- High receptor occupancy was observed for SL-172154 on CD47+ CD4 T cells. Binding of SL-172154 to CD40+ B cells and monocytes led to rapid activation and margination post infusion.
- Cyclical increases in innate and adaptive serum cytokines were consistent with CD40 receptor engagement and activation. There were no appreciable increases in IL-6 or TNFα, nor evidence of bell-shaped dose responses.
- Polarization towards an M1 macrophage phenotype was observed in the tumor biopsies at the higher doses of 3.0 mg/kg and 10.0 mg/kg consistent with the mechanism of action, and was associated with an increase in cytotoxic T cells in tumor nest.
- SL-172154 has been well-tolerated at doses which saturate both CD40 and CD47, with evidence of on-target PD activity which plateaus at dose of ≥ 3.0 mg/kg.
- The PK/PD modeling supports 3.0 mg/kg as an optimal dose for evaluation in combination studies.

Acknowledgements

- We extend our deep appreciation to study participants and their families
- This study is funded by Shattuck Labs, Inc. Austin, TX and Durham, NC, USA. Poster production assistance was provided by LifeSci Advisors LLC.

Ethics Approval

This study is being conducted in full conformity with the Declaration of Helsinki and was approved by all IRBs/ethics committees from each clinical site participating in the study. Specific approval numbers can be provided upon request.

References

Brightwell RM, et al. Gynecol Oncol 2016;143 (2):393.
Gupta V, et al. Cancers 2018;10 (10)366.
Wang H, et al. International journal of molecular sciences 2015;16 (2):3391.

Copies of this poster obtained through Quick Response (QR) Code are for personal use only and may not be reproduced without permission from ASCO or the author of this poster.